Abstract
1101

Telomerase inhibitor imetelstat in combination with the BCL-2 inhibitor venetoclax enhances apoptosis *in vitro* and increases survival *in vivo* in acute myeloid leukemia

Joshua J. Rusbuldt, Leopoldo Luistro, Diana Chin, Melissa Smith, Amy Wong, Margarita Romero, Aleksandra Rizo, Jacqueline Bussolari, Kate Sasser, Fei Huang
Janssen Research & Development

INTRODUCTION

Acute myeloid leukemia (AML) has limited treatment options apart from chemotherapy which ultimately provides only temporary reprieve from tumor burden. As such, improved therapies and novel mechanisms of targeting the disease are needed.

Like many tumors, AML cells overexpress hTERT, the catalytic subunit of telomerase. Telomerase canonically functions to elongate telomere ends and overcome the end replication problem. Telomerase also functions non-canonically to block apoptotic signals and promote self-renewal[1], though these mechanisms are less well understood.

AML cells also overexpress BCL-2, a negative regulator of apoptosis. BCL-2 inhibits pro-apoptotic proteins Bax and Bak thereby overriding death signals and allowing tumor cells to continue growing. BCL-2 and hTERT overexpression both correlate with poor disease prognosis in patients.

Imetelstat is a novel, first-in-class telomerase inhibitor with clinical activity in hematological malignancies[2,3,4]. Venetoclax (ABT-199) is a selective BCL-2 inhibitor approved for CLL and showing clinical benefit in AML[5].

MATERIALS AND METHODS

In vitro investigations in AML cell lines

- Five AML lines were treated with imetelstat and/or ABT-199 for 48 or 96 hours
- Cell viability was then assessed by cytometry using an Annexin V/Propidium Iodide (PI) assay, with viable cells defined as Annexin V^-/PI^-
- Additional samples were collected for RNA and protein extracts to evaluate hTERT expression by RT-qPCR and telomerase activity by qPCR-TRAP

In vivo study in the disseminated MOLM-13 AML model

- Female SCID-beige mice at 6-8 weeks were used as approved by the Institution Animal Care and Use Committee of Janssen R&D, Spring House, PA
- Mice were injected with 1 million MOLM-13 AML cells intravenously via lateral tail vein on Day 0 and randomly assigned to treatment groups of ten mice per group
- Starting on Day 1, imetelstat treatment was administered intraperitoneally every 3 to 4 days for 4 weeks
- Starting on Day 1, ABT-199 treatment was orally administered daily for 28 days
- Mice were monitored 2x weekly for body weight and adverse effects
- Median survival was determined and percent increased life span (% ILS) were calculated compared to vehicle controls

Ex vivo efficacy in purified PBMCs from AML patient blood

- PBMCs were Ficoll purified from whole blood of four AML patients
- Cells were treated for 16 or 40 hours and assessed for viability by flow cytometry as above

HYPOTHESIS

Preclinical data suggest hTERT downregulation induces apoptosis via disruptive interactions between hTERT and BCL-2 in tumor cells[6].

Inhibition of telomerase and BCL-2 in conjunction will yield greater anti-tumor effect in AML as compared to either treatment alone

Schematic at right adapted from reference 6.

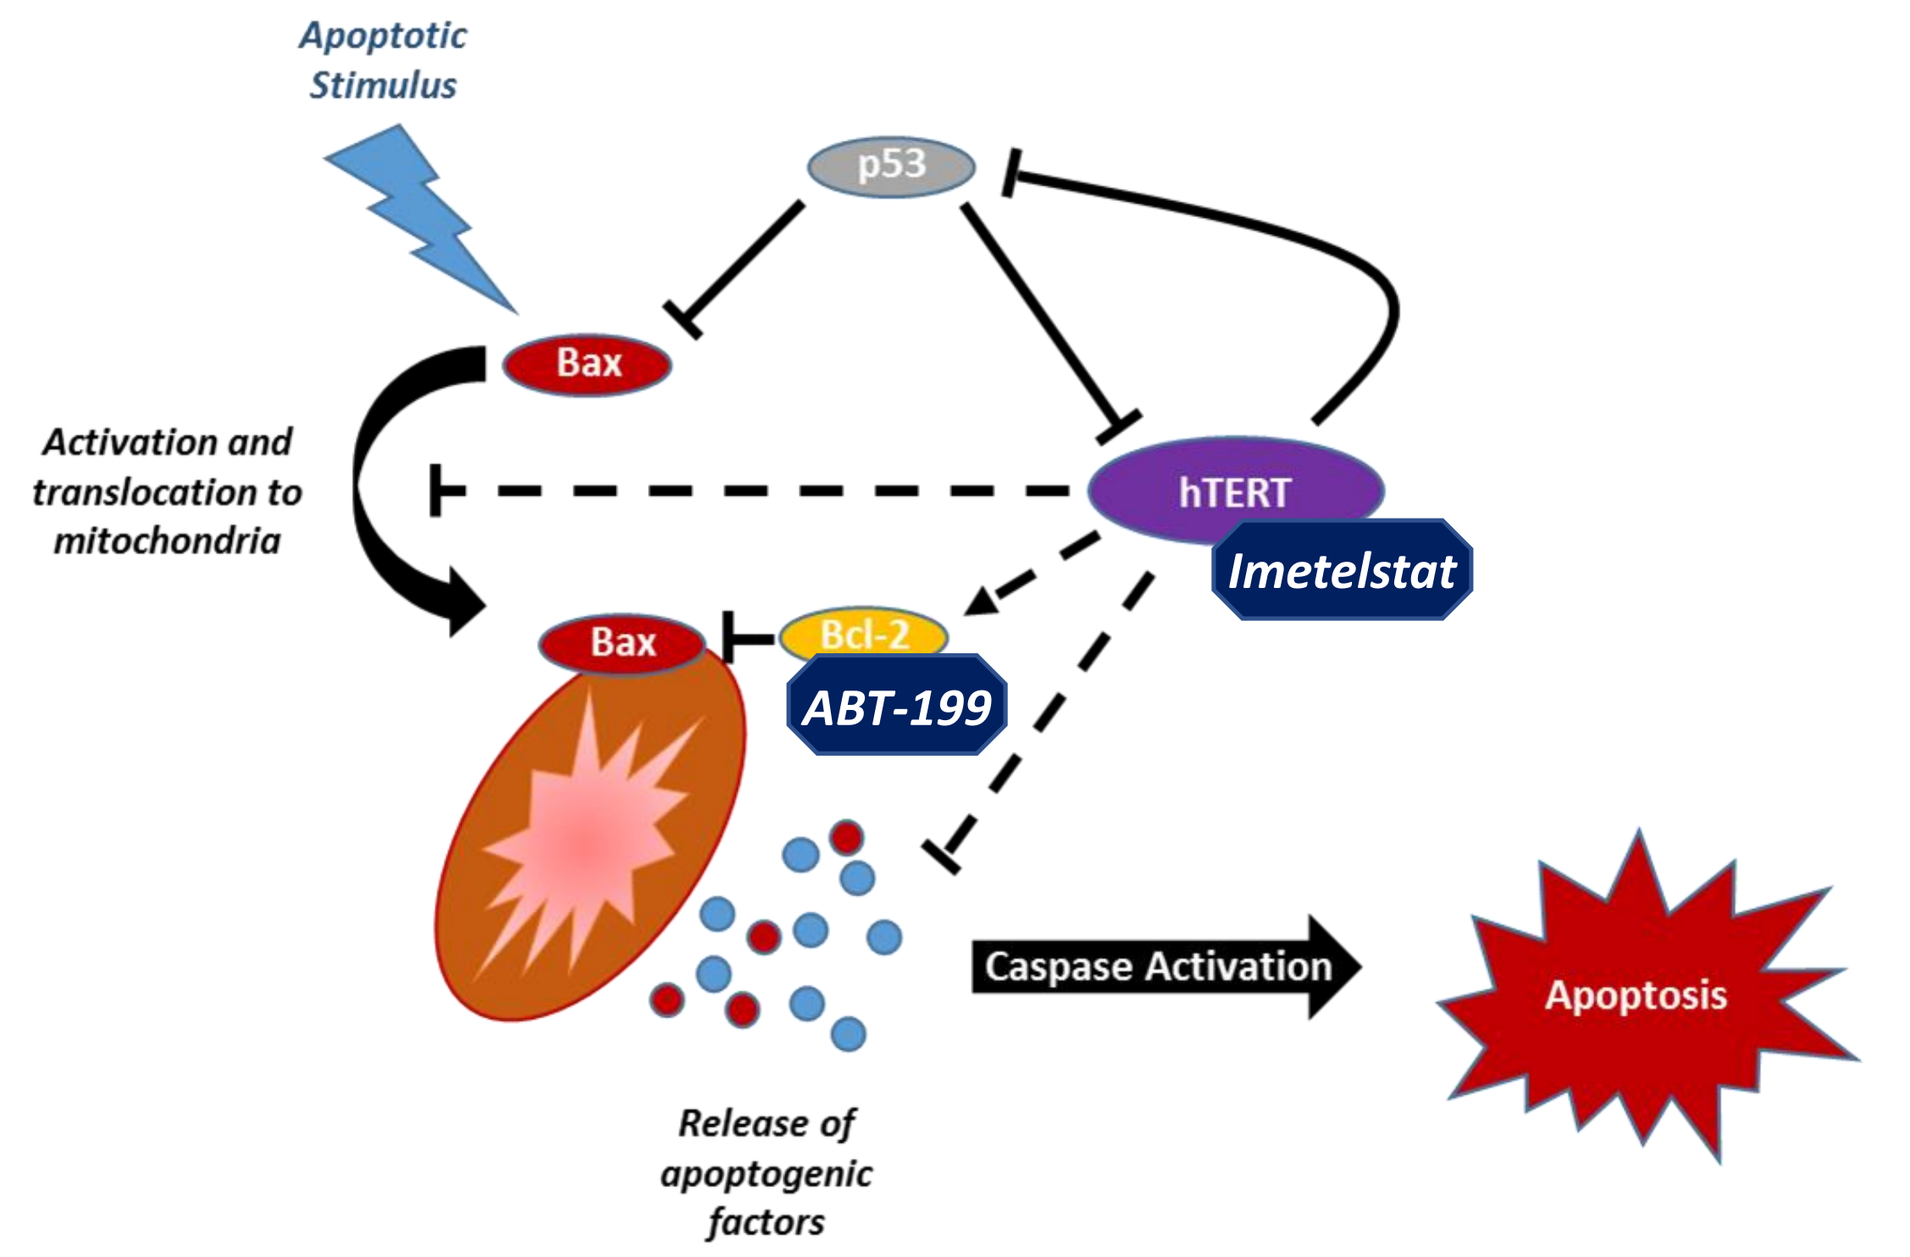
Apoptotic Stimulus
p53
Bax
Activation and translocation to mitochondria
hTERT
Imetelstat
Bax
Bcl-2
ABT-199
Caspase Activation
Apoptosis
Release of apoptogenic factors

