Characterization of Disease, Treatment Patterns, and Outcomes of Patients With Myelofibrosis: Analysis of Two United States Commercial Claims Databases

Maneesha Mehra,[1] Ravi Potluri,[2] Jianming He,[1] Grace Wang,[3] Suneel Mundle,[3] Jacqueline Bussolari[3]

[1]Janssen Global Services LLC, Raritan, NJ; [2]SmartAnalyst, Inc., New York, NY; [3]Janssen Research & Development, LLC, Raritan, NJ

INTRODUCTION

- Myelofibrosis (MF) is a myeloproliferative neoplasm with a heavy symptomatic burden that results in profound negative effects on quality of life.[1]
- Patients with MF may present with splenomegaly, constitutional symptoms, moderate to severe anemia, thrombocytopenia, or leukocytosis.[2]
- Presentation may be primary (PMF) disease arising de novo, secondary following transformation from polycythemia vera or essential thrombocythemia (post-PV/ET sMF), or secondary from diseases such as myelodysplastic syndrome, leukemia, or lymphoma (Other sMF).
- To date, allogeneic stem cell transplantation remains the only potentially curative treatment for MF; however, few patients are eligible for transplant.[1]
- Patients who are not stem cell transplant candidates often require treatment to manage their symptoms.[1]
- Ruxolitinib, an inhibitor of the JAK1/JAK2 tyrosine kinases, is the only approved treatment for patients with intermediate or high-risk PMF or post PV/ET sMF.[3]
- National Comprehensive Cancer Network (NCCN) and European Society of Medical Oncology (ESMO) guidelines both recommend ruxolitinib for low-risk, symptomatic MF[4,5]; alternative recommended treatment approaches include interferon alpha,[4] hydroxyurea,[5] or clinical trial enrollment.[4,5]
- Ruxolitinib or clinical trial enrollment is also recommended for patients with intermediate-2 or high-risk disease, with additional treatments such as erythropoietin-stimulating agents recommended for treating patients with MF-associated anemia.[4,5]
- However, limited information is available on the management of patients who fail or discontinue frontline ruxolitinib.[6]

OBJECTIVE

- The present analysis was conducted to characterize disease, treatment patterns, and outcomes in patients with MF using two US health insurance claims databases.

METHODS

- The Truven Health Analytics MarketScan® (Commercial Claims and Encounters and Truven Medicare) and Optum™ (Optum, Inc, Eden Prairie, MN, USA) integrated virtual electronic health records and claims databases were retrospectively analyzed to identify patients with MF diagnosed between 2006 and 2015.
- Patients aged ≥ 18 years with ≥ 90 days of medical history prior to diagnosis were included.
- Patients were categorized as having PMF, post-PV/ET sMF, or Other sMF based on earliest MF International Classification of Diseases, 9th revision diagnosis code (ICD9, **Table 1**).
- Demographic characteristics, constitutional symptoms (identified by diagnosis code, eg, splenomegaly ICD9 789.2), platelet counts (in a small subset of patients, n = 112), and treatment patterns were summarized.
- Best supportive care may have included erythropoietin ± steroids (danazol, fluoxymesterone, prednisolone, or dexamethasone); steroids (one or multiple steroids) were considered a separate treatment if administered without erythropoietin.
- A treatment line was considered ended if followed by a treatment gap of ≥ 60 days.
- Kaplan-Meier analysis was performed to determine overall survival (OS).
- The effects of specific covariates (age, sex, and presence of splenomegaly) on OS were analyzed using a Cox proportional hazards model.

Table 1. Relevant ICD9 Codes for Diagnosis

	ICD9 Code
PMF	238.76
Post-PV/ET sMF	289.83 with prior history of PV (238.4)/ET (238.7)
Other sMF	sMF (289.83) without prior history of PV (238.4)/ET (238.7)

RESULTS

Patients

- 6982 patients in the Truven and Optum databases met the inclusion criteria; baseline characteristics are presented in **Table 2**.
- Median age at diagnosis was 66 years (interquartile range, 58-78 years).
 - 52% (n = 3,650) were aged > 65 years.
- More than half of patients were male (53%; n = 3,673).
- Most included patients were categorized as having Other sMF.
 - PMF: 23% (n = 1,637);
 - Post-PV/ET sMF: 14% (n = 956);
 - Other sMF: 63% (n = 4,389).

Splenomegaly

- At the time of index diagnosis (± 90 days), 11% (n = 749) of patients had splenomegaly (**Table 2**).
 - An additional 3% (n = 227) developed splenomegaly > 90 days following index diagnosis.

Platelets

- 112 patients had available baseline platelet counts (-90 to +180 days of index MF diagnosis; **Table 2**).
 - Most had platelet counts > 100,000/μL (78%; n = 87).

RESULTS (continued)

Table 2. Select Characteristics of Included Patients

	Optum (n = 859)	Truven (n = 6123)	Combined (N = 6,982)
Total patients, n (%)			
PMF	212 (25)	1,425 (23)	1,637 (23)
Post-PV/ET sMF	116 (14)	840 (14)	956 (14)
Other sMF	531 (62)	3,858 (63)	4,389 (63)
Age, mean (range)			
PMF	67 (22-84)	67 (20-102)	67 (20-102)
Post-PV/ET sMF	69 (33-84)	66 (24-97)	66 (24-97)
Other sMF	67 (20-85)	67 (18-103)	67 (18-103)
Males, n/n (%)			
PMF	117/212 (55)	829/1,425 (58)	946/1,637 (58)
Post-PV/ET sMF	62/116 (53)	401/840 (48)	463/956 (48)
Other sMF	283/531 (53)	1,981/3,858 (51)	2,264/4,389 (52)
Patients with splenomegaly, n/n (%)			
± 90 days within index MF diagnosis	91/859 (11)	658/6,123 (11)	749/6,982 (11)
> 90 days within index MF diagnosis	0	227/6,123 (4)	227/6,982 (3)
At any time	91/859 (11)	1,148/6,123 (19)	1,239/6,982 (18)
Patients with platelet counts available, n	89	23	112
Patients with platelet count within 90 days prior and 180 days following index MF diagnosis, n/n (%)			
< 50,000/μL	9/89 (10)	1/23 (4)	10/112 (9)
50,000-75,000/μL	11/89 (12)	2/23 (9)	13/112 (12)
76,000-100,000/μL	1/89 (1)	1/23 (4)	2/112 (2)
> 100,000/μL	68/89 (76)	19/23 (83)	87/112 (78)

