DISEASE MODIFYING ACTIVITY OF IMETELSTAT IN PATIENTS WITH HEAVILY TRANSFUSED NON-DEL(5Q) LOWER-RISK MYELODYSPLASTIC NEOPLASMS RELAPSED/REFRACTORY/INELIGIBLE FOR ERYTHROPOIESIS-STIMULATING AGENTS IN IMerge PHASE III

soho2023 ELEVENTH ANNUAL MEETING

MDS-605

V. SANTINI[1], U. PLATZBECKER[2], P. FENAUX[3], M.A. SEKERES[4], M.R. SAVONA[5], Y.F. MADANAT[6], M. DÍEZ-CAMPELO[7], D. VALCÁRCEL-FERREIRAS[8], T. ILLMER[9], A. JONÁŠOVÁ[10], P. BĚLOHLÁVKOVÁ[11], L. SHERMAN[12], T. BERRY[12], S. DOUGHERTY[12], S. SHAH[12], Q. XIA[12], L. PENG[12], L. SUN[12], Y. WAN[12], F. HUANG[12], A. IKIN[12], S. NAVADA[12], F. FELLER[12], A.M. ZEIDAN[13*], R.S. KOMROKJI[14*]

[1]Azienda Ospedaliero Universitaria Careggi, University of Florence, Florence, Italy; [2]Leipzig University Hospital, Leipzig, Germany; [3]Hôpital Saint-Louis, Université de Paris 7, Paris, France; [4]Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL, USA; [5]Vanderbilt-Ingram Cancer Center, Vanderbilt University Medical Center, Nashville, TN, USA; [6]Harold C. Simmons Comprehensive Cancer Center, UT Southwestern Medical Center, Dallas, TX, USA; [7]University Hospital of Salamanca, Salamanca, Spain; [8]Hospital Universitari Vall d'Hebron, Barcelona, Spain; [9]Hematology Private Practice, Dresden, Germany; [10]General Hospital, Prague, Czech Republic; [11]Charles University Hospital Hradec Kralove, Czech Republic; [12]Geron Corporation, Parsippany, NJ, USA; [13]Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA; [14]Moffitt Cancer Center, Tampa, FL, USA. *Contributed equally.

INTRODUCTION

- Imetelstat is a first-in-class direct and competitive inhibitor of telomerase that specifically targets MDS clones with abnormally high telomerase activity, enabling the recovery of effective hematopoiesis[1-4]
- In the phase 2 part of the IMerge study, patients with LR-MDS who were heavily RBC transfusion dependent, R/R to or ineligible for ESAs, non-del(5q), and naive to lenalidomide and HMA achieved durable and continuous RBC-TI when treated with imetelstat,[5] with an 8-week RBC-TI rate of 42% and a median TI duration of 86 weeks
- The phase 3 results from IMerge in the same patient population were highly consistent with the phase 2 efficacy results, with an 8-week RBC-TI rate of 40% and median TI duration approaching 1 year, a 24-week RBC-TI of 28%, and a 1-year RBC-TI of 18%[6]
- This poster presents the analysis of phase 3 results on the reduction of MDS clones with imetelstat vs placebo, suggesting potential disease-modifying activity of imetelstat

Figure 1. IMerge Phase 3 Trial Design (MDS3001; NCT02598661)