RESULTS

In vitro combination synergy assessment

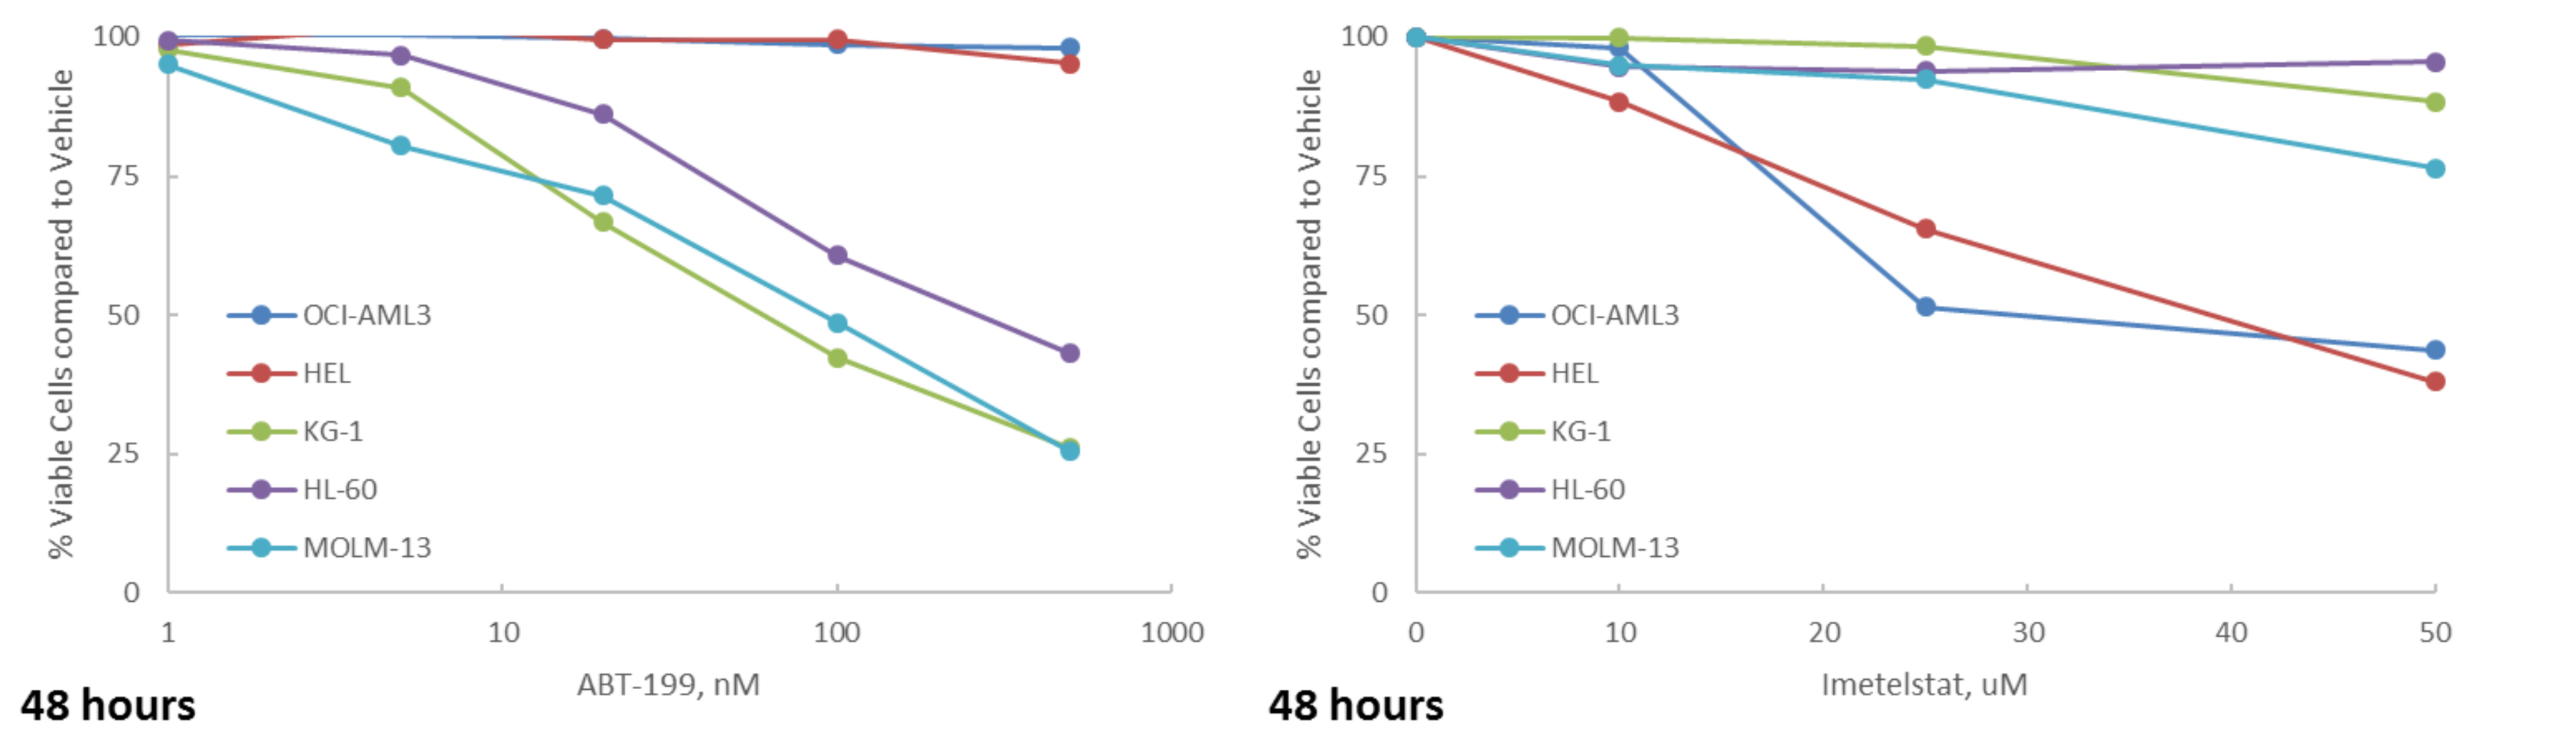
% Viable Cells compared to Vehicle
OCI-AML3
HEL
KG-1
HL-60
MOLM-13
ABT-199, nM
48 hours
Imetelstat, uM
48 hours

