Poster number: 4256

Efficacy and Safety of Imetelstat in RBC Transfusion-Dependent IPSS Low/Int-1 MDS Relapsed/Refractory to Erythropoiesis-Stimulating Agents (ESA) (IMerge)

Pierre Fenaux[1], Azra Raza[2], Edo Vellenga[3], Uwe Platzbecker[4], Valeria Santini[5], Irina Samarina[6], Koen Van Eygen[7], María Díez-Campelo[8], Mrinal M. Patnaik[9], Laurie Jill Sherman[10], Libo Sun[11], Helen Varsos[11], Esther Rose[11], Aleksandra Rizo[11], David P. Steensma[12]

[1]Hôpital St Louis, Paris, France; [2]Columbia Presbyterian, New York, NY, USA; [3]UMCG, Groningen, Netherlands; [4]Universitätsklinikum Carl Gustav Carus Dresden, Germany; [5]AOU Careggi, University of Florence, Italy; [6]Emergency Hospital of Dzerzhinsk, Nizhny Novgorod, Russia; [7]AZ Groeninge – Oncology Centre, Kortrijk, Belgium; [8]Hosp. Clinico Univ. De Salamanca, Spain; [9]Mayo Clinic Rochester, Rochester, MN, USA; [10]Janssen Research & Development, LLC, Spring House, PA, USA; [11]Janssen Research & Development, LLC, Raritan, NJ, USA; [12]Dana-Farber Cancer Institute, Boston, MA, USA

BACKGROUND

- MDS: characterized by clonal myeloproliferation arising from malignant progenitor cell clones that have shorter telomeres and multiple clonal genetic abnormalities
- Telomerase activity (TA) and expression of human telomerase reverse transcriptase (hTERT): significantly increased in MDS and believed to play a critical role in dysregulated cell growth, enabling continued and uncontrolled proliferation of malignant progenitor cell clones
 - o Higher TA and hTERT and shorter telomere length: poor prognostic features for patients with low-risk MDS, leading to shorter overall survival
- Limited treatment options for anemia in lower-risk MDS that has relapsed after or is refractory to ESA therapy
- Imetelstat: novel, first-in-class telomerase inhibitor that targets cells with short telomere lengths and active telomerase (**Figure 1**) and has clinical activity in myeloid malignancies.[1-3] Targeting MDS clones with imetelstat has potential to improve outcomes, including anemia, in MDS relapsed/refractory to ESA therapy
- IMerge: ongoing 2-part, global, phase 2/3 study of imetelstat in red blood cell (RBC) transfusion-dependent (TD), ESA-relapsed/refractory lower risk MDS. Part 1 consists of an open-label, single-arm design with imetelstat monotherapy
 - o Here we report safety and efficacy findings from 32 patients enrolled in Part 1
 - o Subgroup analysis of patients naïve to lenalidomide and hypomethylating agent (HMA) treatment and without del(5q) are also presented, as current results suggest improved efficacy among these patients