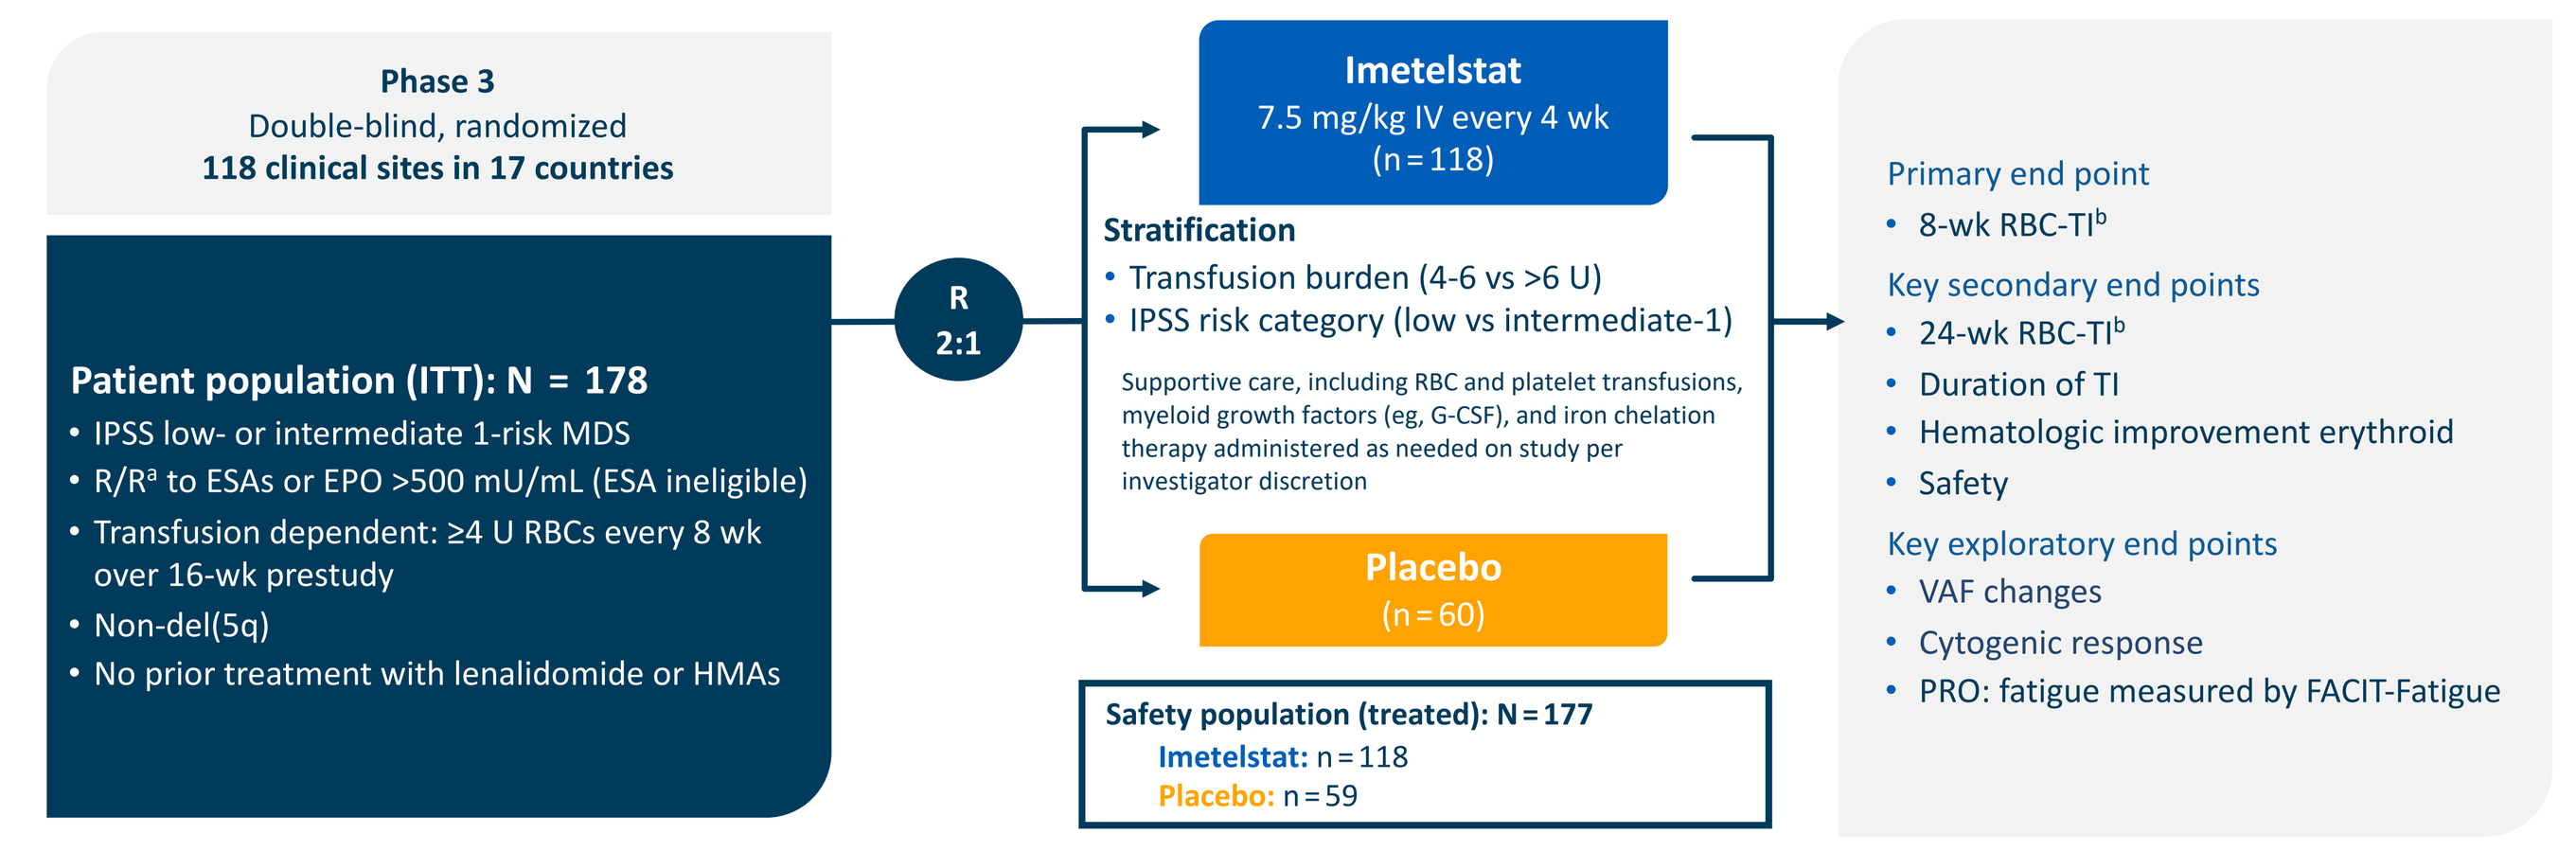
Phase 3
Double-blind, randomized
118 clinical sites in 17 countries
Patient population (ITT): N = 178
• IPSS low- or intermediate 1-risk MDS
• R/Rª to ESAs or EPO >500 mU/mL (ESA ineligible)
• Transfusion dependent: ≥4 U RBCs every 8 wk over 16-wk prestudy
• Non-del(5q)
• No prior treatment with lenalidomide or HMAs
R 2:1
Imetelstat
7.5 mg/kg IV every 4 wk
(n = 118)
Stratification
• Transfusion burden (4-6 vs >6 U)
• IPSS risk category (low vs intermediate-1)
Supportive care, including RBC and platelet transfusions, myeloid growth factors (eg, G-CSF), and iron chelation therapy administered as needed on study per investigator discretion
Placebo
(n = 60)
Safety population (treated): N = 177
Imetelstat: n = 118
Placebo: n = 59
Primary end point
• 8-wk RBC-TIᵇ
Key secondary end points
• 24-wk RBC-TIᵇ
• Duration of TI
• Hematologic improvement erythroid
• Safety
Key exploratory end points
• VAF changes
• Cytogenic response
• PRO: fatigue measured by FACIT-Fatigue

[a]Received ≥8 weeks of ESA treatment (epoetin alfa ≥40,000 units, epoetin beta ≥30,000 units, darbepoetin alfa 150 µg, or equivalent per week) without Hb rise ≥1.5 g/dL or decreased RBC transfusion requirement ≥4 U per 8 weeks or transfusion dependence or reduction in Hb level by ≥1.5 g/dL after hematologic improvement from ≥8 weeks of ESA treatment. [b]Proportion of patients without any RBC transfusion for ≥8 consecutive weeks since entry to the trial (8-week TI); proportion of patients without any RBC transfusion for ≥24 consecutive weeks since entry to the trial (24-week TI).