Figure 1. Single-agent response of AML cell lines *in vitro*
Three of five lines are sensitive to ABT-199 at 48 hour exposure (left), consistent with the literature[5]. ABT-199 insensitive lines show moderate sensitivity to imetelstat (right).

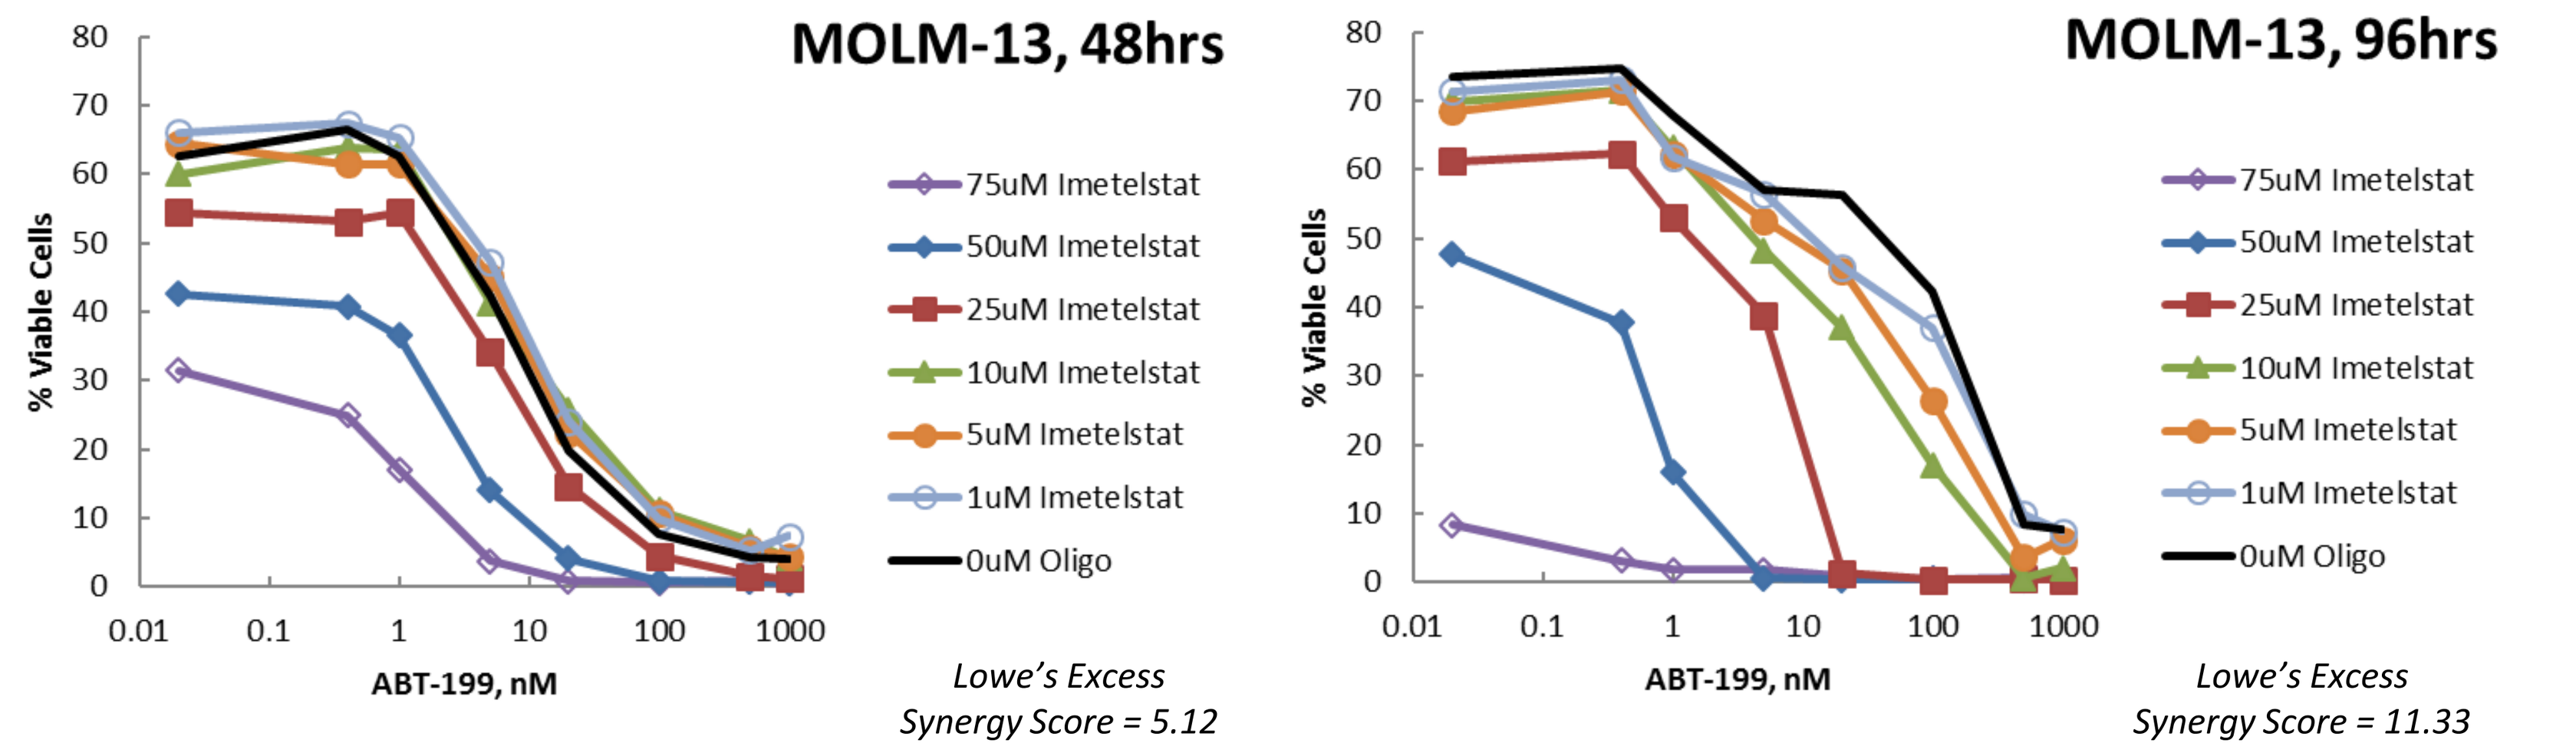
MOLM-13, 48hrs
% Viable Cells
75uM Imetelstat
50uM Imetelstat
25uM Imetelstat
10uM Imetelstat
5uM Imetelstat
1uM Imetelstat
0uM Oligo
ABT-199, nM
Lowe's Excess Synergy Score = 5.12
MOLM-13, 96hrs
ABT-199, nM
Lowe's Excess Synergy Score = 11.33

Figure 2. Imetelstat in combination with ABT-199 is synergistic in AML cells *in vitro*
Imetelstat showed both dose and time dependent activity in enhancing cell killing of ABT-199. Synergy scores are calculated using the Horizon Chalice™ Analyzer Software[7].