Figure 1. Imetelstat binds to RNA template, preventing maintenance of telomeres

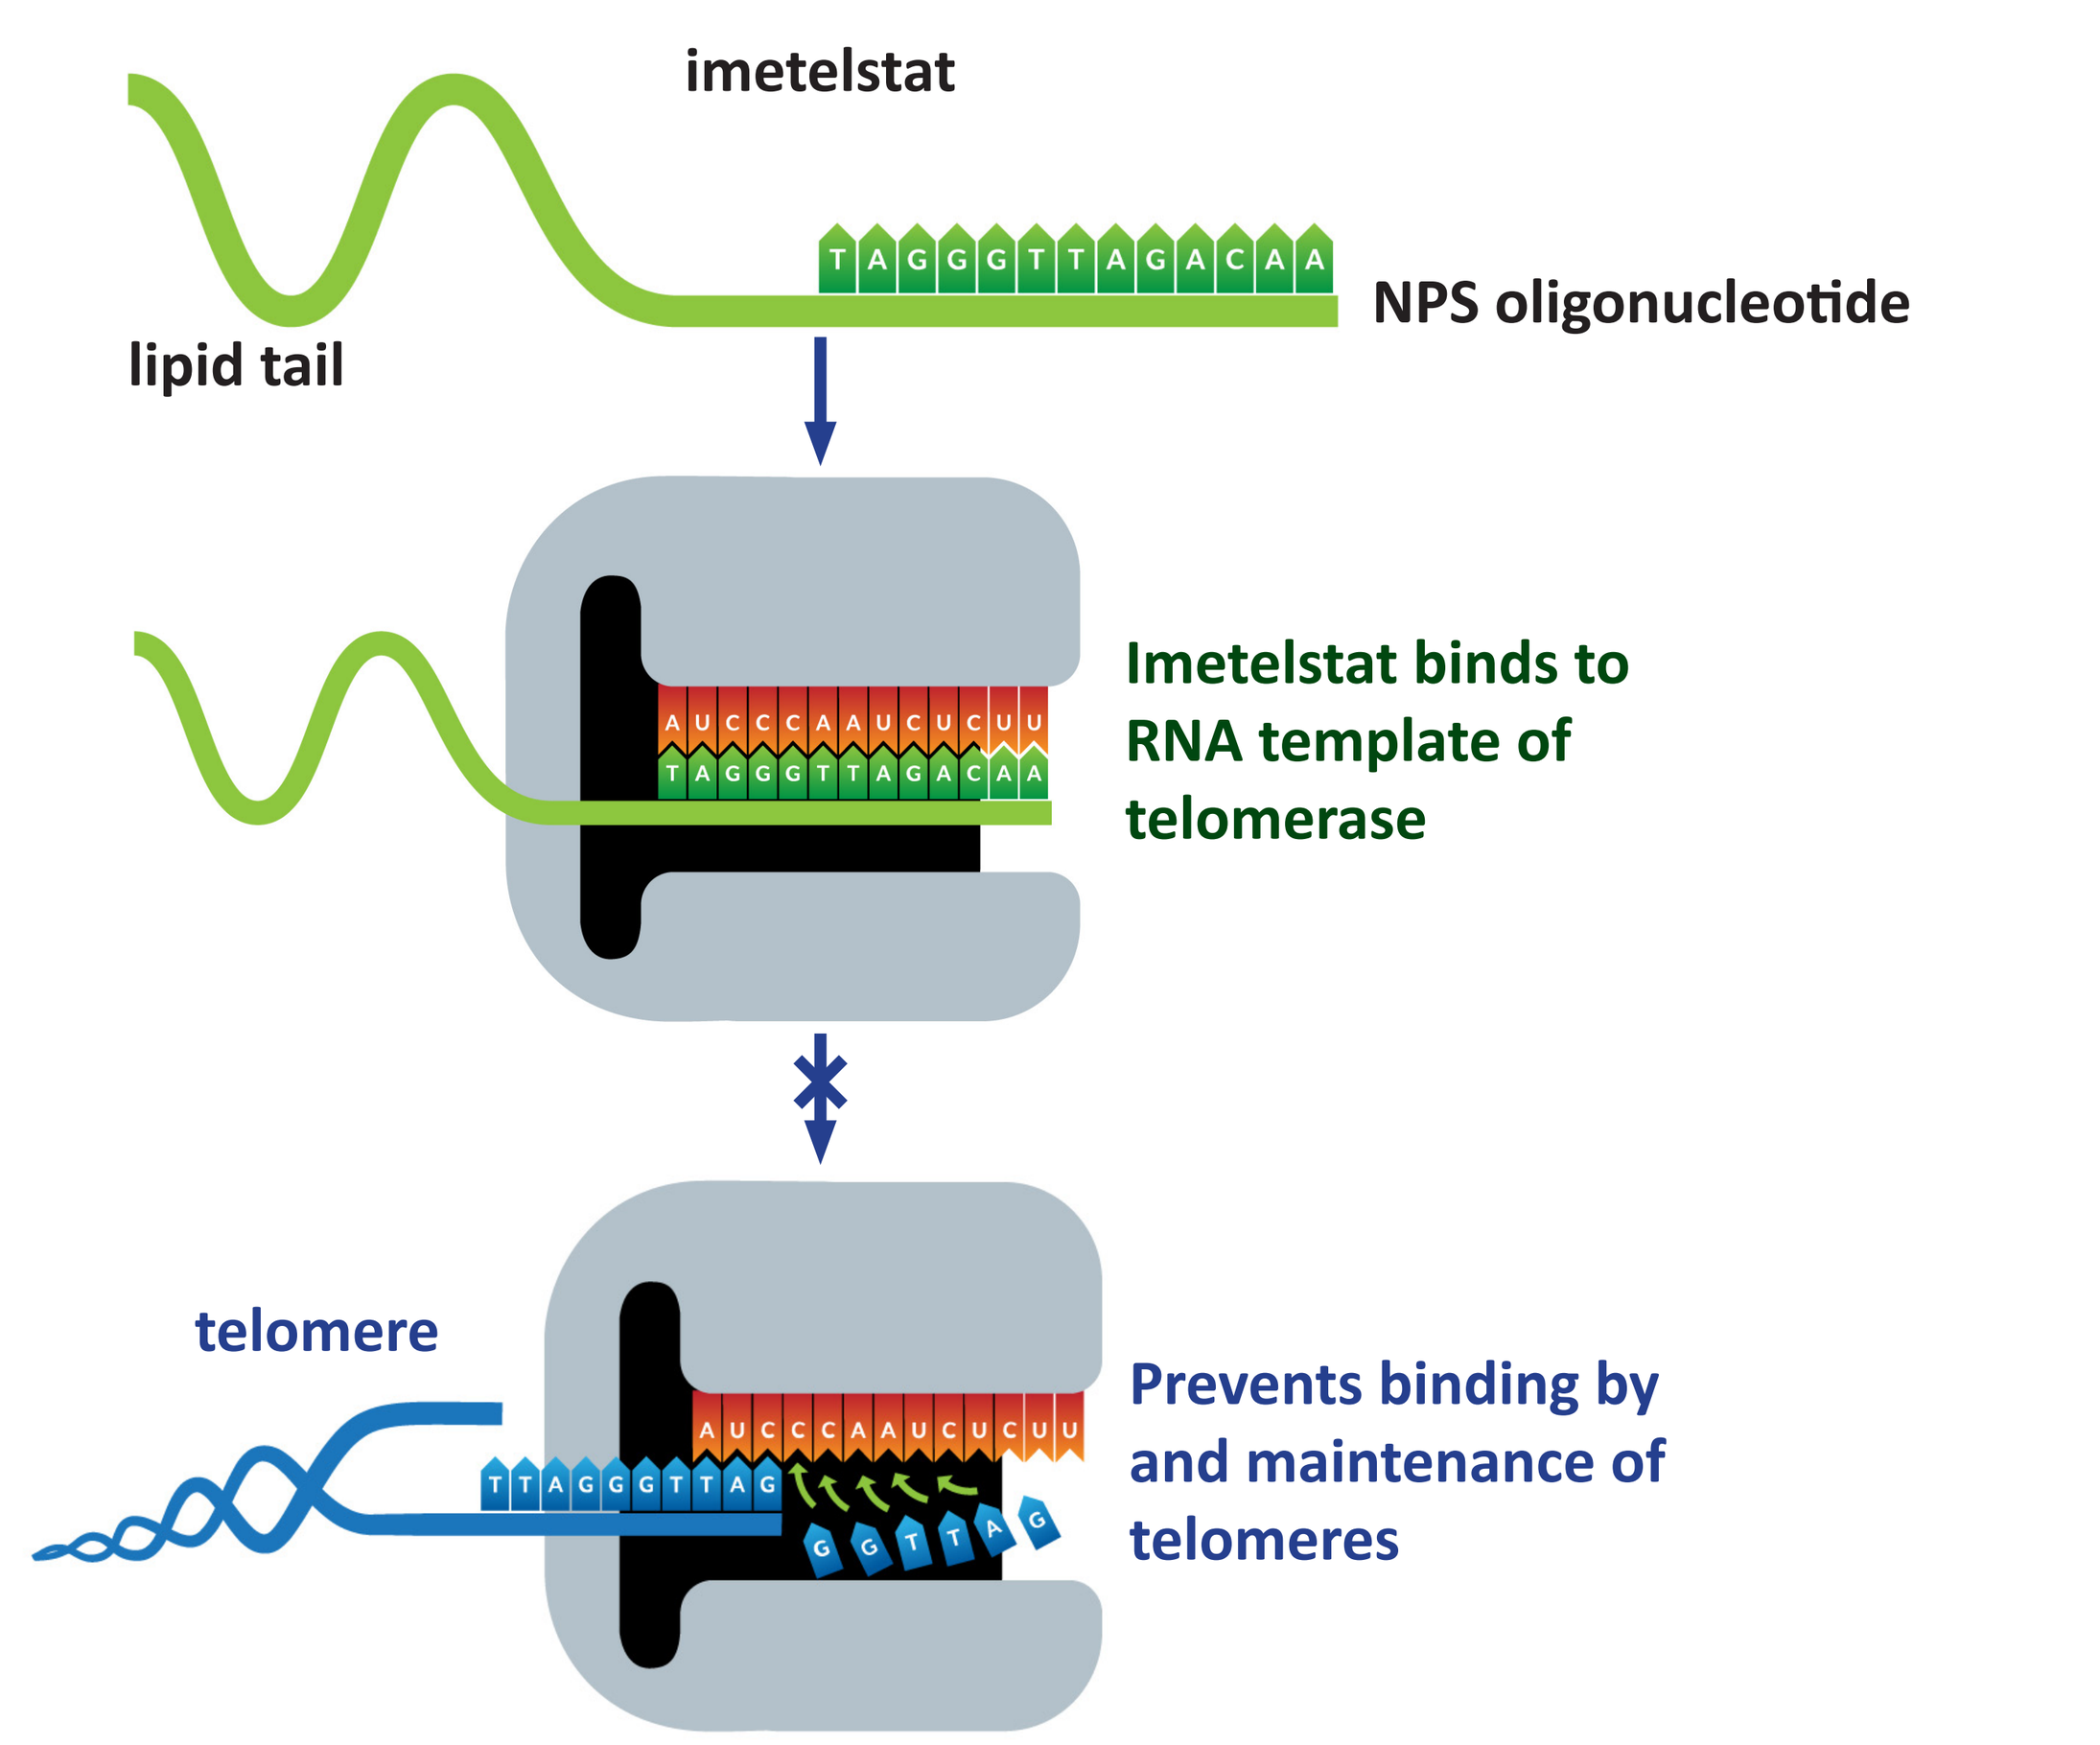

imetelstat
lipid tail
TAGGGTTAGACAA
NPS oligonucleotide
AUCCCAAUCUCUU
TAGGGTTAGACAA
Imetelstat binds to RNA template of telomerase
telomere
TTAGGGTTAG
GGTTAG
Prevents binding by and maintenance of telomeres

METHODS

Eligibility

- Adults diagnosed with MDS; IPSS Low or Int-1
- TD, defined as a RBC transfusion requirement of ≥4 units over 8 weeks prior to study entry
- ESA relapsed or refractory following at least 8 weeks of weekly epoetin alfa 40,000 U or darbepoetin alfa 150 mcg (or equivalent) or serum erythropoietin (sEPO) >500 mU/mL
- Any prior therapy (including lenalidomide or HMAs) allowed
 - o Patients with the del(5q) karyotype were allowed to enter irrespective of prior treatment
- ECOG 0-2
- ANC ≥1.5 x 10^9/L and platelets ≥75 x 10^9/L independent of growth factor or transfusion support
- AST, ALT and ALP ≤2.5 times the upper limit of normal (x ULN), total bilirubin ≤3 x ULN and direct bilirubin ≤2 x ULN (unless due to Gilbert's syndrome)

Treatment

- Imetelstat administered as a 2-hour IV infusion every 4 weeks at a starting dose of 7.5 mg/kg, following premedication with an antihistamine and corticosteroid. Dose escalation to 9.4 mg/kg permitted for insufficient response after at least 3 cycles at the initial dose, provided that ANC and platelet nadirs had not dropped below 1.5 x 10^9/L and 75 x 10^9/L, respectively, and no grade ≥3 non-hematological toxicity
- Supportive care, including transfusion and myeloid growth factors as clinically indicated, permitted

Endpoints and Analysis

- Primary endpoint: rate of RBC transfusion-independence (TI) lasting ≥8 weeks
- Key secondary endpoints:
 - o Safety
 - o Rate of ≥24-week TI
 - o Time to and duration of TI