AIM

- To evaluate cytogenetic response, VAF changes, and clinical correlates in phase 3 of the IMerge study

METHODS

- In the IMerge phase 3 trial, a double-blind, randomized (2:1), placebo-controlled study conducted at 118 global sites between 2019 and 2022, patients with heavily RBC-TD ESA-relapsed/refractory/ineligible non-del(5q) LR-MDS naive to lenalidomide/HMA were randomized to receive imetelstat 7.5 mg/kg (n = 118) or placebo (n = 60) every 4 weeks until disease progression, unacceptable toxicity, withdrawal of consent, or lack of response
- The main outcomes measures were cytogenetic response assessed by BM aspirates collected every 24 weeks posttreatment in patients with baseline cytogenetic abnormalities. VAF changes posttreatment assessed by blood samples collected every 12 weeks in patients with ≥5% VAF at baseline and ≥1 postbaseline assessment

RESULTS

- Demographics and disease characteristics were balanced between study arms (**Table 1**)
 - The study comprised 118 and 60 patients in the imetelstat and placebo arms, respectively

Table 1. Demographics and Disease Characteristics

Characteristic	Imetelstat (n = 118)	Placebo (n = 60)
Age, median (range), y	72 (44-87)	73 (39-85)
Male, n (%)	71 (60)	40 (67)
Time since diagnosis, median (range), y	3.5 (0.1-26.7)	2.8 (0.2-25.7)
WHO classification, n (%)		
RS+	73 (62)	37 (62)
RS−	44 (37)	23 (38)
IPSS risk category, n (%)		
Low	80 (68)	39 (65)
Intermediate-1	38 (32)	21 (35)
Pretreatment Hb level, median (range),[a] g/dL	7.9 (5.3-10.1)	7.8 (6.1-9.2)
Prior RBC transfusion burden, median (range), RBC U/8 wk	6 (4-33)	6 (4-13)
Prior RBC transfusion burden, n (%)		
≥4 to ≤6 U/8 wk	62 (53)	33 (55)
>6 U/8 wk	56 (48)	27 (45)
sEPO, median (range), mU/mL	174.9 (6.0-4460.0)	277.0 (16.9-5514.0)
sEPO level, n (%)[b]		
≤500 mU/mL	87 (74)	36 (60)
>500 mU/mL	26 (22)	22 (37)
Prior ESA, n (%)	108 (92)	52 (87)
Prior luspatercept, n (%)[c]	7 (6)	4 (7)

DCO date, October 13, 2022. [a]Average of all Hb values in the 8 weeks before the first dose date, excluding values within 14 days after a transfusion, which was considered to be influenced by transfusion. [b]Data missing for 5 patients in the imetelstat group and 2 in the placebo group. [c]Insufficient number of patients previously treated with luspatercept to draw conclusions about the effect of imetelstat treatment in such patients.

- Most common AEs with imetelstat were hematologic and of short duration (**Table 2**)
 - Median duration of grade 3-4 thrombocytopenia and neutropenia was <2 weeks; >80% of events were reversible to grade ≤2 within 4 weeks (**Table 3**)
 - 41 Patients (34.7%) in the imetelstat group and 2 patients (3.4%) in the placebo group had ≥1 dose of a myeloid growth factor mostly within cycles 2-4
 - Clinical consequences of grade 3-4 infection and bleeding were low and similar for imetelstat and placebo

Table 2. AEs With Imetelstat vs Placebo

Hematologic AEs (≥10% of patients), n (%)	Imetelstat (n = 118)		Placebo (n = 59)	
	Any grade	Grade 3-4	Any grade	Grade 3–4
Thrombocytopenia	89 (75)	73 (62)	6 (10)	5 (8)
Neutropenia	87 (74)	80 (68)	4 (7)	2 (3)
Anemia	24 (20)	23 (19)	6 (10)	4 (7)
Leukopenia	12 (10)	9 (8)	1 (2)	0

Table 3. Duration of Cytopenias

Grade 3-4 cytopenias (per laboratory value)	Imetelstat (n = 118)	Placebo (n = 59)
Thrombocytopenia		
Duration, median (range), wk	1.4 (0.1-12.6)	2.0 (0.3-11.6)
Resolved within 4 wk, %	86.3	44.4
Neutropenia		
Duration, median (range), wk	1.9 (0-15.9)	2.2 (1.0-4.6)
Resolved within 4 wk, %	81.0	50.0

- Primary end point of 8-week RBC-TI rate was significantly higher with imetelstat vs placebo
 - The ≥8-week RBC-TI rate was 40% in the imetelstat vs 15% in the placebo group (**Fig. 2**)