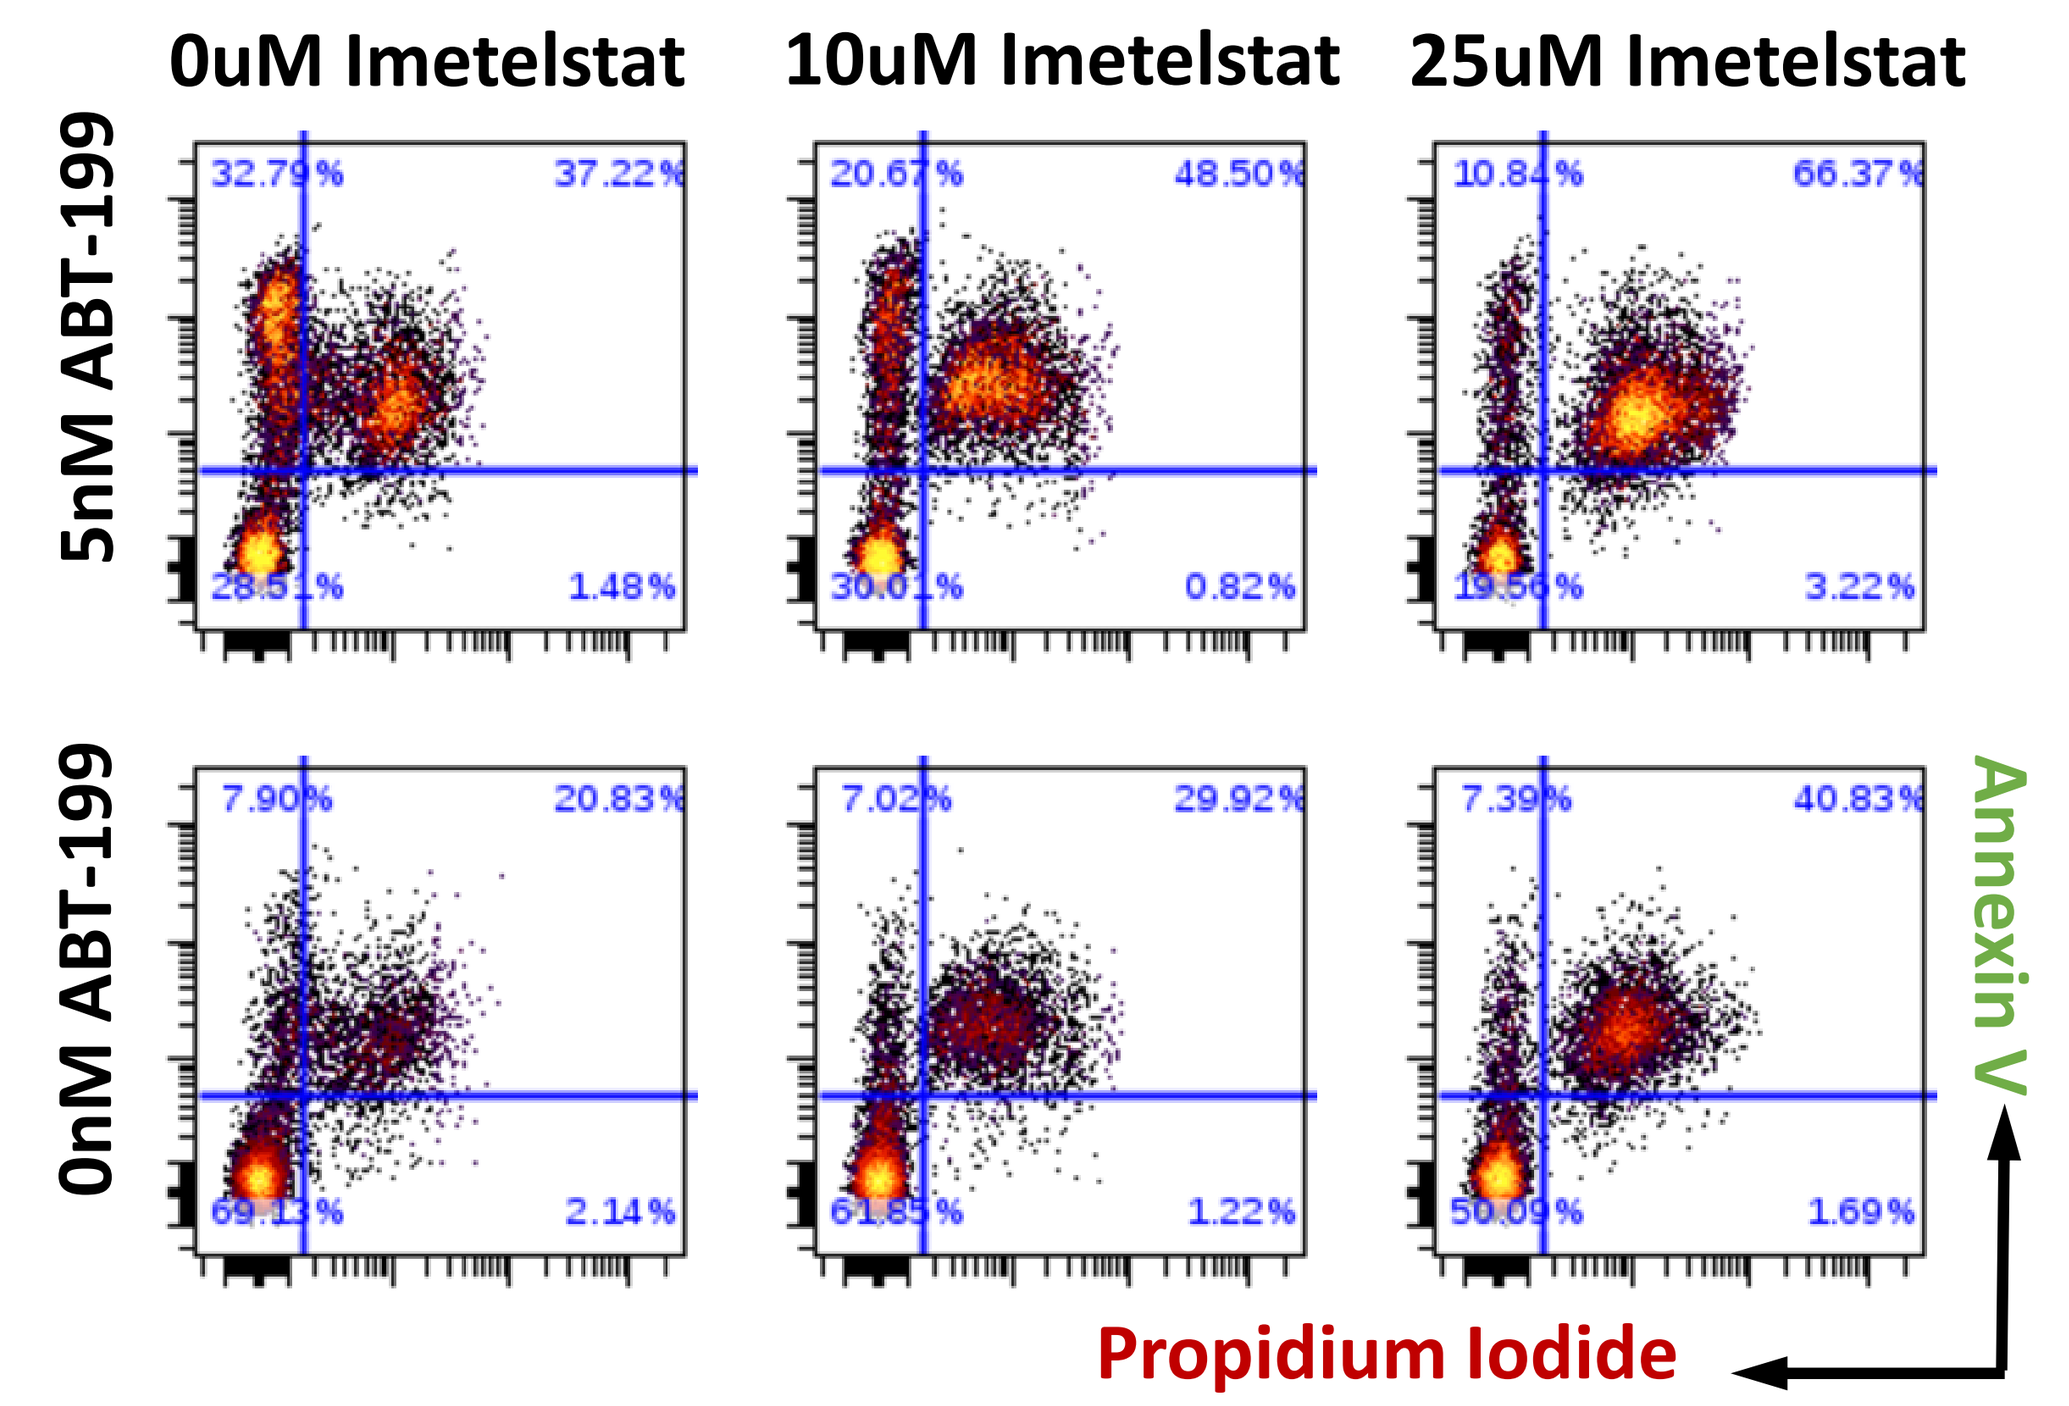
0uM Imetelstat
10uM Imetelstat
25uM Imetelstat
5nM ABT-199
0nM ABT-199
Annexin V
Propidium Iodide