 - o Hematologic improvement (HI) rate
 - o Rate of complete response (CR) and partial response (PR) per IWG

RESULTS

Patients

- Baseline median RBC transfusion burden, 6 units/8 weeks (range: 4–14)

Table 1. Baseline characteristics (N=32)

Median age (range), y	68.5 (46-83)
Male, n (%)	16 (50)
ECOG PS 0-1, n (%)	29 (91)
IPSS risk, n (%)	
Low	19 (59)
Intermediate-1	13 (41)
Karyotype, n (%)	
Normal	17 (53)
Any abnormality	11 (34)
del(5q)	7 (22)
Unknown (missing or no growth)	4 (13)
WHO category, n (%)	
RARS/RCMD-RS	16 (50)
All others	16 (50)
sEPO >500 mU/mL, n (%)	13* (43)
Prior ESA, n (%)	28 (88)
Prior lenalidomide, n (%)	12 (38)
Prior decitabine or azacitidine, n (%)	8 (25)
Naïve to lenalidomide and HMA and non-del(5q), n (%)	13 (41)

*Of 30 patients with sEPO levels reported.

Exposure

- Median follow-up for this analysis: 66.1 weeks
- Median number of treatment cycles: 6.5 (range: 1–20 cycles)
- Sixteen patients (50%) had dose reductions and 19 patients (59%) had cycle delays due to adverse events
- Seven patients had imetelstat dose escalation to 9.4 mg/kg

Efficacy

Table 2. Key Efficacy Outcomes

	All Treated (N=32)	Lenalidomide and HMA naïve and Non-del (5q) (n=13)
Rate of ≥8-week TI, n (%)	12* (38)	7 (54)
Mean relative reduction from baseline transfusion burden, %	-64	-71
Rate of ≥24-week TI, n (%)	5 (16)	4 (31)
Median time to onset of TI, weeks	8.1	8.3
Median duration of TI, weeks	23.1	42.9
Erythroid HI rate, n (%)	20† (63)	9† (69)
CR + mCR + PR (per IWG), n (%)	4 (13)	3 (23)

*Results are based on the Oct 16th 2017 data snapshot, at which one ≥8-week TI had not been fully confirmed based on communication with investigator; †Includes patients with a transfusion reduction of ≥4 units during the best 8-week on-study interval, as well as those with a hemoglobin increase from pretreatment level.