Figure 2. 8-Week to 1-Year RBC-TI Rate With Imetelstat vs Placebo

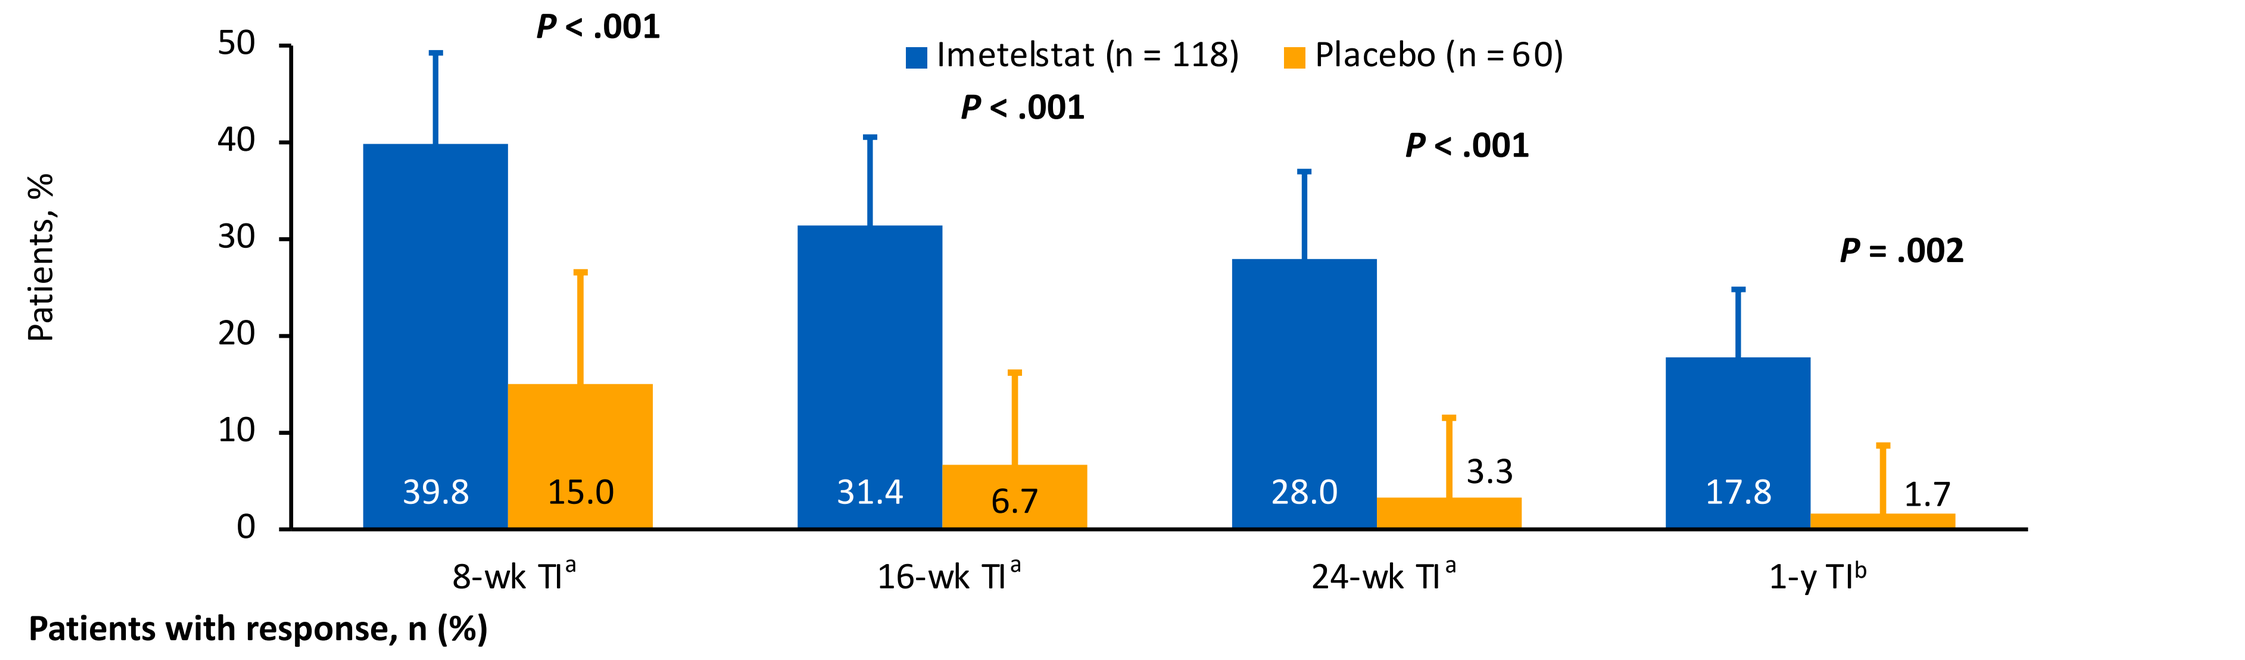
Imetelstat (n = 118)
Placebo (n = 60)
Patients, %
P < .001
P < .001
P < .001
P = .002
39.8 15.0
31.4 6.7
28.0 3.3
17.8 1.7
8-wk TIª
16-wk TIª
24-wk TIª
1-y TIᵇ

Patients with response, n (%) 95% CI	8-wk TI[a]	16-wk TI[a]	24-wk TI[a]	1-y TI[b]
Imetelstat	47 (39.8) 30.9-49.3	37 (31.4) 23.1-40.5	33 (28.0) 20.1-37.0	21 (17.8) 11.4-25.9
Placebo	9 (15.0) 7.1-26.6	4 (6.7) 1.9-16.2	2 (3.3) 0.4-11.5	1 (1.7) 0.0-8.9

Primary end point 8-week TI and the first secondary end point 24-week TI were statistically significant by the study's prespecified gate-keeping testing procedure. P Value determined by the Cochran-Mantel-Haenszel test, with stratification for prior RBC transfusion burden (≥4 to ≤6 vs >6 RBC U per 8 weeks during a 16-week period prerandomization) and baseline IPSS risk category (low vs intermediate-1) applied to randomization. [a]DCO date, October 13, 2022. [b]DCO date, January 13, 2023.

- A higher percentage of patients achieved cytogenetic response with imetelstat vs placebo (**Table 4**)
 - Among patients with cytogenetic abnormalities at baseline, the cytogenetic response rate was 35% (9 of 26) in the imetelstat group and 15% (2 of 13) in the placebo group
 - Among cytogenetic responders, 89% of patients (8 of 9) in the imetelstat group and 50% (1 of 2) in the placebo group also achieved 8-week RBC-TI

Table 4. Cytogenetic Response With Imetelstat vs Placebo

Cytogenetic response[a]	Imetelstat (n = 118)	Placebo (n = 60)
Patients with baseline cytogenetic abnormality based on central laboratory review, n (%)[b]	26 (22)	13 (22)
Cytogenetic best response, n (%)[c,d]		
Cytogenetic CR	5 (19)	1 (8)
Cytogenetic PR	4 (15)	1 (8)
Cytogenetic CR or PR criteria not met	5 (19)	5 (39)
Not evaluable	12 (46)	6 (46)
Cytogenetic CR or PR, n (%)[d]	9 (35)	2 (15)
95% CI[e]	17-56	2-45
% Difference (95% CI)[f]	19 (−16 to 44)	
***P* Value[g]**	0.216	

[a]Cytogenetic testing was done centrally, and the cytogenetic response was assessed by IRC. [b]Percentages calculated using the number of patients in each treatment group as the denominator. [c]Only patients considered for IRC adjudication are those assessed as having baseline cytogenetic abnormality by the IRC based on central laboratory data. [d]Percentages calculated using the number of patients with a baseline cytogenetic abnormality per central laboratory review within each treatment group as the denominator. [e]Exact Clopper-Pearson CI. [f]Wilson score CI. [g]P Value derived from the Cochran-Mantel-Haenszel test controlling for prior RBC transfusion burden (≤6 vs >6 U RBC) and IPSS risk group (low vs intermediate-1) applied to randomization.