Treatment

- Overall, median follow-up time for patients who had a line of therapy was 607 days.
- Overall, 57% (n = 3,950) of the 6,982 included patients received any/frontline treatment or supportive care (**Table 3**).
 - The most common frontline treatment or supportive care approaches were steroids alone in 27% of patients (n = 1,053) and hydroxyurea alone in 21% of patients (n = 811).
 - Ruxolitinib ± other treatment was given frontline to 12% (n = 488) of patients.
 - Median duration of treatment with frontline ruxolitinib was 7 months (interquartile range, 4-15 months).
 - Of the patients who discontinued/failed frontline ruxolitinib, 85% did not go on to another line of therapy.
- A total of 19% (n = 1,305) of the 6,982 included patients received second-line treatment or supportive care (**Table 4**).
 - As in the frontline setting, the most common second-line treatment or supportive care approaches were steroids alone in 27% (n = 348) and hydroxyurea alone in 21% (n = 276) of the 1,305 patients who received any second-line treatment.
 - Ruxolitinib ± other treatment was given in the second-line setting to 12% (n = 157) of the 1,305 patients who received any second-line treatment.
 - Median duration of treatment with second-line ruxolitinib was 6 months (interquartile range, 3-13 months).

Table 3. Distribution of Frontline Treatments

	Optum (n = 448)	Truven (n = 3,502)	Combined (n = 3,950)
Best supportive care ± other treatment, n (%)	50 (11)	753 (22)	803 (20)
Chemotherapy ± other treatment, n (%)	33 (7)	291 (8)	324 (8)
Hydroxyurea ± other treatment, n (%)	170 (38)	964 (28)	1,134 (29)
Ruxolitinib ± other treatment, n (%)	63 (14)	425 (12)	488 (12)
Radiation ± other treatment, n (%)	11 (2)	88 (3)	99 (3)
Splenectomy ± other treatment, n (%)	1 (< 1)	48 (1)	49 (1)
Steroids only, n (%)	120 (27)	933 (27)	1,053 (27)

Note: Best supportive care may have included erythropoietin ± steroids (danazol, fluoxymesterone, prednisolone, or dexamethasone); steroids (one or multiple steroids) were considered a separate treatment if administered without erythropoietin.

Table 4. Distribution of Second-Line Treatments

	Optum (n = 217)	Truven (n = 1,088)	Combined (n = 1,305)
Best supportive care ± other treatment, n (%)	20 (9)	232 (21)	252 (19)
Chemotherapy ± other treatment, n (%)	13 (6)	75 (7)	88 (7)
Hydroxyurea ± other treatment, n (%)	79 (36)	323 (30)	402 (31)
Ruxolitinib ± other treatment, n (%)	38 (18)	119 (11)	157 (12)
Radiation ± other treatment, n (%)	9 (4)	37 (3)	46 (4)
Splenectomy ± other treatment, n (%)	2 (1)	10 (1)	12 (1)
Steroids only, n (%)	56 (26)	292 (27)	348 (27)

Note: Best supportive care may have included erythropoietin ± steroids (danazol, fluoxymesterone, prednisolone, or dexamethasone); steroids (one or multiple steroids) were considered a separate treatment if administered without erythropoietin.

Survival

- Median OS for patients who received frontline ruxolitinib was 30 months compared with 22 months for patients receiving non-ruxolitinib treatment (hazard ratio [HR] = 0.7; 95% confidence interval, 0.6-0.8; **Figure 1**).
- Of the 488 patients who received frontline ruxolitinib, 23% (n = 112) went on to receive ≥ 1 further treatment.
 - Among these 112 patients who received further treatment following frontline ruxolitinib, 44% (n = 49) received a second-line regimen that also included ruxolitinib.
- Median OS among patients (n = 430) who failed or discontinued frontline ruxolitinib was 7 months (**Figure 2**).
- Median OS among patients who received frontline and second-line ruxolitinib was longer than that among patients who received frontline ruxolitinib and went on to a different second-line treatment or no treatment (30 months vs 14 months; $P < 0.05$; **Figure 3**).