- The primary endpoint of RBC TI lasting ≥8-weeks achieved in 12/32 (38%) patients
- 5/32 (16%) achieved 24-week TI (**Figure 2A, 2B and Figure 3**)
 - o These patients also achieved sustained hemoglobin increases by at least 1.5 g/dL over 8 weeks (HI-E Hb)
 - o Their median duration of TI (65.1 weeks) now exceeds one year
- 20/32 patients (63%) had an erythroid HI (**Figure 2A and Figure 2B**)
- In the subset of patients who were naïve to lenalidomide and HMAs and who lacked del(5q), 8-week and 24-week TI rates were 54% and 31%, respectively (higher than in the overall population) and the erythroid HI rate was 69% (similar to that reported in the overall population)
- CR and marrow CR (mCR) were each reported for 2 patients and there were no PRs, for a CR+PR+mCR rate of 13%
 - o One CR and both mCR were in the subset of patients who were naïve to lenalidomide and HMAs and who lacked del(5q)
- 8-week TI did not differ based on the presence of ringed sideroblasts (RS):
 - o 38% (6/16) for RS+ and 38% (6/16) for RS-
- Response appeared to be independent of sEPO level; of 30 patients with baseline sEPO level reported:
 - o 41% (7/17) with sEPO level ≤500 mU/L achieved ≥8-week TI
 - o 38% (5/13) with sEPO level >500 mU/L achieved ≥8-week TI

Figure 2. Waterfall plot of A) the longest transfusion-free interval and B) absolute change in transfusion amount in the best 8-week interval.