- More patients treated with imetelstat vs placebo had ≥50% reduction in central BM RS
 - A higher percentage of patients treated with imetelstat than placebo had a ≥50% reduction in central BM RS (**Fig. 3**)
 - Reduction in BM RS associated with TI response: RBC-TI responders were enriched in patients achieving a ≥50% reduction in central BM RS
- More patients treated with imetelstat vs placebo had ≥50% reductions in mutations of interest
 - Mutations on 36 MDS-associated genes were tested by NGS at baseline and after treatment
 - Among patients with evaluable mutation data, the maximum reductions in VAF of the *SF3B1, TET2, DNMT3A,* and *ASXL1* genes were greater with imetelstat than placebo (**Fig. 4**)
- VAF reduction correlated with improved outcomes in patients treated with imetelstat
 - Among patients treated with imetelstat who achieved ≥50% *SF3B1* and ≥50% *TET2* VAF reduction, 83% were 8-week RBC-TI responders (**Fig. 5**)
 - ≥24-Week and ≥1-year RBC-TI responders were also enriched in patients achieving ≥50% reduction in *SF3B1* VAF or ≥50% reduction in *TET2* VAF (**Fig. 5**)
 - VAF reduction with imetelstat correlated with longer RBC-TI duration and increased Hb levels (**Fig. 6**)

Figure 3. Percent of Patients With ≥50% Reduction in Central BM RS With Imetelstat vs Placebo

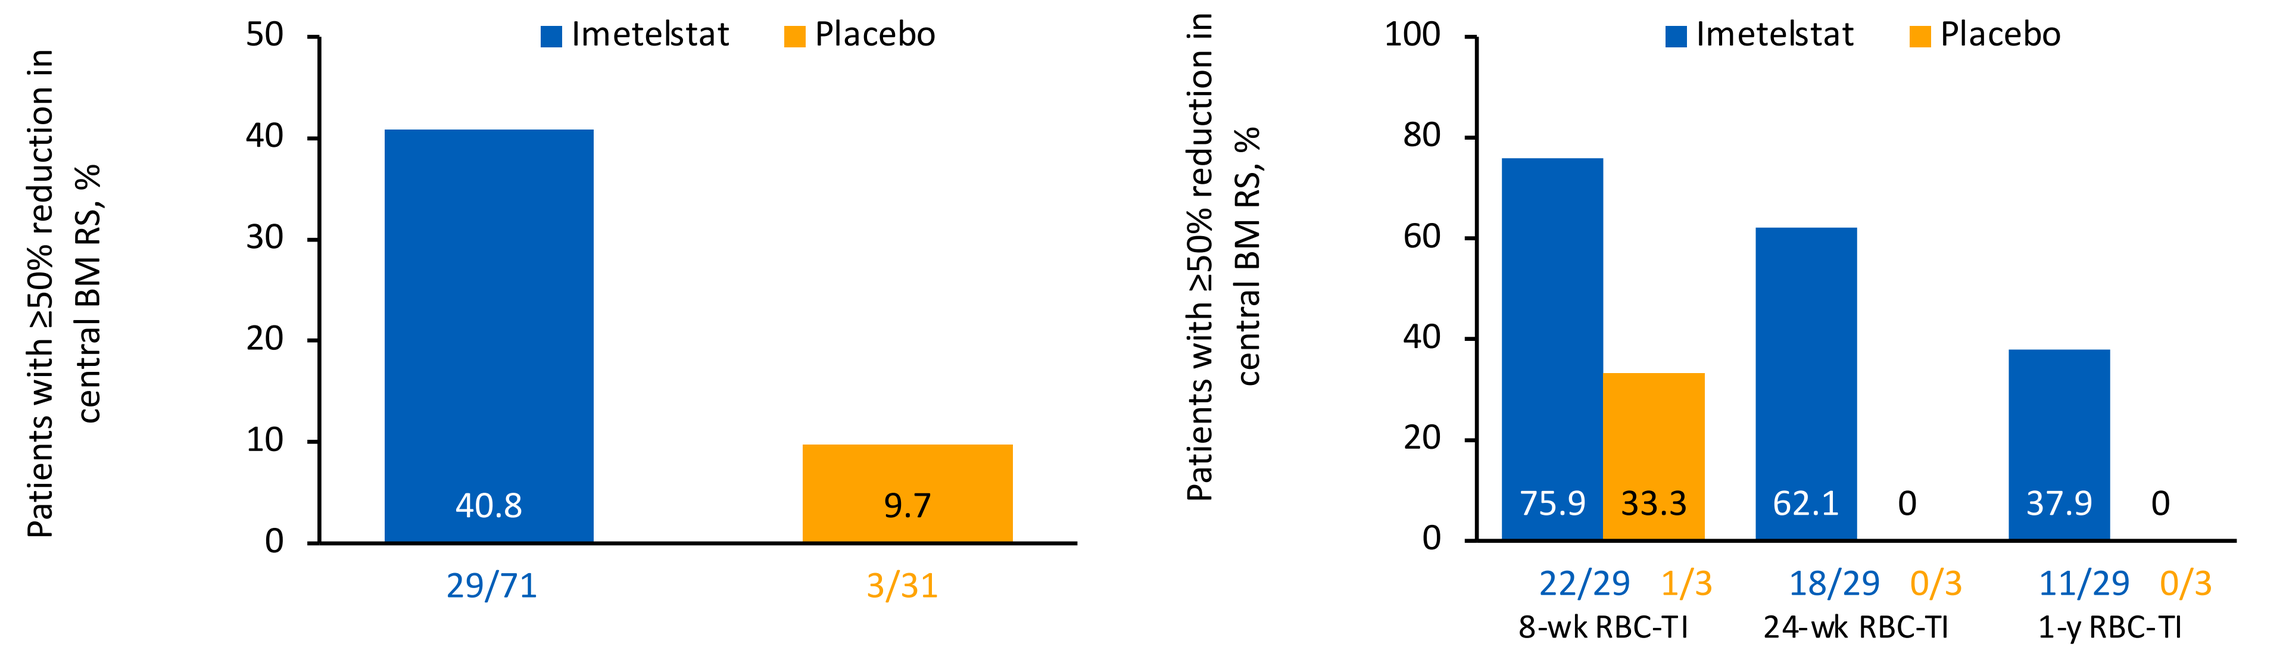
Imetelstat
Placebo
Patients with ≥50% reduction in central BM RS, %
40.8
9.7
29/71
3/31
75.9 33.3
62.1 0
37.9 0
22/29 1/3
18/29 0/3
11/29 0/3
8-wk RBC-TI
24-wk RBC-TI
1-y RBC-TI