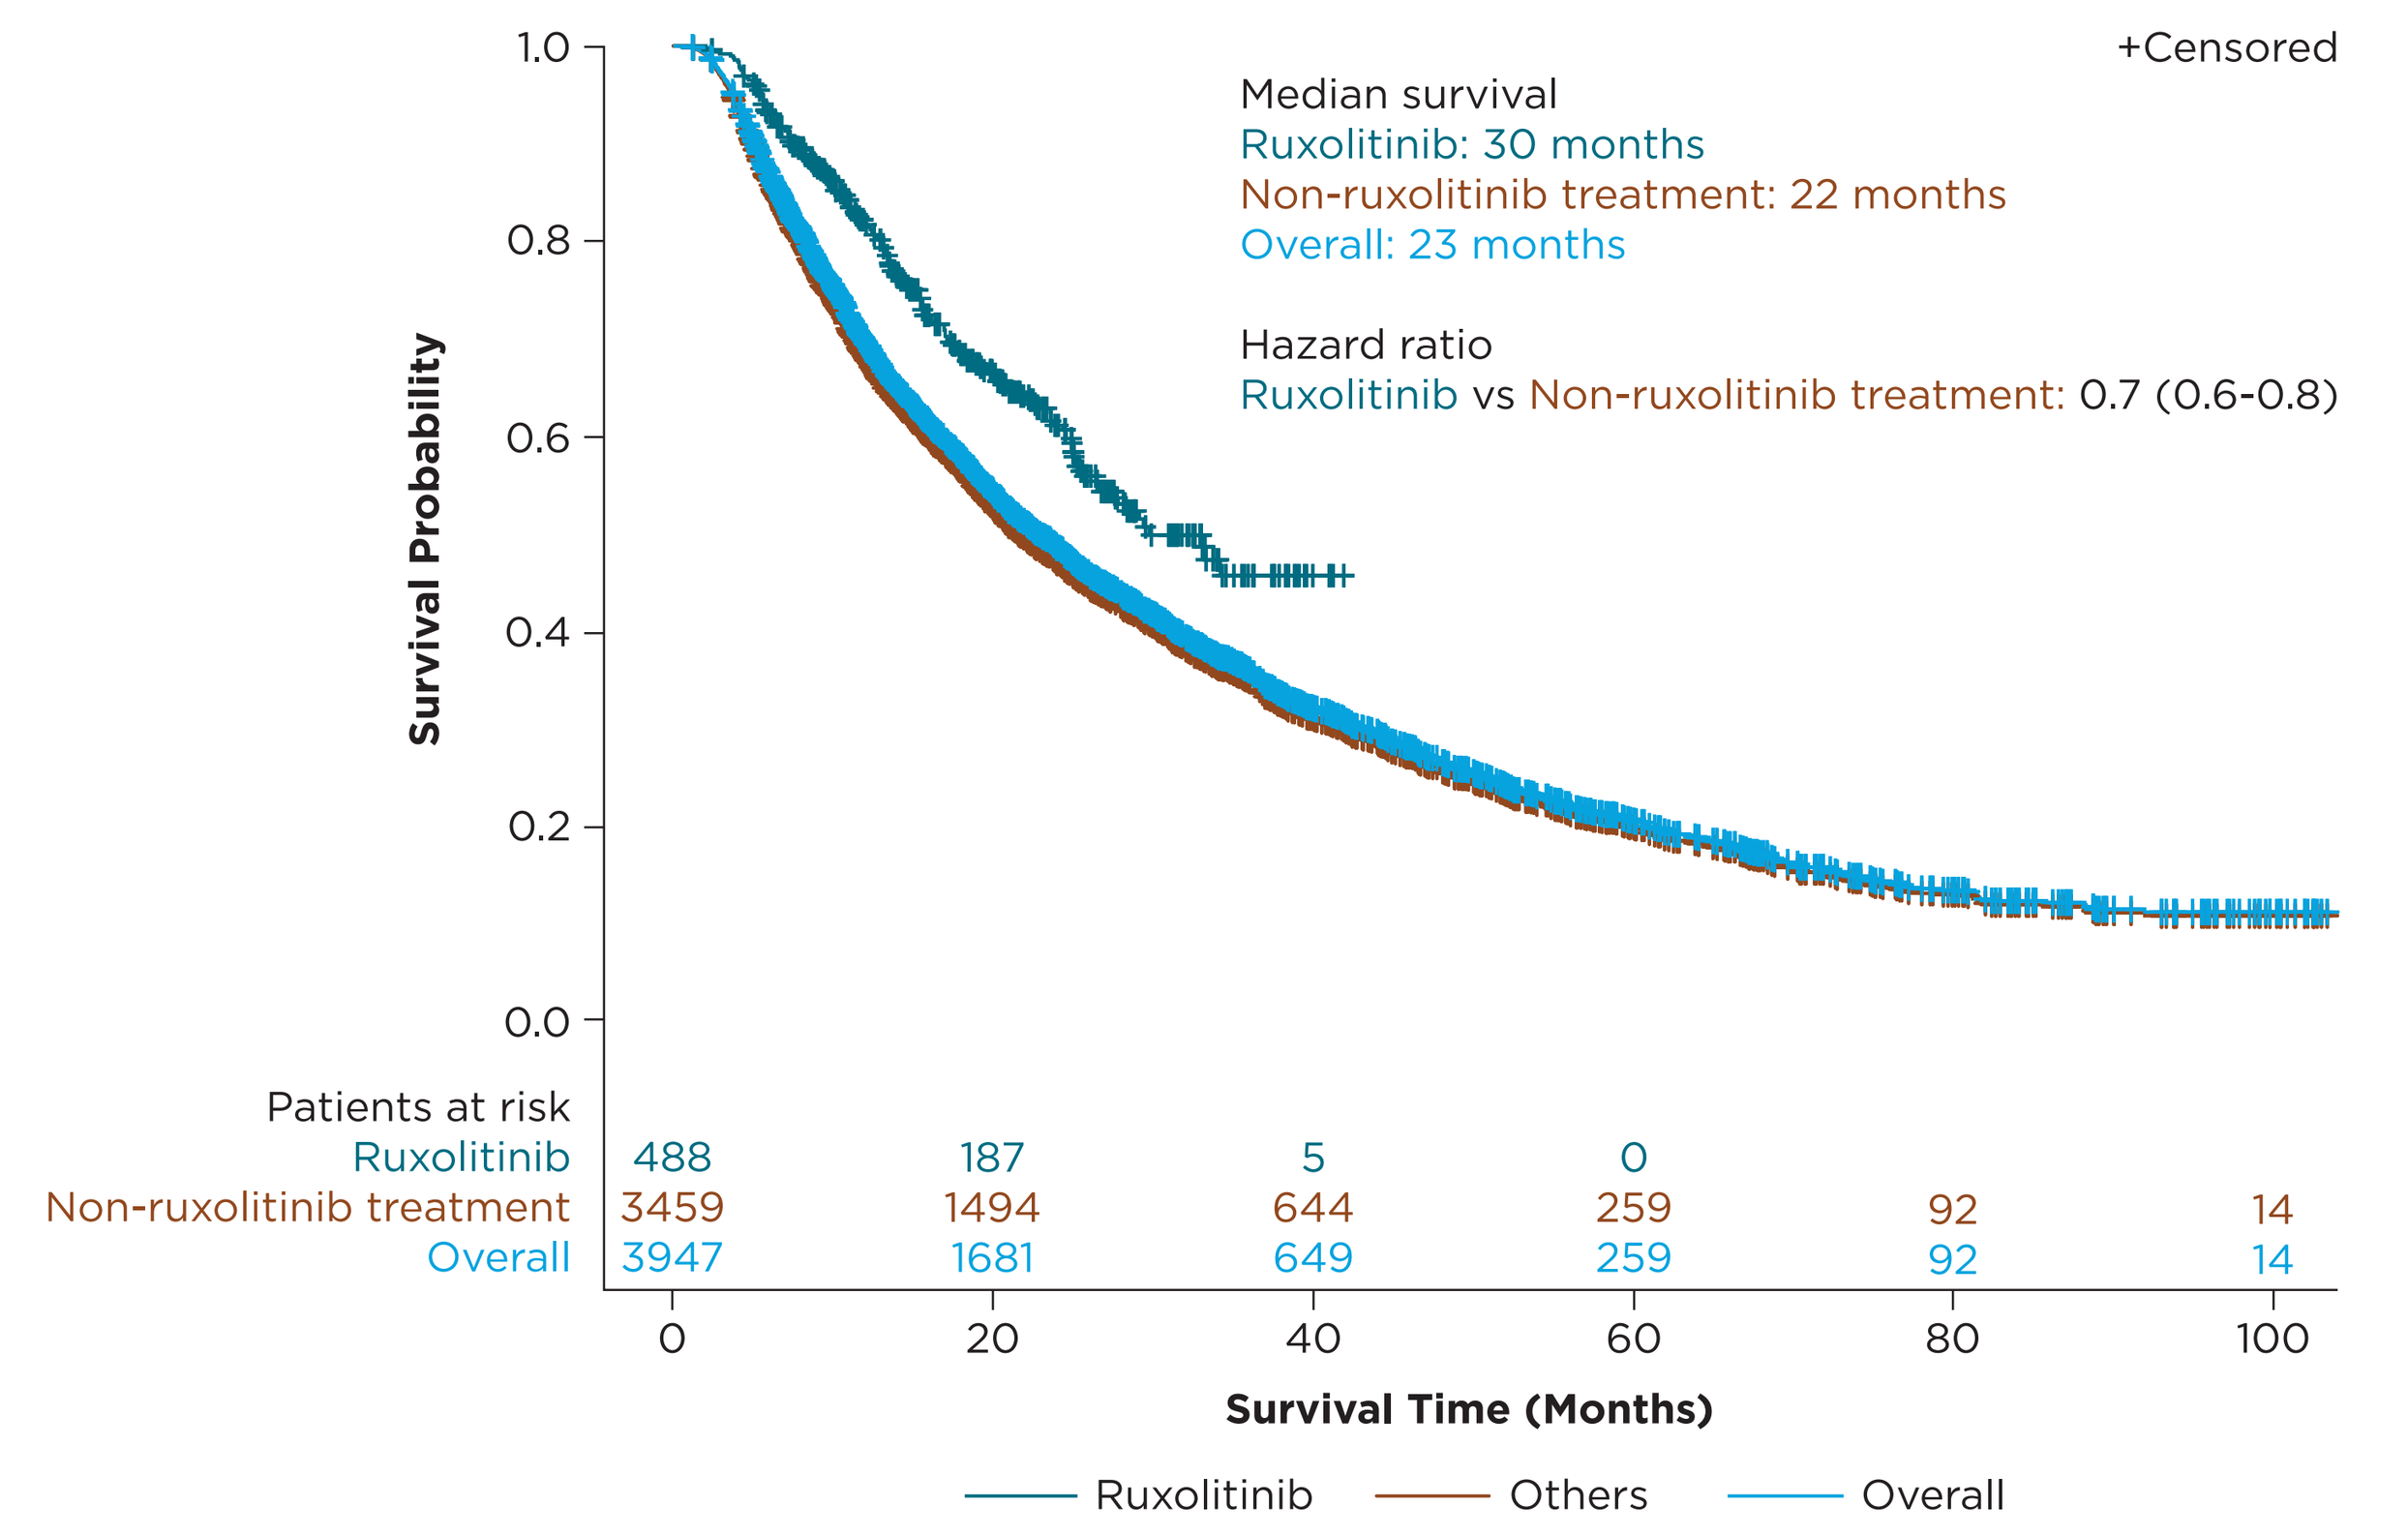
+Censored
Median survival
Ruxolitinib: 30 months
Non-ruxolitinib treatment: 22 months
Overall: 23 months
Hazard ratio
Ruxolitinib vs Non-ruxolitinib treatment: 0.7 (0.6-0.8)
Survival Probability
Patients at risk
Ruxolitinib 488 187 5 0
Non-ruxolitinib treatment 3459 1494 644 259 92 14
Overall 3947 1681 649 259 92 14
0 20 40 60 80 100
Survival Time (Months)
Ruxolitinib Others Overall