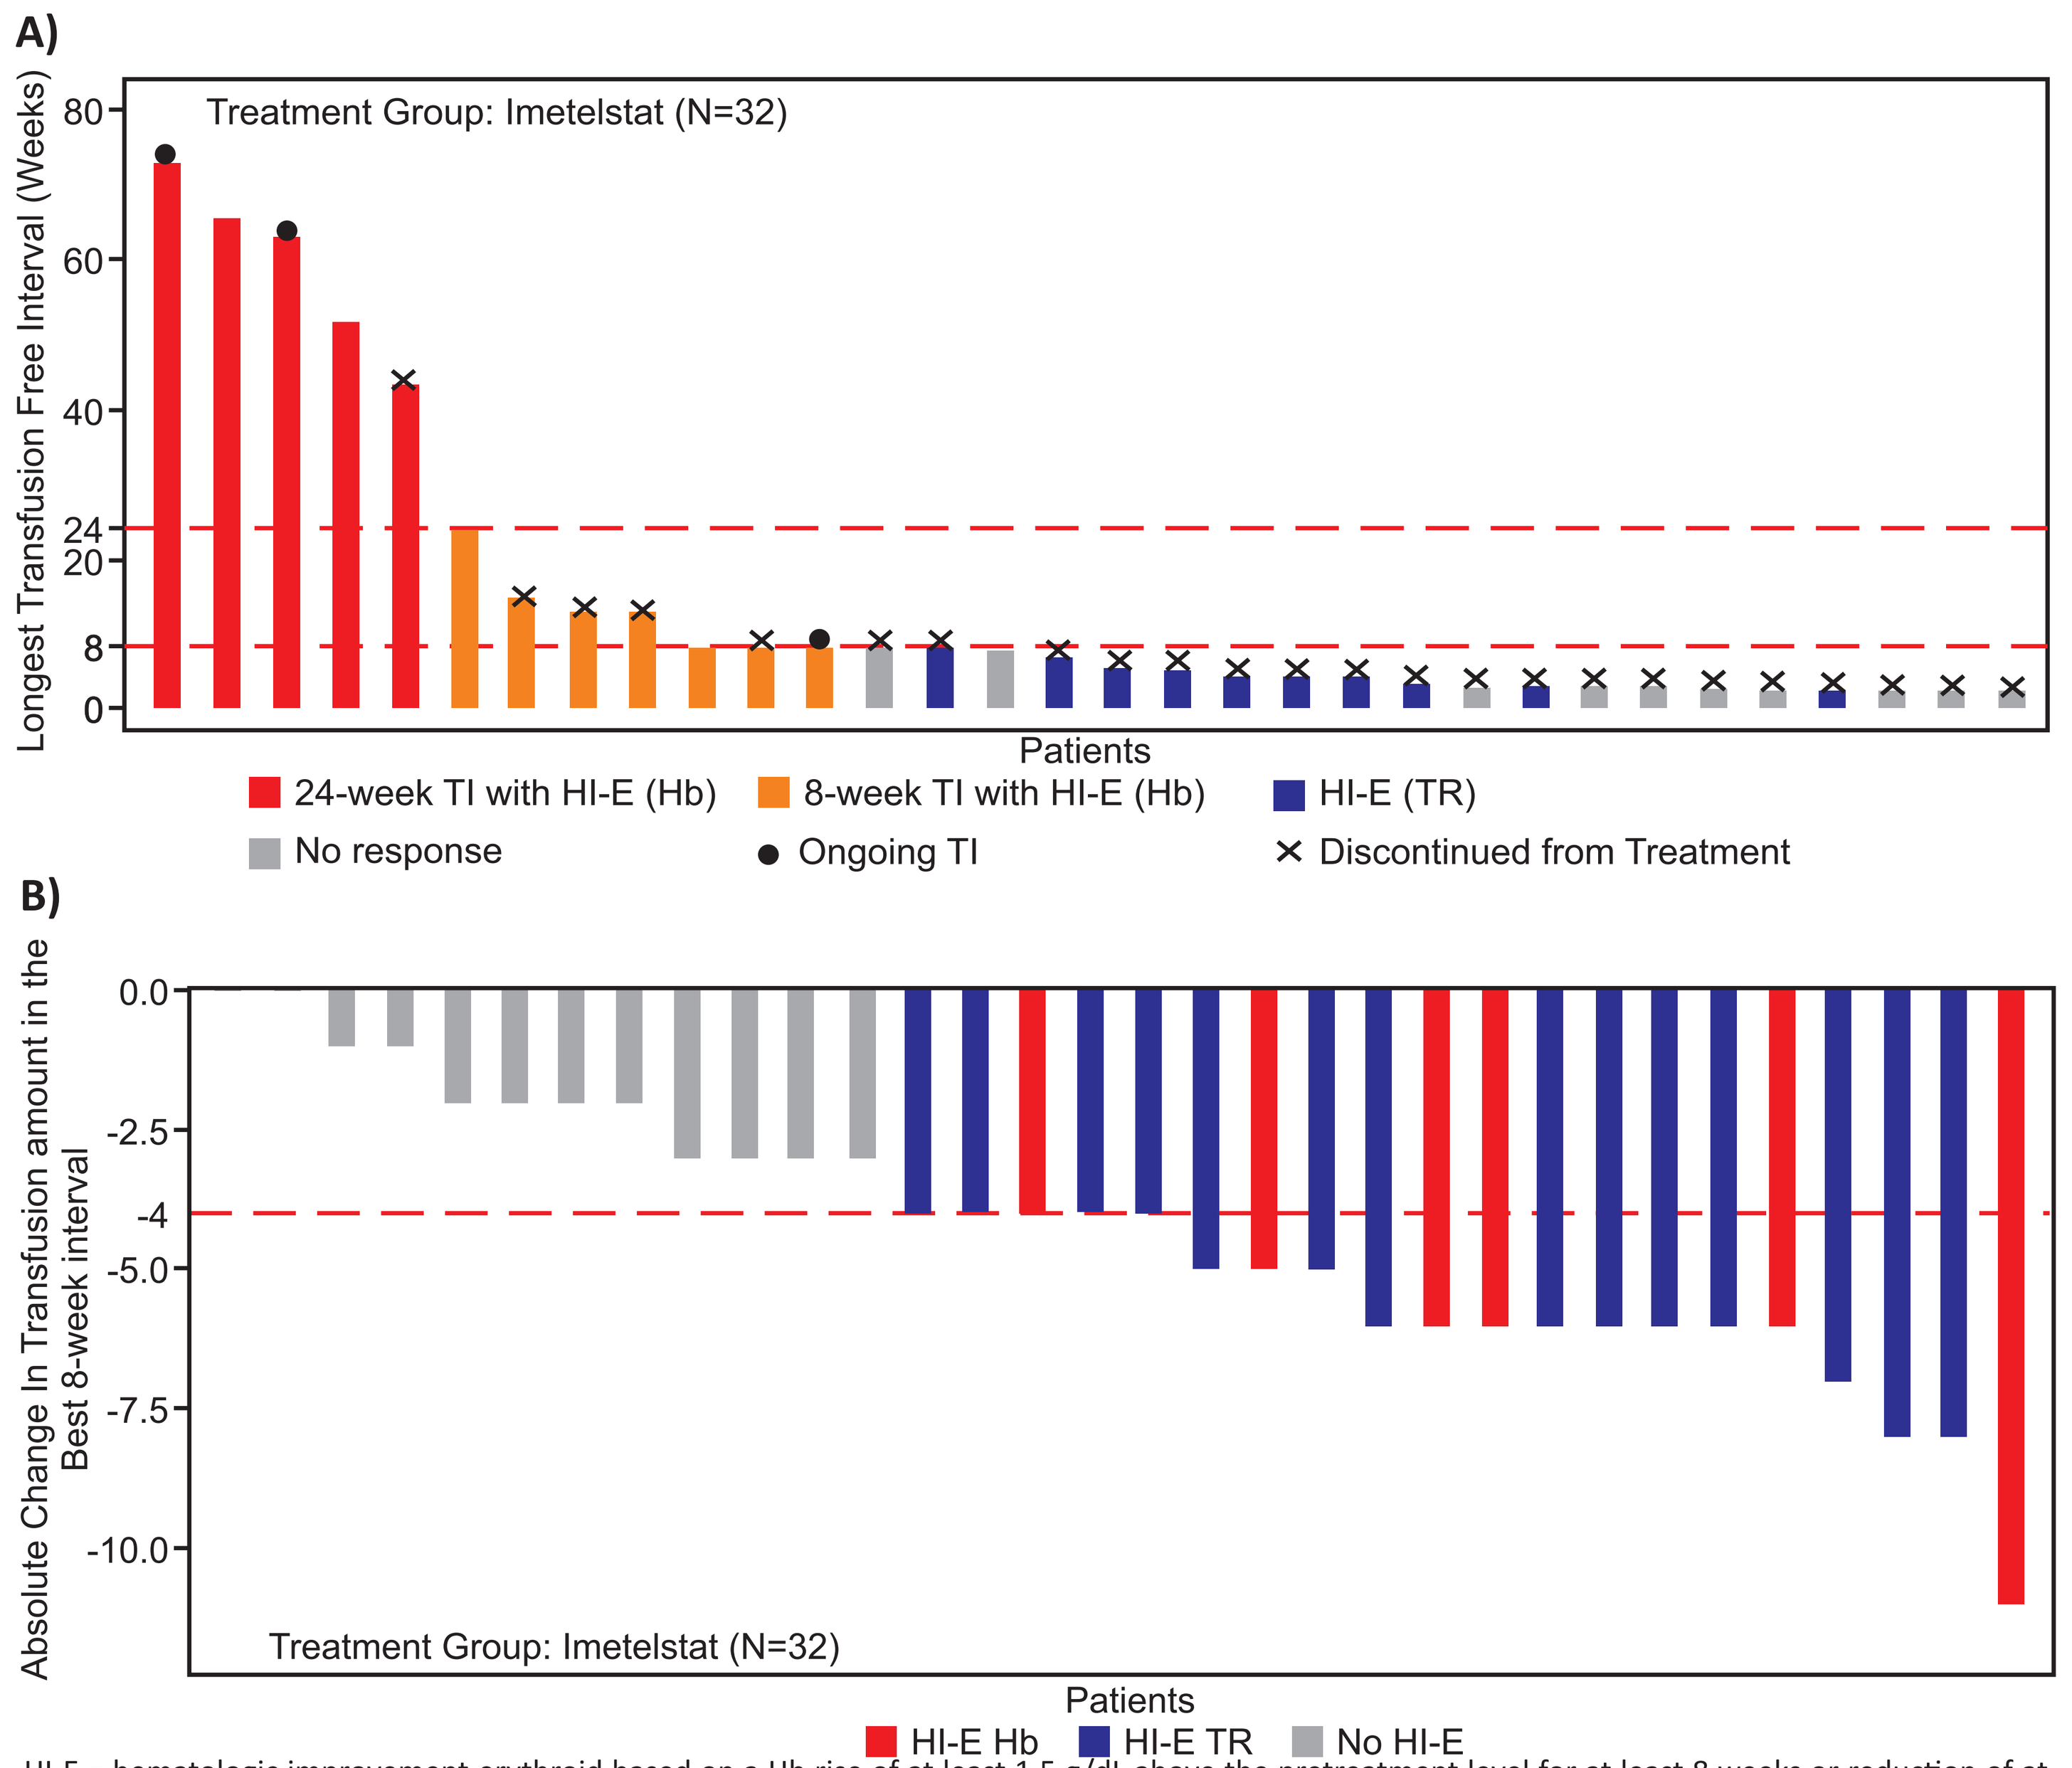

A)
Treatment Group: Imetelstat (N=32)
Longest Transfusion Free Interval (Weeks)
80
60
40
24
20
8
0
Patients
24-week TI with HI-E (Hb)
8-week TI with HI-E (Hb)
HI-E (TR)
No response
Ongoing TI
Discontinued from Treatment
B)
Absolute Change In Transfusion amount in the Best 8-week interval
0.0
-2.5
-4
-5.0
-7.5
-10.0
Treatment Group: Imetelstat (N=32)
Patients
HI-E Hb
HI-E TR
No HI-E