Figure 3. Synergistic induction of apoptosis by combining ABT-199 with imetelstat
Imetelstat pushes early apoptotic cells (Annexin V+/PI-) induced by ABT-199 into late apoptosis and cell death (Annexin V+/PI+). Dot plots show differences in cells treated with 5nM ABT-199 in combination with 10 μM and 25 μM imetelstat versus untreated controls.

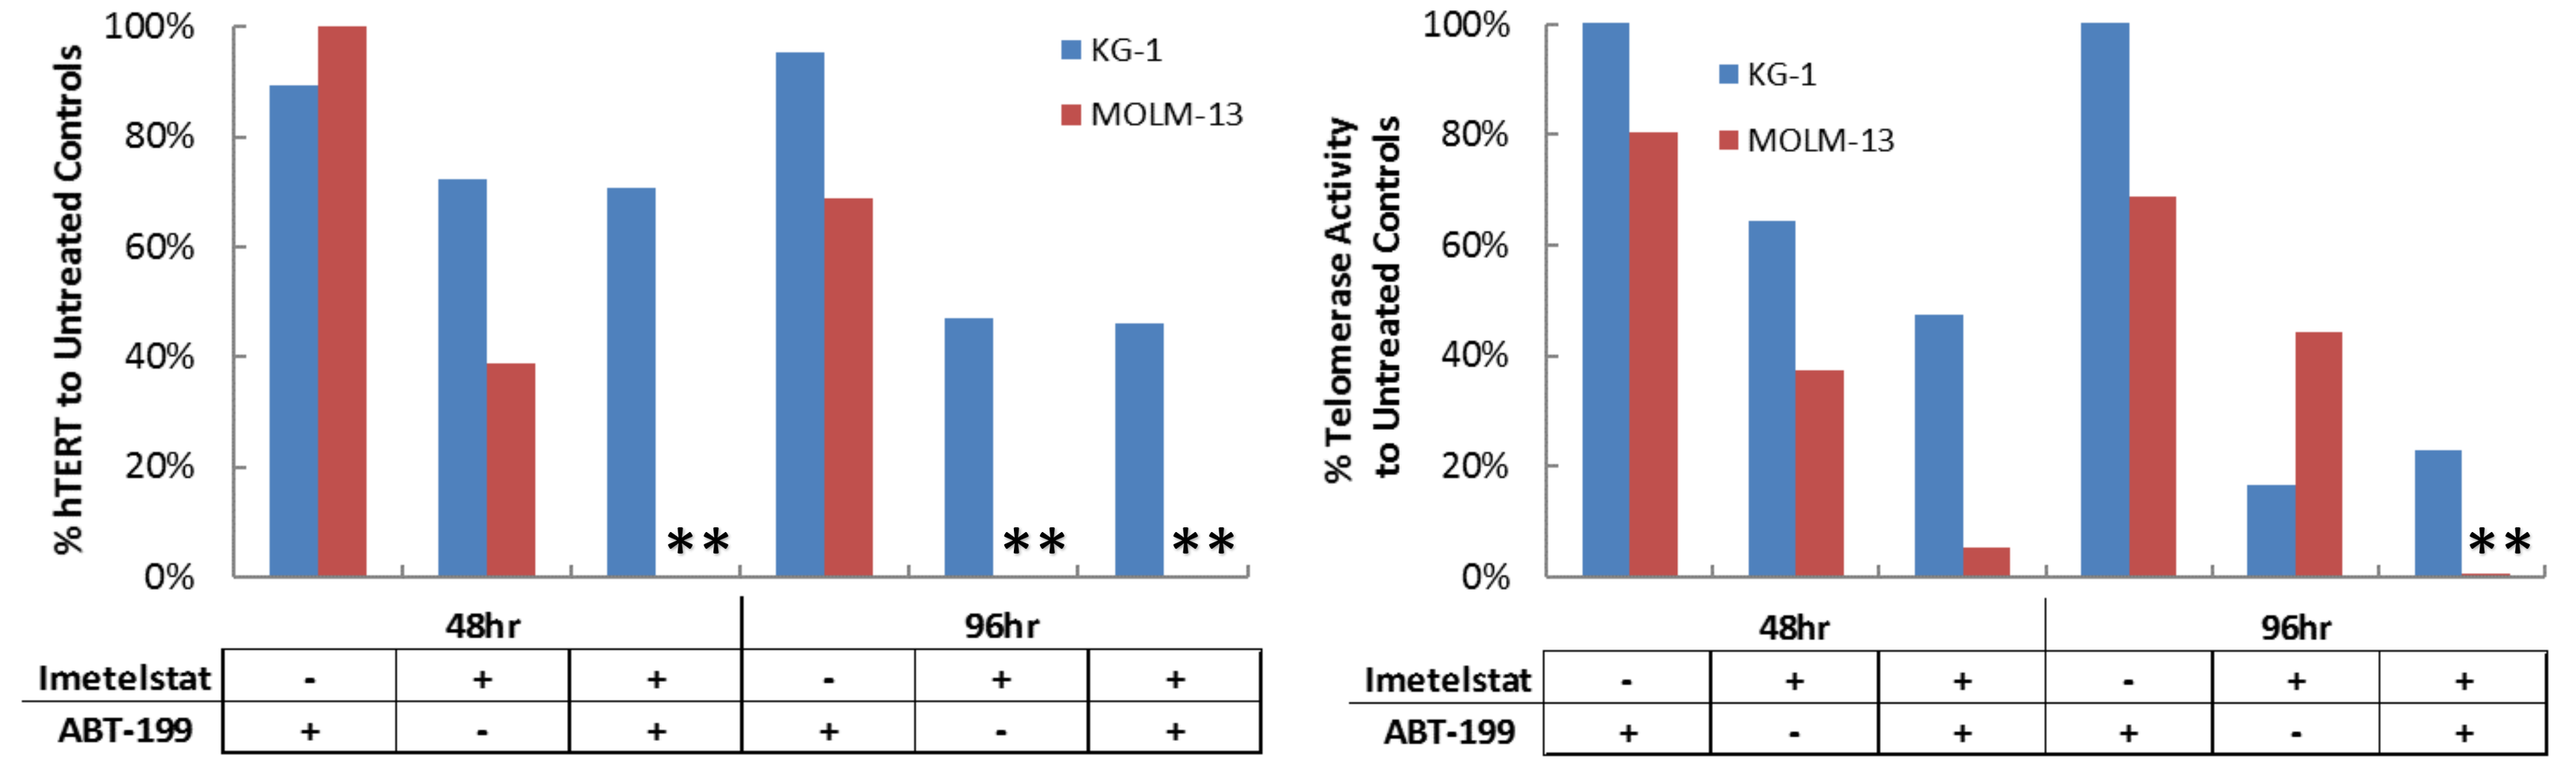
% hTERT to Untreated Controls
KG-1
MOLM-13
48hr
96hr
Imetelstat
ABT-199
% Telomerase Activity to Untreated Controls

Figure 4. hTERT expression and telomerase activity are modulated by the combination
hTERT expression (left) and telomerase activity (right) were greatly reduced by imetelstat, only minimally by ABT-199 *in vitro*, but both are further strongly reduced by combining both drugs. ** = values at or below assay limit of detection.