Figure 1. Survival of Patients With MF Who Received Frontline Treatments

Represented patients were stratified based on their first line of therapy. Patients were included in the "Ruxolitinib" category if they received a ruxolitinib regimen; otherwise, they were included in the "Non-ruxolitinib treatment" category. If date of death was known, it was treated as an event. Time to event was calculated as the time from start of frontline treatment to event/censoring. If date of death was unknown and last insurance claim was > 180 days prior to end of study, the date of last claim was considered date of death. If the last claim was ≤ 180 days prior to end of study, the patient was censored at either last claim date or end of study, whichever was later.

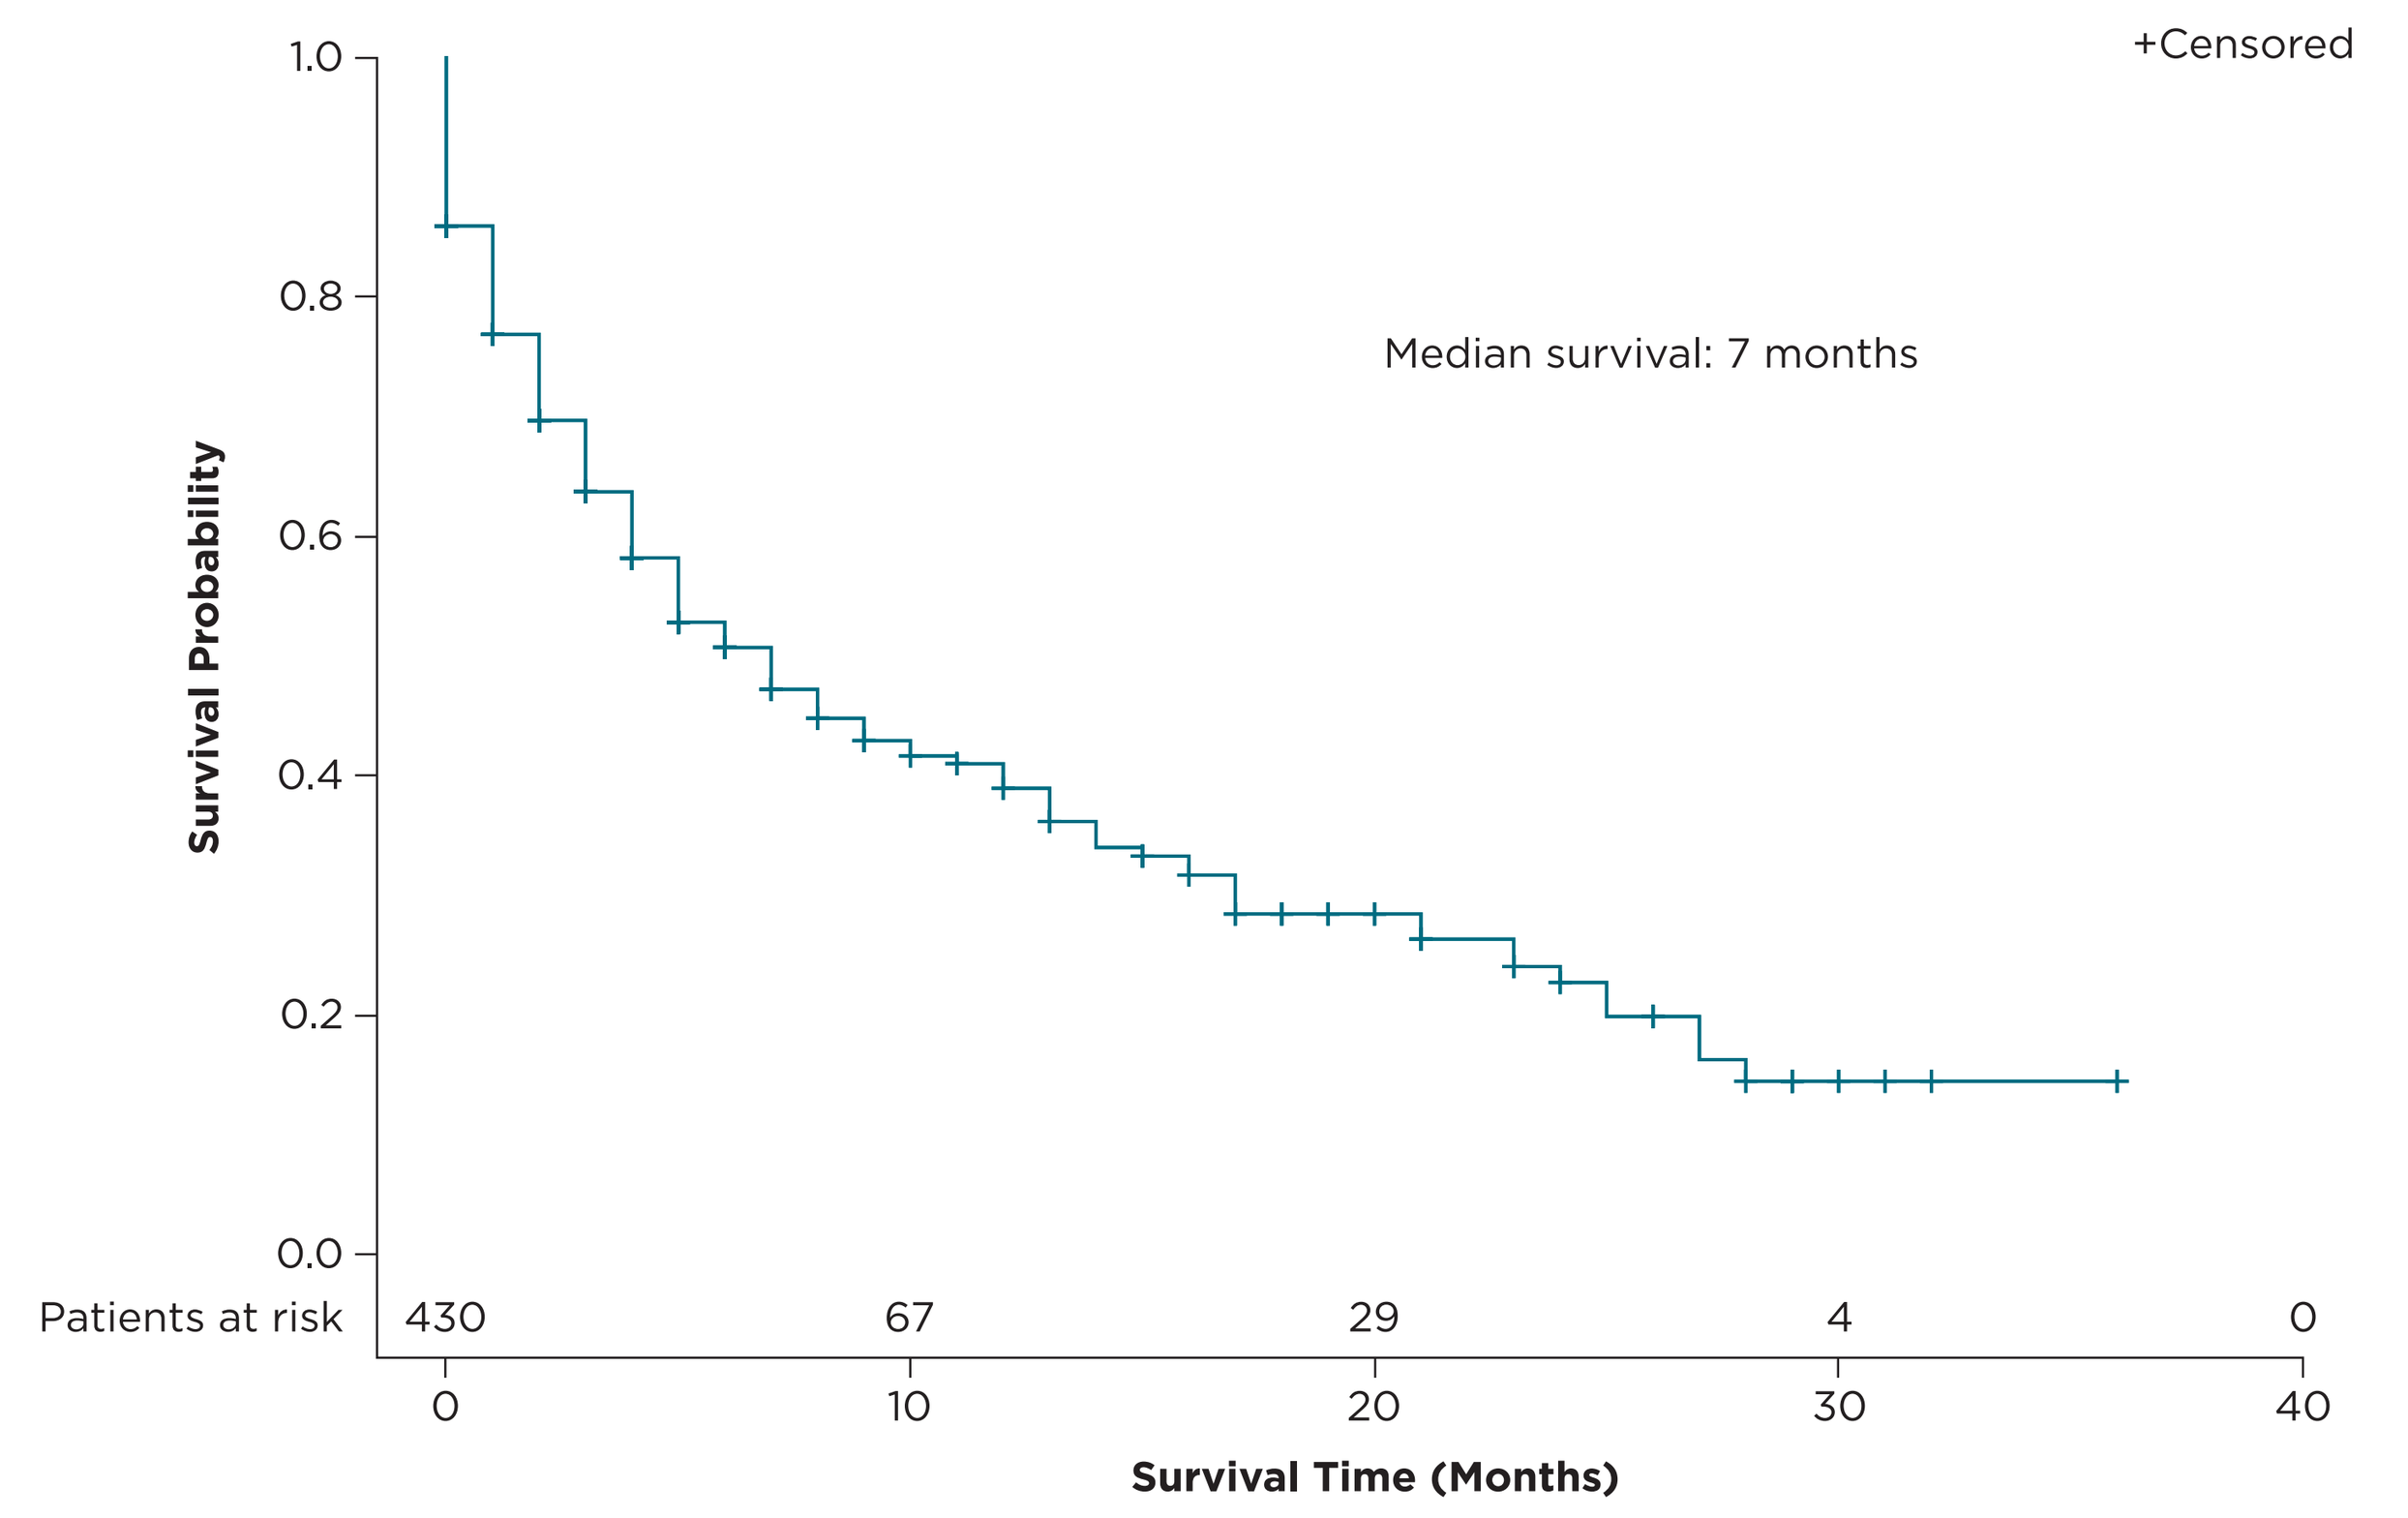
+Censored
Median survival: 7 months
Survival Probability
Patients at risk 430 67 29 4 0
0 10 20 30 40
Survival Time (Months)