Assessment was evaluated by central pathology review. Patients with baseline RS ≥15% and ≥1 postbaseline assessment were considered.

Figure 4. Percent of Patients With ≥50% Reduction in VAFs With Imetelstat vs Placebo

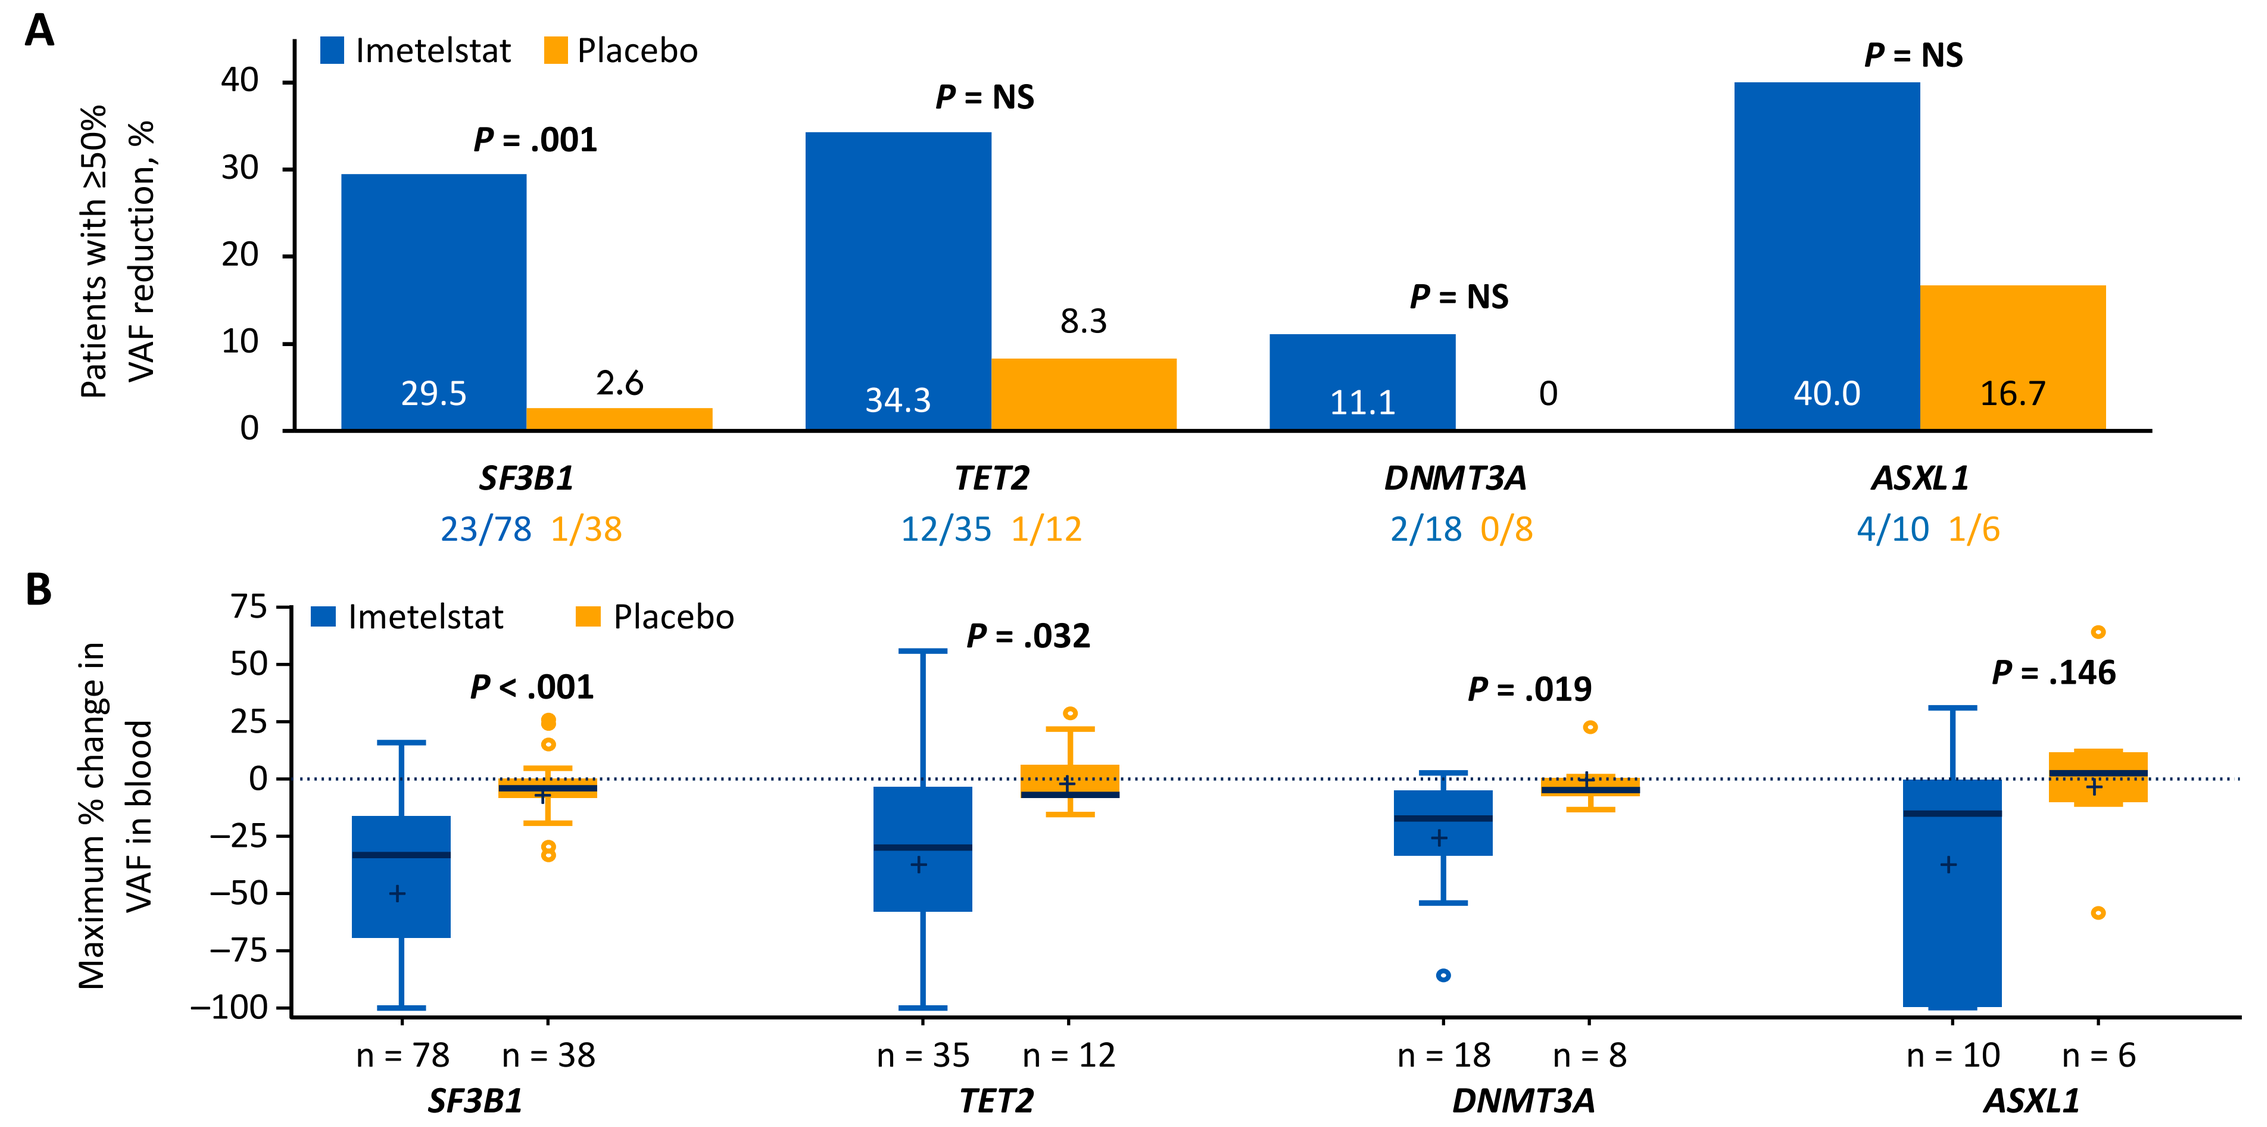
A
Imetelstat
Placebo
Patients with ≥50% VAF reduction, %
P = .001
P = NS
P = NS
P = NS
29.5 2.6
34.3 8.3
11.1 0
40.0 16.7
SF3B1
23/78 1/38
TET2
12/35 1/12
DNMT3A
2/18 0/8
ASXL1
4/10 1/6
B
Maximum % change in VAF in blood
P < .001
P = .032
P = .019
P = .146
n = 78 n = 38
n = 35 n = 12
n = 18 n = 8
n = 10 n = 6