In vivo efficacy in mouse xenograft model

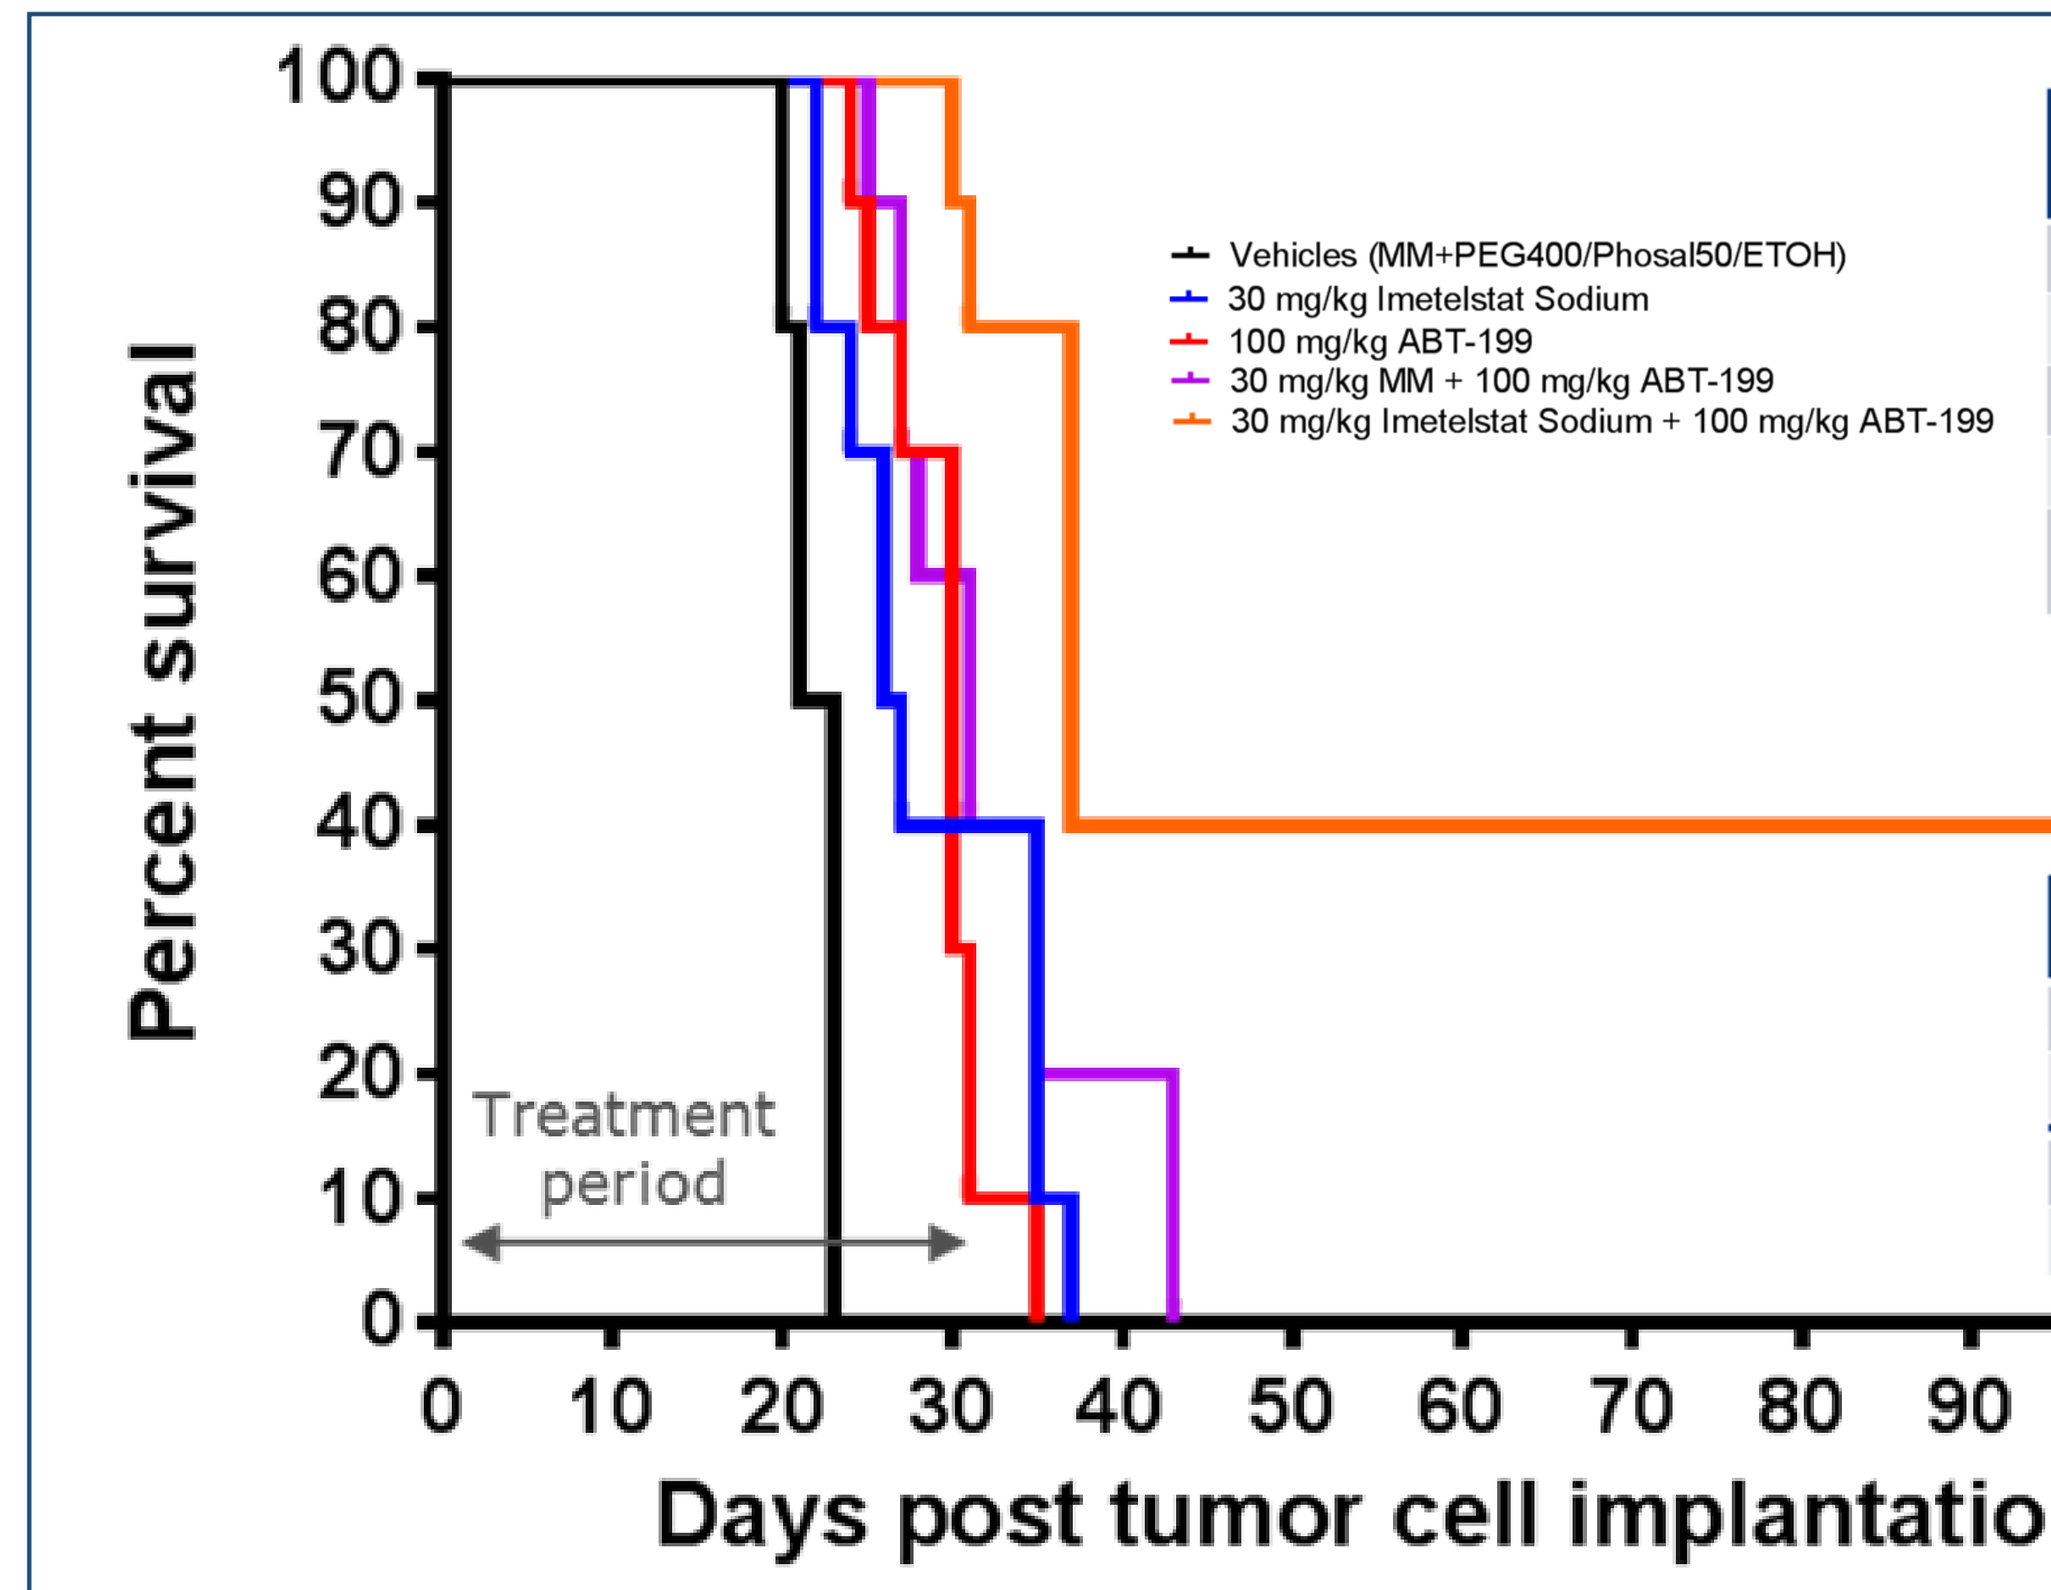
Percent survival
Days post tumor cell implantation
Treatment period
Vehicles (MM+PEG400/Phosal50/ETOH)
30 mg/kg Imetelstat Sodium
100 mg/kg ABT-199
30 mg/kg MM + 100 mg/kg ABT-199
30 mg/kg Imetelstat Sodium + 100 mg/kg ABT-199