Figure 2. Survival of Patients With MF Who Discontinued or Failed Frontline Ruxolitinib

Represented patients received frontline ruxolitinib and either went on to a different treatment in second line (15%) or received no further treatment (85%). If date of death was known, it was treated as an event. Time to event was calculated as the time from end of frontline treatment to event/censoring. If date of death was unknown and last insurance claim was > 90 days prior to end of study, the date of last claim was considered date of death. If the last claim was ≤ 90 days prior to end of study, the patient was censored at either last claim date or end of study, whichever was later.

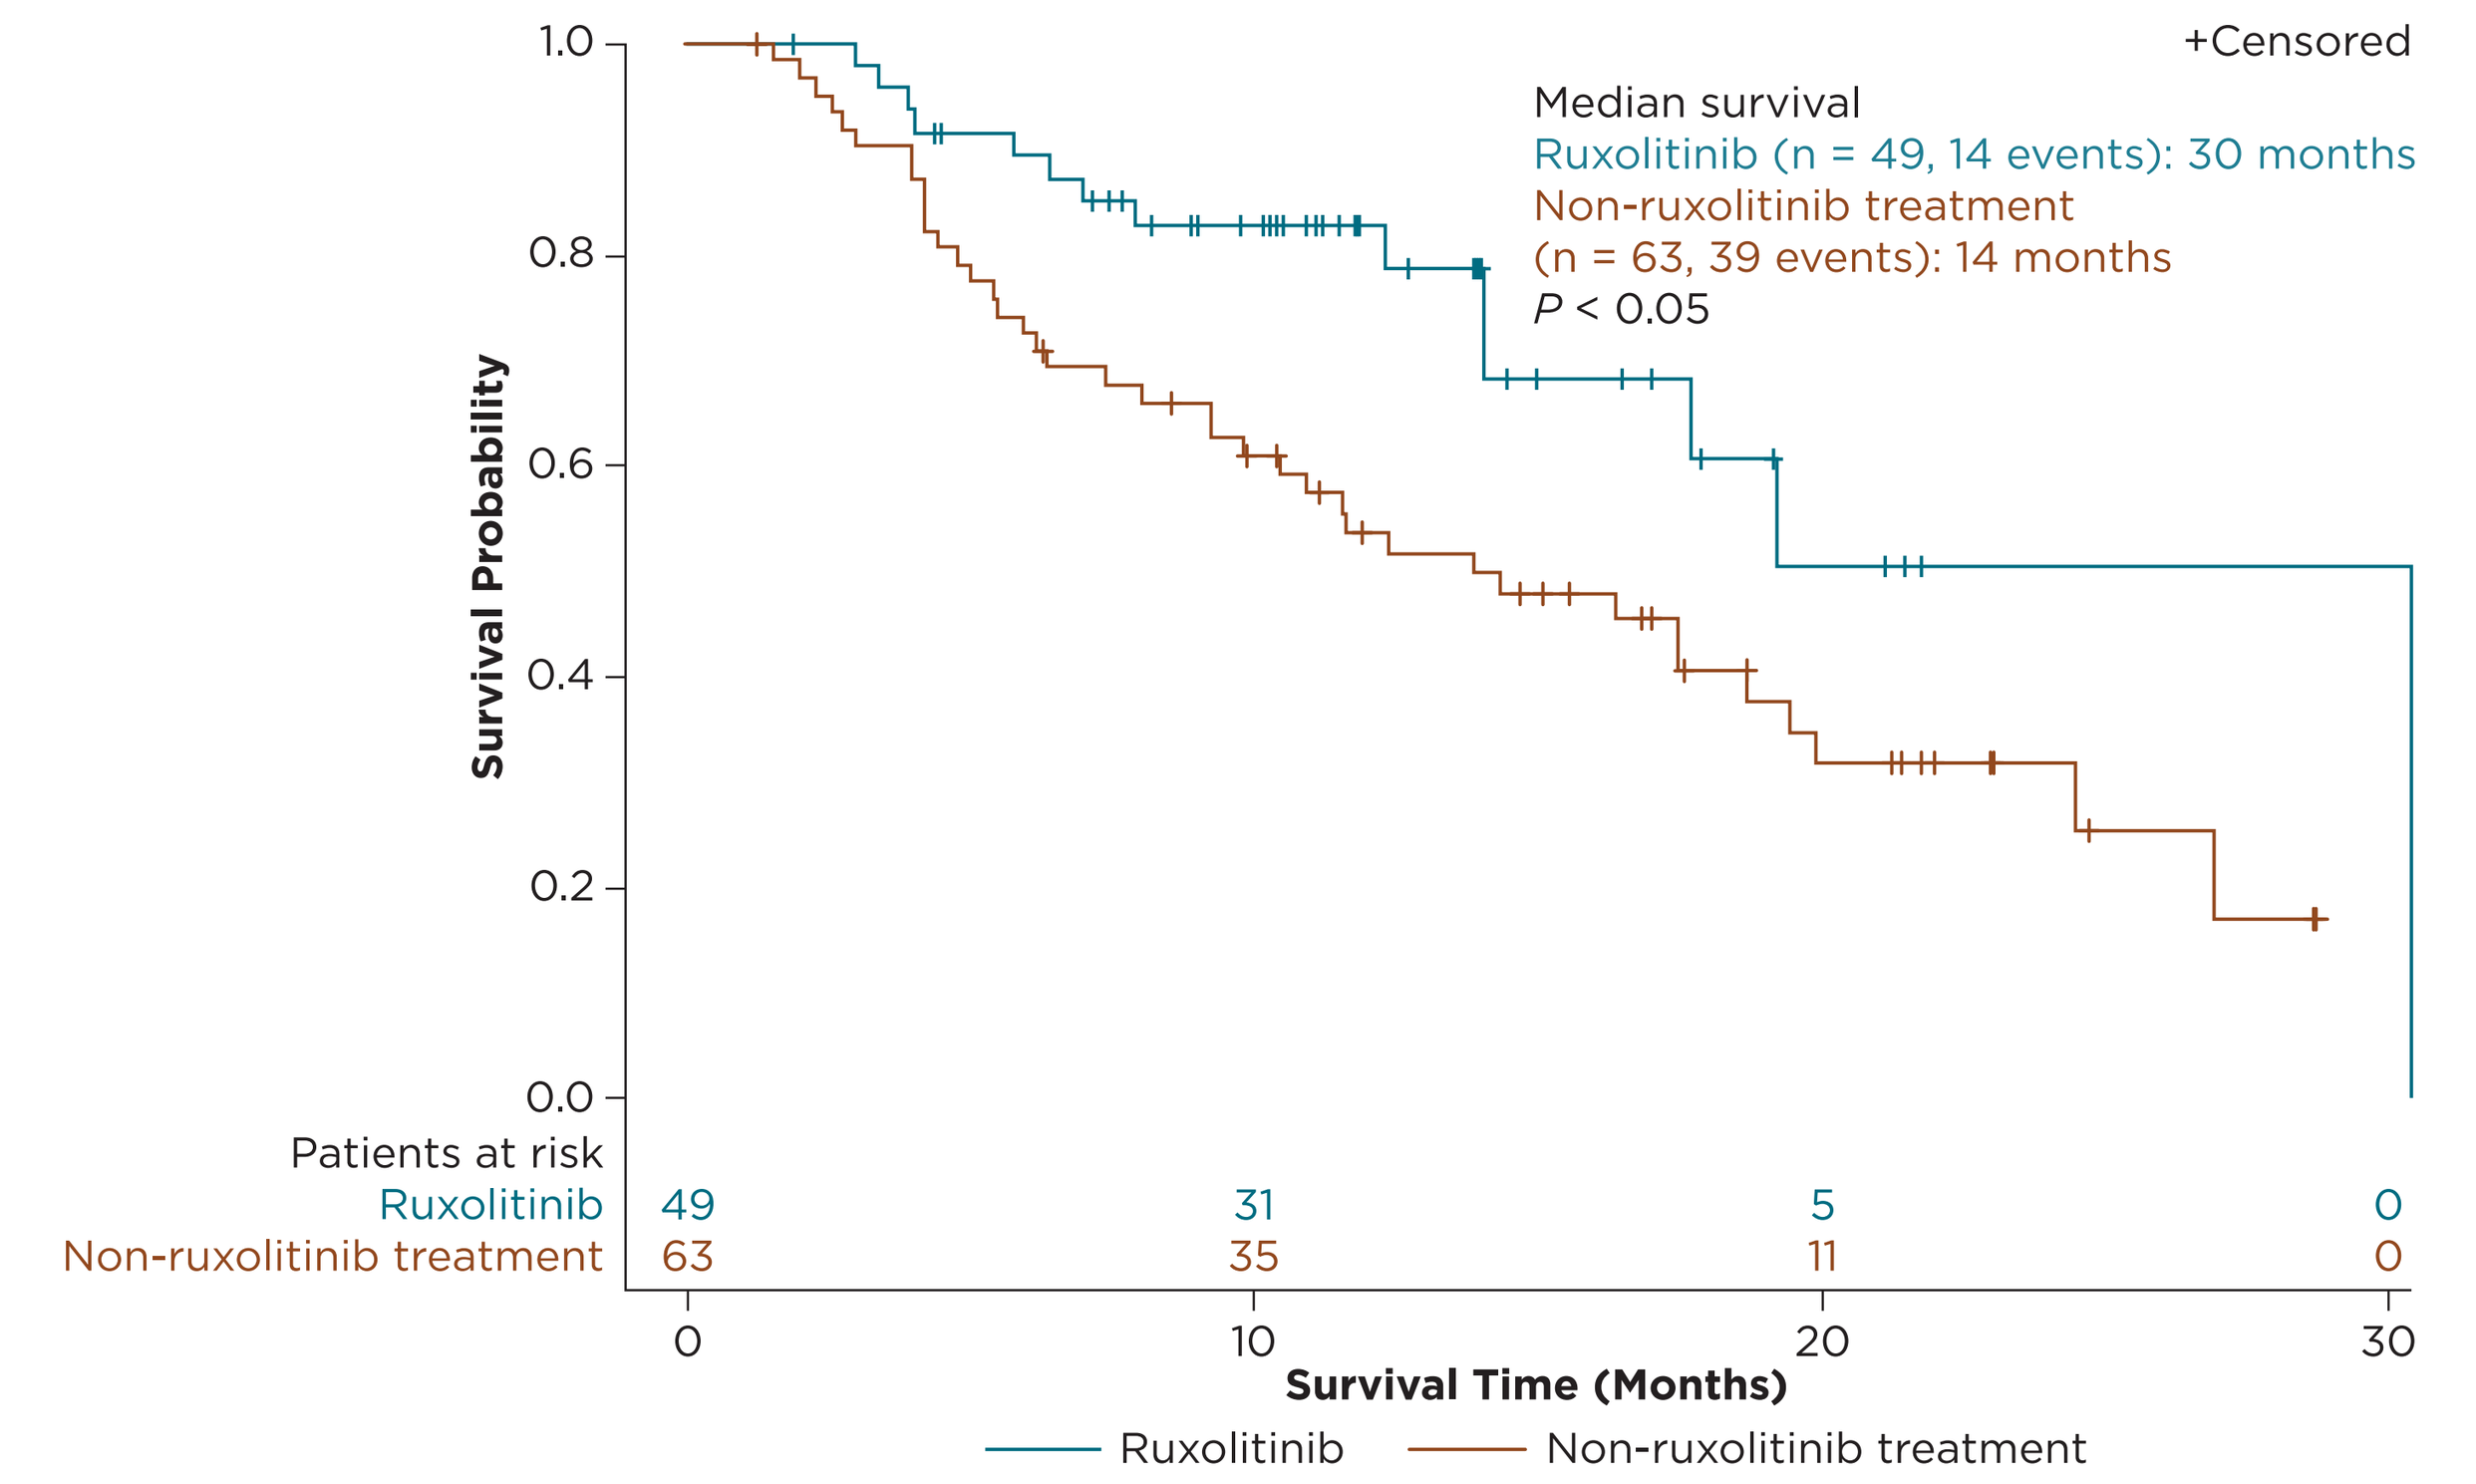
+Censored
Median survival
Ruxolitinib (n = 49, 14 events): 30 months
Non-ruxolitinib treatment
(n = 63, 39 events): 14 months
P < 0.05
Survival Probability
Patients at risk
Ruxolitinib 49 31 5 0
Non-ruxolitinib treatment 63 35 11 0
0 10 20 30
Survival Time (Months)
Ruxolitinib Non-ruxolitinib treatment