HI-E = hematologic improvement-erythroid based on a Hb rise of at least 1.5 g/dL above the pretreatment level for at least 8 weeks or reduction of at least 4 units of RBC transfusions/8 weeks compared with the prior RBC transfusion burden (criterion adapted from IWG 2006); HI-E Hb = HI-E with sustained rise in hemoglobin by at least 1.5 g/dL over 8 weeks; TI = transfusion independence; TR = transfusion reduction by at least 4 units over 8 weeks.

Figure 3. Example of a 24-week TI responder.

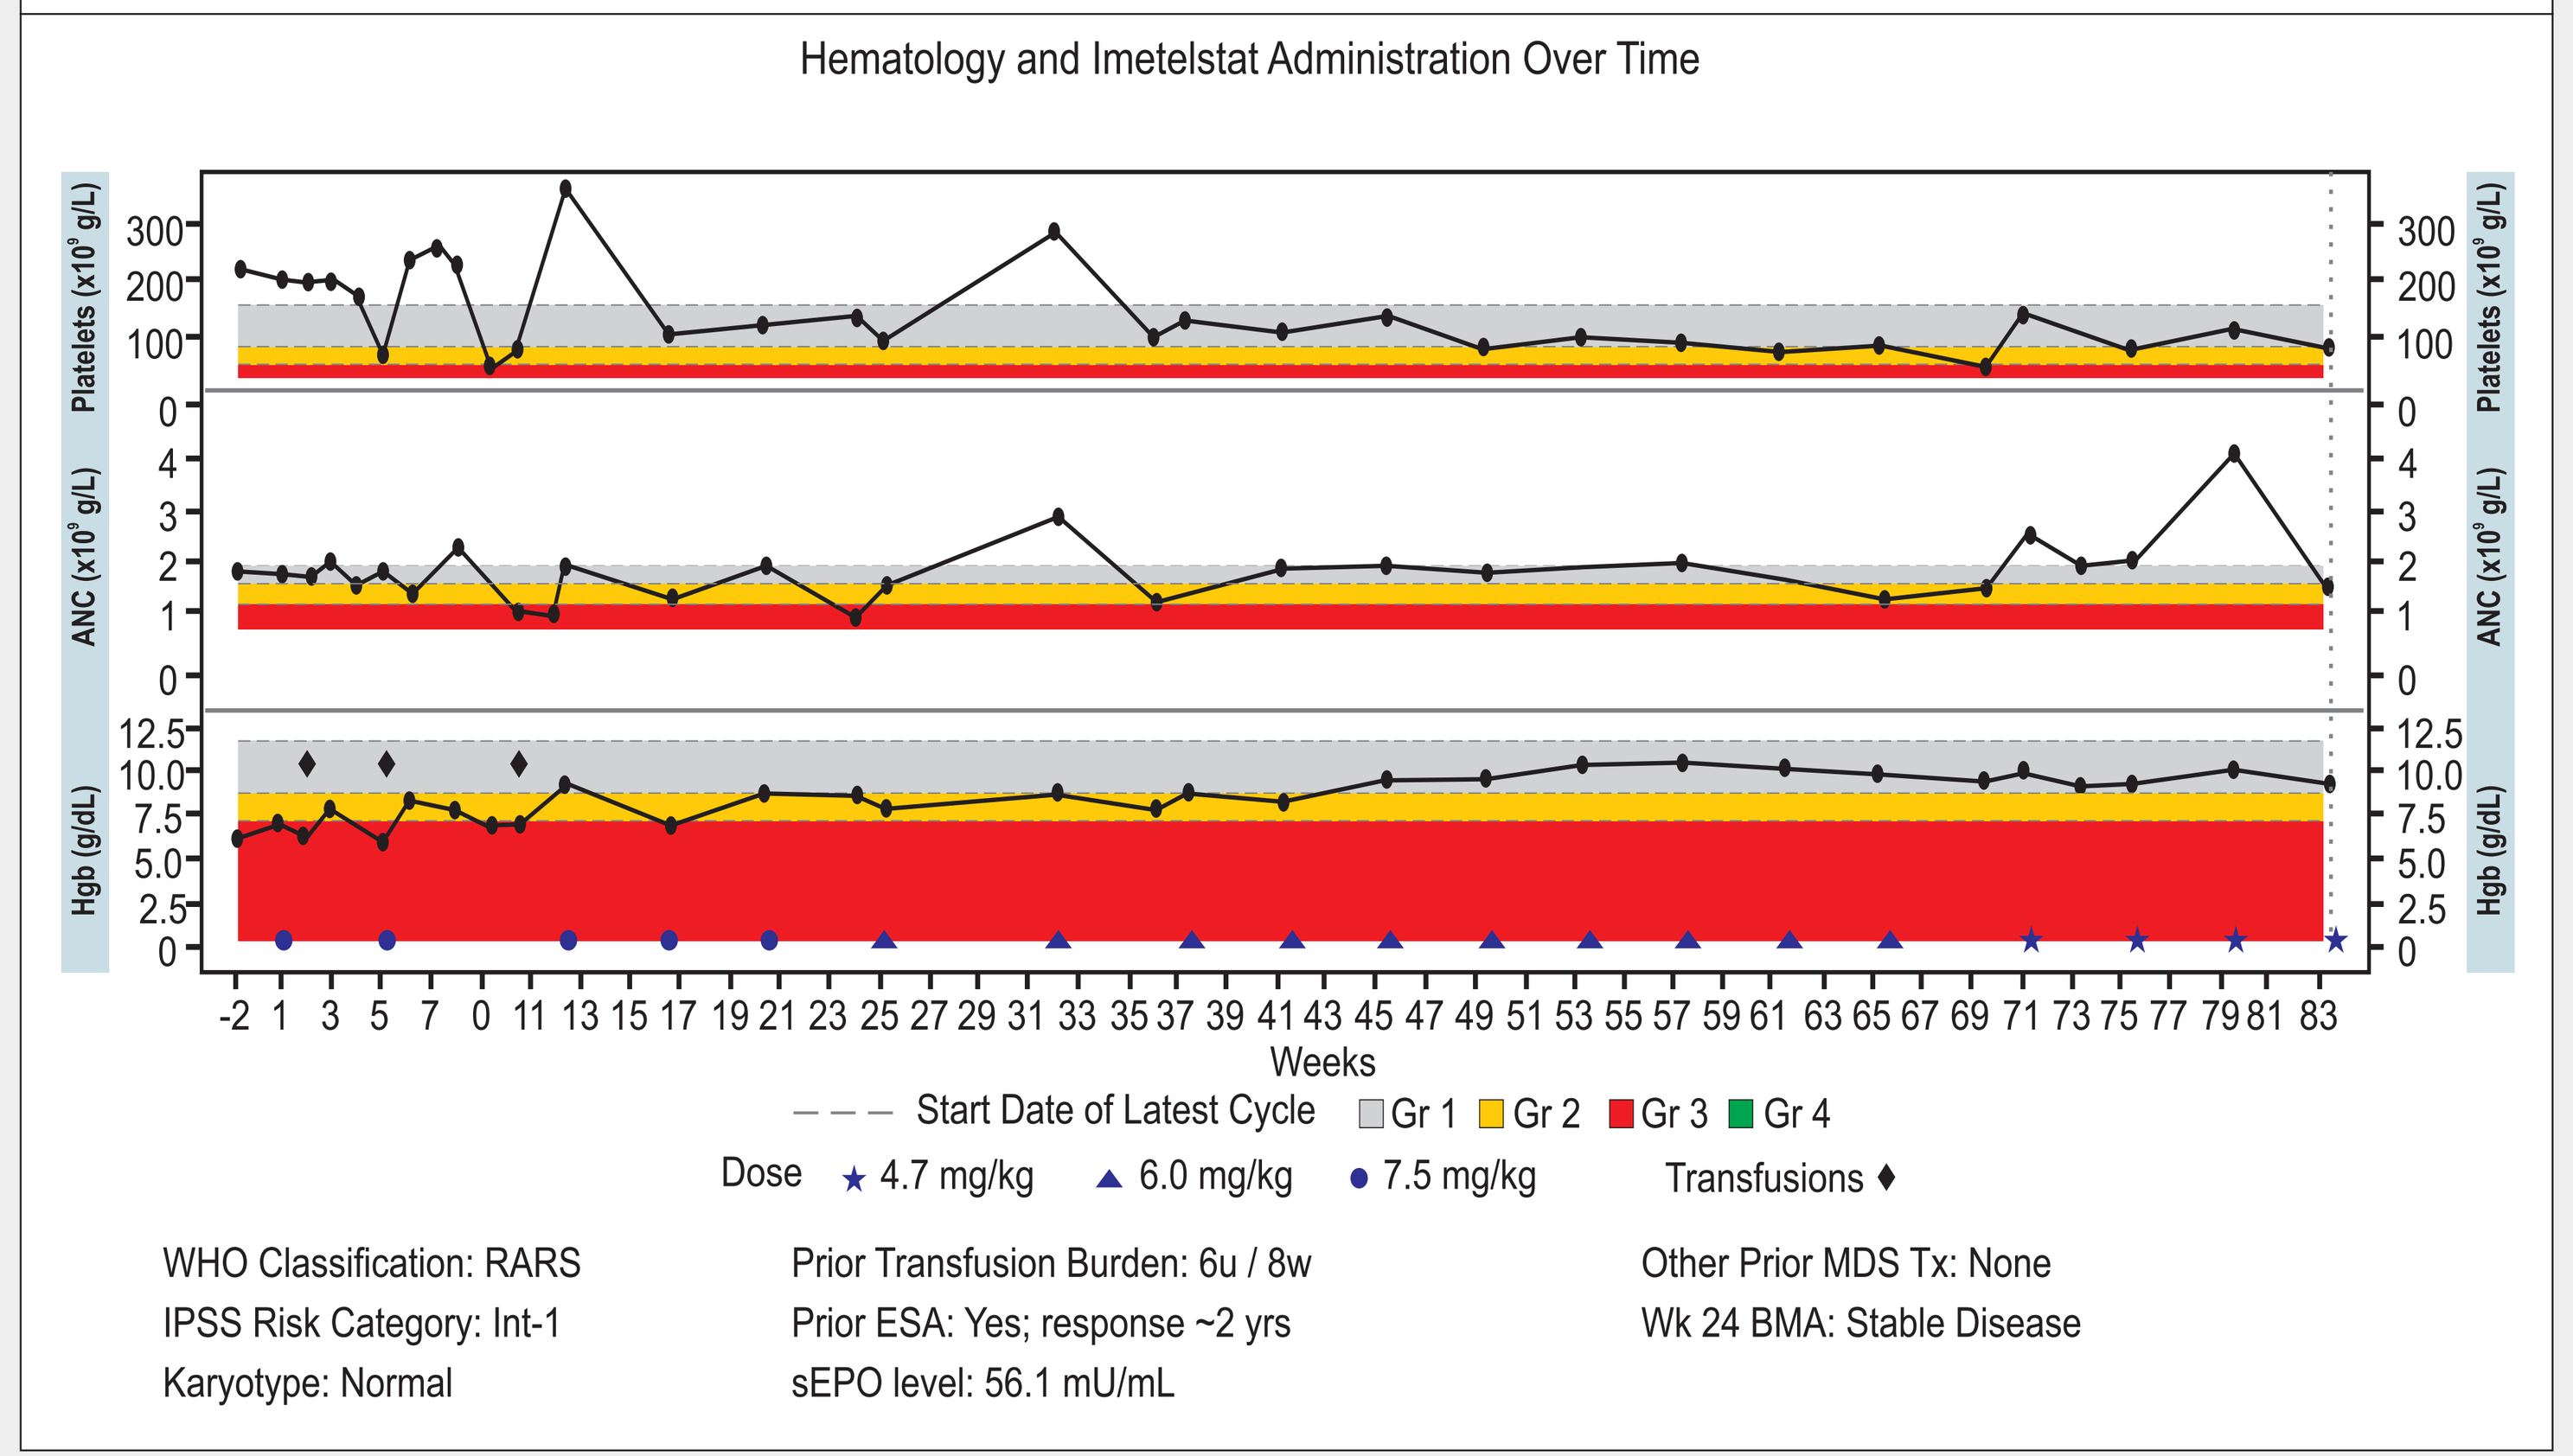

Hematology and Imetelstat Administration Over Time
Platelets (x10⁹ g/L)
ANC (x10⁹ g/L)
Hgb (g/dL)
Weeks
Start Date of Latest Cycle
Gr 1
Gr 2
Gr 3
Gr 4
Dose
4.7 mg/kg
6.0 mg/kg
7.5 mg/kg
Transfusions
WHO Classification: RARS
IPSS Risk Category: Int-1
Karyotype: Normal
Prior Transfusion Burden: 6u / 8w
Prior ESA: Yes; response ~2 yrs
sEPO level: 56.1 mU/mL
Other Prior MDS Tx: None
Wk 24 BMA: Stable Disease