Group	Median Survival (days)	%ILS	p*
Vehicles	22	---	---
Imetelstat sodium	26.5	20.4	0.0009
ABT-199	30	36.3	0.0001
MM + ABT-199	31	40.9	0.0001
Imetelstat sodium + ABT-199	37	68.1	0.0001

Group	Median Survival (days)	%ILS	p
Imetelstat	26.5	---	---
Imetelstat + ABT-199	37	39.6	0.0011
ABT-199	30	---	---
Imetelstat + ABT-199	37	23.3	0.0001

Figure 5. Combining drugs yields enhanced survival in MOLM-13 mouse model of AML
Imetelstat plus ABT-199 was well tolerated by SCID-beige mice – morbidity was observed for single agent and controls only due to tumor growth.
Survival curve and increased life span analysis show that the combination yields the greatest outcome, with 4 mice remaining alive 80 days after drug treatment ended.
MM = mismatch oligonucleotide control

Ex vivo proof of concept

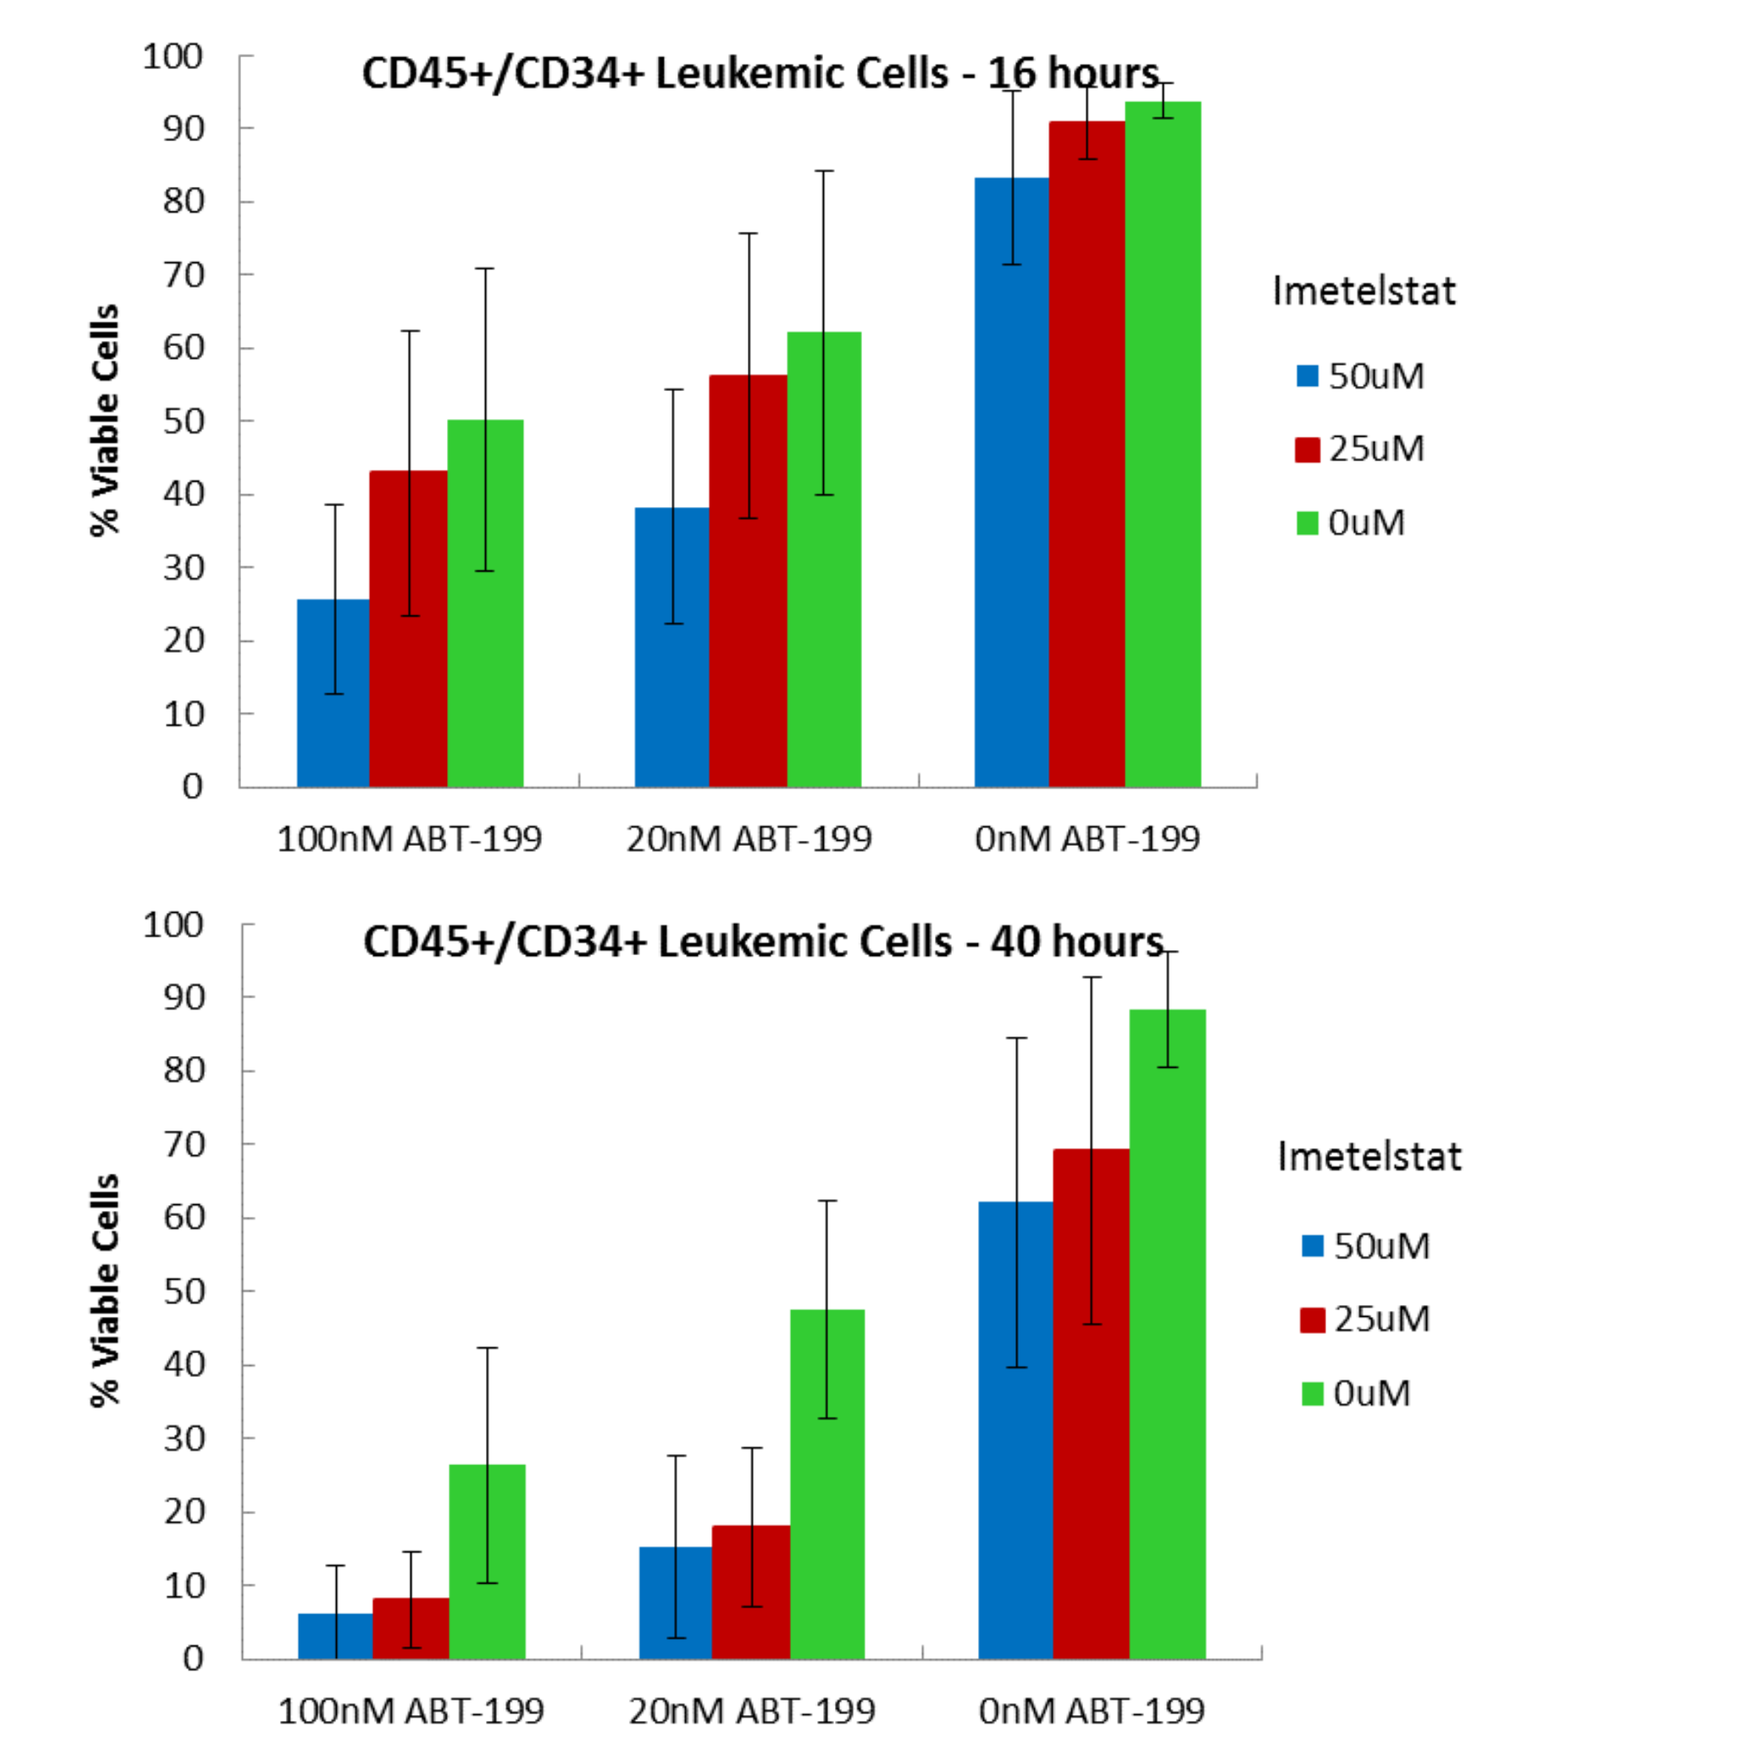
CD45+/CD34+ Leukemic Cells - 16 hours
CD45+/CD34+ Leukemic Cells - 40 hours
% Viable Cells
Imetelstat
50uM
25uM
0uM
100nM ABT-199
20nM ABT-199
0nM ABT-199

Figure 6. Imetelstat in combination with ABT-199 reduces the viability of AML leukemic cells *ex vivo*