Figure 3. Survival of Patients Receiving Second-Line Therapy Who Had Received Frontline Ruxolitinib

Represented patients received frontline ruxolitinib and either went on to ruxolitinib or a different treatment, in second line. Time to event was calculated as the time from start of second-line treatment to event/censoring. If date of death was unknown and last insurance claim was > 90 days prior to end of study, the date of last claim was considered date of death. If the last claim was ≤ 90 days prior to end of study, the patient was censored at either last claim date or end of study, whichever was later.

- Survival after failure or discontinuation of ruxolitinib was not statistically significantly associated with the covariates tested.
 - Sex (HR = 1.03; $P = 0.85$);
 - Age (< 65 vs ≥ 65 years; HR = 0.88; $P = 0.50$);
 - Presence of splenomegaly (-90 days before index diagnosis to any point after index diagnosis; HR = 0.87; $P = 0.48$).

CONCLUSIONS

- Most patients diagnosed with MF were aged ≥ 65 years and had neither splenomegaly nor thrombocytopenia at baseline.
- Limitations of these results include:
 - Potential information bias since a physician may fail to report a symptom such as splenomegaly;
 - The small number of patients with available platelet counts;
 - Ruxolitinib received approval in 2011 from the US Food and Drug Administration for treating patients with MF[3]; therefore, its use was not approved for the entire time frame included in the present database analysis (diagnosis between 2006 and 2015).
- In the present database analysis, many patients (43%) received no treatment or supportive care.
- Although only a fraction of patients received ruxolitinib, frontline treatment with ruxolitinib was associated with favorable median OS.
- Most patients (85%) who failed or discontinued frontline ruxolitinib received no further treatment.
- However, median OS was greatly reduced (7 months) once patients failed or discontinued ruxolitinib.
- Additional treatment options for patients who discontinue ruxolitinib are needed.

REFERENCES

1. Chow V, et al. *Onco Targets Ther.* 2016;9:2655-2665.
2. Tefferi A, et al. *Blood.* 2013;122:1395-1398.
3. Jakafi (ruxolitinib) [prescribing information]. Wilmington, DE: Incyte Corporation. March 2016.
4. NCCN Clinical Practice Guidelines in Oncology. Myeloproliferative Neoplasms v2.2017. National Comprehensive Cancer Network (NCCN). Accessed October 25, 2016.
5. Vannucchi AM, et al. *Ann Oncol.* 2015;26(Suppl 5):v85-v99.
6. Jabbour E, et al. *Blood* (Presented at American Society of Hematology Annual Meeting). 2013;122: Abstract 1584.

ACKNOWLEDGMENTS

The authors would like to thank Hemanth Kanakamedala of SmartAnalyst Inc. for additional assistance with data analyses.
The study was funded by Janssen Research & Development, LLC. Writing assistance was provided by Tamara Fink, PhD, of PAREXEL, with funding from Janssen Global Services, LLC.

DISCLOSURES

MM, JH, GW, SM, and JB are employed by and hold stock in Janssen Research & Development. RP is an employee of SmartAnalyst Inc, a company contracted by Janssen to carry out the analysis. All authors participated in data interpretation, development, and approval of the abstract and poster presentation.

An electronic version of the poster can be viewed by scanning the QR code. The QR code is intended to provide scientific information for individual reference. The PDF should not be altered or reproduced in any way.

Poster presented at the American Society of Hematology (ASH) 58th Annual Meeting, December 3-6, 2016, San Diego, California.

Sponsored by Janssen Research & Development, LLC