Safety

- Cytopenias, particularly neutropenia and thrombocytopenia, were the most frequently reported adverse events overall and in the subset who were naïve to lenalidomide and HMAs and lacked del(5q) (**Table 3**)
- The subset had a lower incidence of grade ≥3 neutropenia relative to the overall population but similar grade ≥3 thrombocytopenia (**Table 4**)
 - o In most cases, grade ≥3 cytopenias were reversible within 4 weeks without clinical sequelae, and patients were able to continue imetelstat treatment after dose modification
- 1 patient (of 22 with neutropenia) experienced neutropenic fever and 2 patients (of 18 with thrombocytopenia) had grade 3 thrombocytopenia concurrent with grade 1 bleeding events that were both considered related to imetelstat; these events recovered without sequelae
- 28 patients (88%) had liver function test (LFT) elevations by at least one grade
 - o These events were generally grade 1 or 2 and reversible
 - o Four patients (including 3 in the subset of patients who were naïve to lenalidomide and HMAs and who lacked del[5q]) had grade 3 worsening of AST and/or ALT, and 1 of these patients had grade 3 worsening of bilirubin; all of which were reversible

Table 3. Most Common Treatment-Emergent Adverse Events (≥10% of Patients in All Treated Patients)

	All Treated (N=32)	Lenalidomide and HMA naïve and Non-del(5q) (n=13)
Patients with ≥1 treatment-emergent AEs, n (%)	31 (97)	12 (92)
Neutropenia	22 (69)	7 (54)
Thrombocytopenia	18 (56)	8 (62)
Headache	8 (25)	2 (15)
ALT increased	6 (19)	3 (23)
AST increased	5 (16)	3 (23)
Leukopenia	5 (16)	2 (15)
Muscle spasms	5 (16)	2 (15)
Anemia	4 (13)	2 (15)
Asthenia	4 (13)	4 (31)
Constipation	4 (13)	2 (15)
Cough	4 (13)	1 (8)
Diarrhea	4 (13)	1 (8)
Dyspnea	4 (13)	2 (15)
Influenza like illness	4 (13)	1 (8)
Nausea	4 (13)	2 (15)
Peripheral edema	4 (13)	2 (15)
Viral URI	4 (13)	4 (31)

Table 4. Maximum Grade Change in Cytopenias From Baseline

	All Treated (N=32)	Lenalidomide and HMA naïve and Non-del(5q) (n=13)
Neutrophils, n (%)		
No worsening	4 (13)	3 (23)
1	3 (9)	1 (8)
2	4 (13)	2 (15)
3	8 (25)	2 (15)
4	13 (41)	5 (38)
Platelets, n (%)		
No worsening	7 (22)	3 (23)
1	2 (6)	1 (8)
2	7 (22)	2 (15)
3	10 (31)	5 (38)
4	6 (19)	2 (15)

CONCLUSIONS

- Safety and efficacy data for the 32 patients in Part 1 of Study MDS3001 support continued investigation of imetelstat using the current dosing regimen of 7.5 mg/kg every 4 weeks
- Adverse events with imetelstat (mostly cytopenias) predictable, manageable, and reversible
- 8-week RBC TI in 38% and erythroid HI in 63% of IPSS Low/Int-1 RBC transfusion dependent MDS patients relapsed/refractory to ESA
- Durable 24-week TI, with sustained rises in Hb, in 16% of patients
- 54% RBC TI in the 13 patients without del(5q) and without prior exposure to either lenalidomide or HMA (compared to 38% in overall population) and responses were more durable (24-week TI rate of 31%)
 - o Based on higher response rates observed in this subgroup, the protocol has been amended to include only patients who meet these criteria
 - o The study is ongoing and actively recruiting (Protocol number: 63935937MDS3001; ClinicalTrials.gov: NCT02598661)

Reference:
1. Baerlocher GM, et al. N Engl J Med 2015;373:920-928; 2. Tefferi A, et al. N Engl J Med 2015;373:908-919; 3. Tefferi A, et al. Blood Cancer J 2016;6:e405.

Acknowledgements: Dr. Laurie Orloski (independent medical writer) provided writing assistance and Dr. Harry Ma (Janssen Research & Development, LLC.) provided additional editorial support for this poster. The authors thank all the patients for their participation in this study and acknowledge the collaboration and commitment of all investigators and their staff.

Disclosures: P. Fenaux–honoraria and research funding from Amgen, Astex, Celgene, Janssen, and Novartis; A. Raza–speakers bureaus for Novartis, Genoptix, and Onconova and research funding from Kura Oncology, Janssen, Celgene, Syros, and Onconova; E. Vellenga–no financial relationships to disclose; U. Platzbecker–consultancy, honoraria, and research funding from Celgene, Janssen, and Acceleron and consultancy and research funding from Novartis; V. Santini–honoraria from Celgene, Janssen, and Novartis, research funding from Celgene, consultancy for Janssen, Abbvie, and Otsuka, and advisory committees for Abbvie and Amgen; I. Samarina–research funding from Janssen; K. Van Eygen–consultancy, honoraria, and research funding from Janssen; M. Díez-Campelo–consultancy for Celgene and Novartis and research funding from Celgene, Novartis, and Janssen-Cilag; L. Sherman, L. Sun, H. Varsos, E. Rose, A. Rizzo–employment and equity ownership in Janssen; D. Steensma–consultancy for Janssen, Pfizer, Onconova, H3 Biosciences, Takeda, Celgene, Amgen, Pfizer, and Novartis, equity ownership in Incyte, research funding from Janssen, and advisory committees for Amgen, Pfizer, and Novartis.

This study was funded by Janssen Research & Development LLC and Geron Corporation.

Poster Presented at the 59th American Society of Hematology (ASH) Annual Meeting & Exposition, December 9-12, 2017, Atlanta, GA