Leukemic blasts from AML patients' PBMCs showed dose response to ABT-199 alone, and enhanced blast death was observed when combining with imetelstat. Mean ± standard deviation of four patient samples.

CONCLUSIONS

- Three of five investigated AML lines exhibited sensitivity to ABT-199; two lines showed moderate sensitivity to imetelstat
- Imetelstat enhanced apoptosis induced by ABT-199 in AML cell lines *in vitro* and AML patient samples *ex vivo;* with greater synergistic activity observed at longer treatment times
- Both hTERT expression and telomerase activity were greatly decreased by imetelstat treatment and further reduced by combination with ABT -199
- Prolonged survival was achieved *in vivo* when combining imetelstat with ABT-199; four mice remained alive ~80 days post end of treatment suggesting a potential cure in these animals

REFERENCES

[1]Li Y and Tergaonkar V. Canc Res 2014; 74: 1639
[2]Rongqing, P et al. Canc Res 2014; 4: 362
[3]Baerlocher G et al. NEJM 2015; 373: 920
[4]Tefferi A et al. NEJM 2015; 373: 908
[5] Tefferi A et al. Blood 2016; 6: e405
[6]Bellot G and Wang X. "Apoptosis", 2013
[7]http://cwr.horizondiscovery.com/analyze.jsp