(A and B) Analyses included patients in the ITT population with a detectable mutant allele for the indicated gene (≥5%) before treatment and ≥1 postbaseline mutation assessment. (A) Ratios underneath the bars represent the number of patients with ≥50% VAF reduction as numerator and the total number of patients with detectable assessment (≥5% VAF) in specified mutation at baseline and any postbaseline mutation assessment as denominator. P Value based on Cochran-Mantel-Haenszel test stratified for prior RBC transfusion burden (≤6 U or >6 U of RBC every 8 weeks) and baseline IPSS risk score (low or intermediate-1). (B) Comparison between each treatment group in the maximum percentage change from baseline in mutant VAF of the indicated gene. P Value based on the 2-sample t-test.

Figure 5. RBC-TI Responders in the Imetelstat Group Were Enriched in Patients Achieving ≥50% Reduction in *SF3B1* (A) and *TET2* (B) VAF

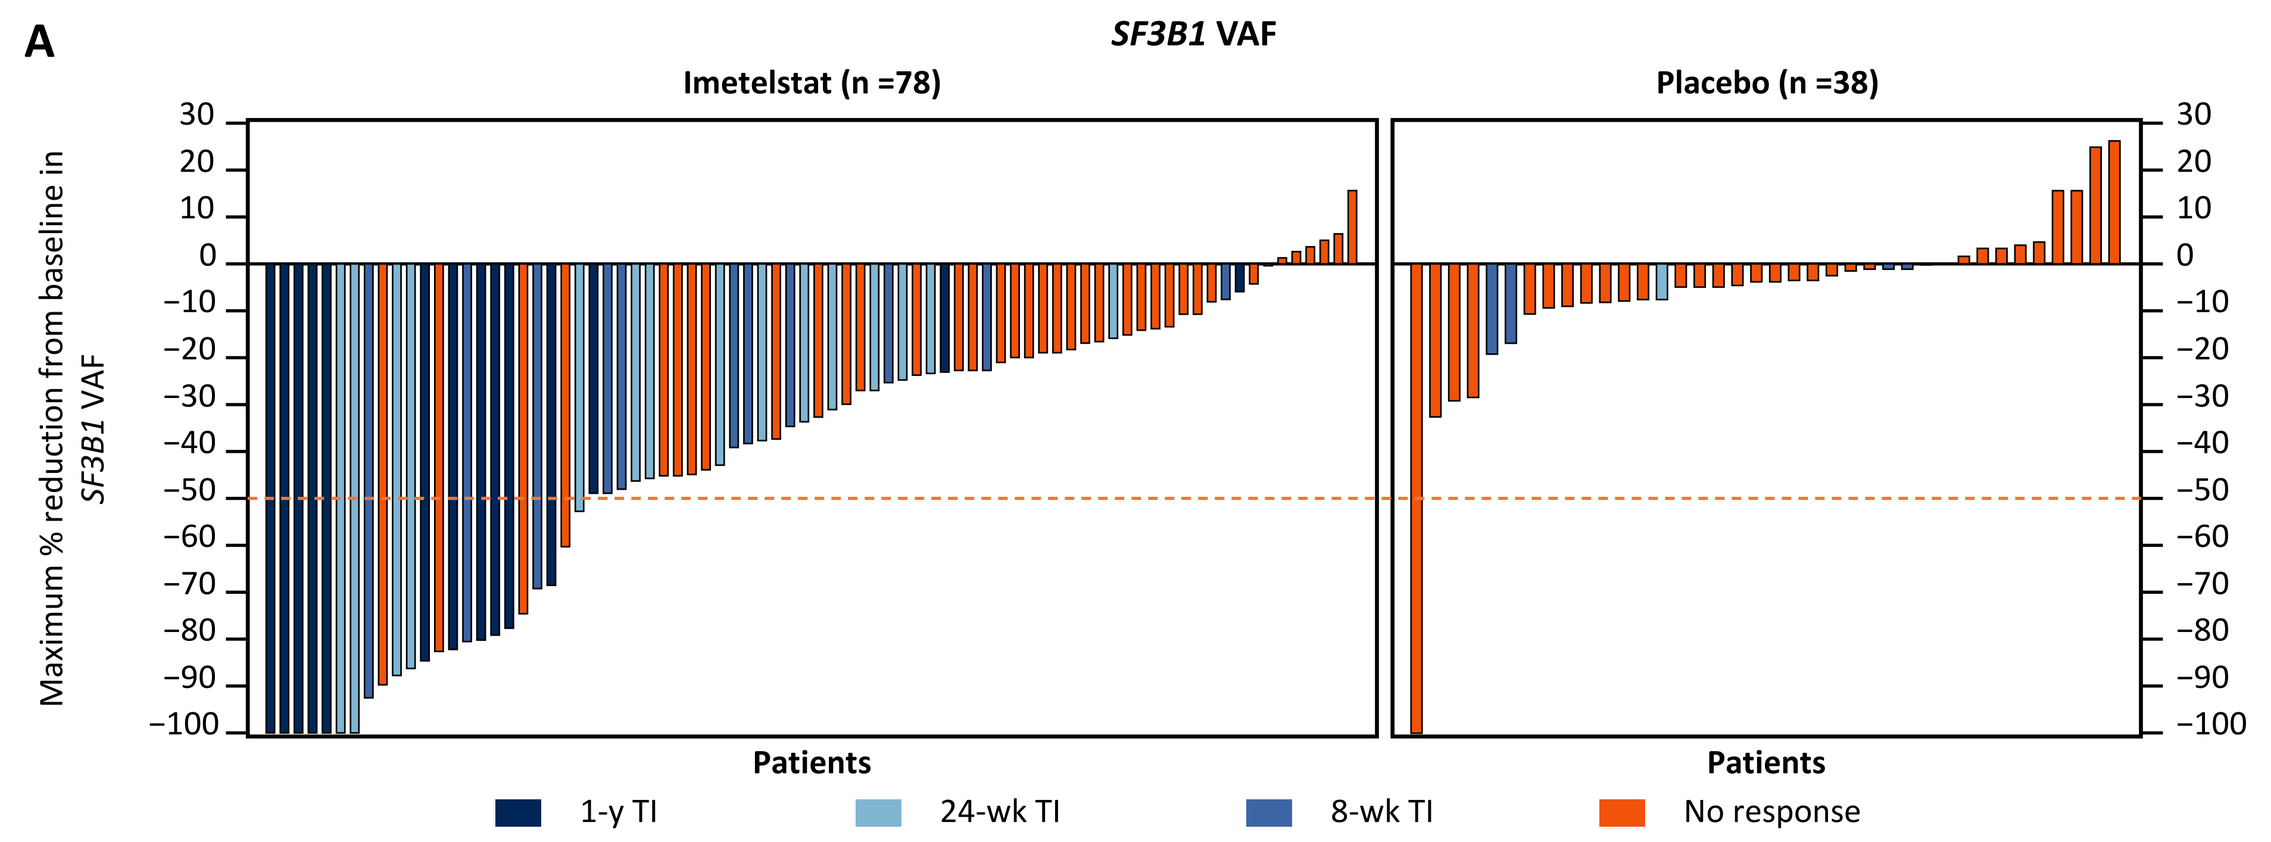
A
SF3B1 VAF
Imetelstat (n =78)
Placebo (n =38)
Maximum % reduction from baseline in SF3B1 VAF
Patients
1-y TI
24-wk TI
8-wk TI
No response

Patients, n (%)	*SF3B1* VAF ≥50% reduction with imetelstat			
	Yes (n = 23)	No (n = 55)	Total (N = 78)	*P* Value (Fisher exact test)
8-wk RBC-TI				
Yes	19 (82.6)	21 (38.2)	40 (51.3)	<.001
No	4 (17.4)	34 (61.8)	38 (48.7)	
24-wk RBC-TI				
Yes	16 (69.6)	13 (23.6)	29 (37.2)	<.001
No	7 (30.4)	42 (76.4)	49 (62.8)	
1-y RBC-TI				
Yes	11 (47.8)	3 (5.5)	14 (17.9)	<.001
No	12 (52.2)	52 (94.5)	64 (82.1)	

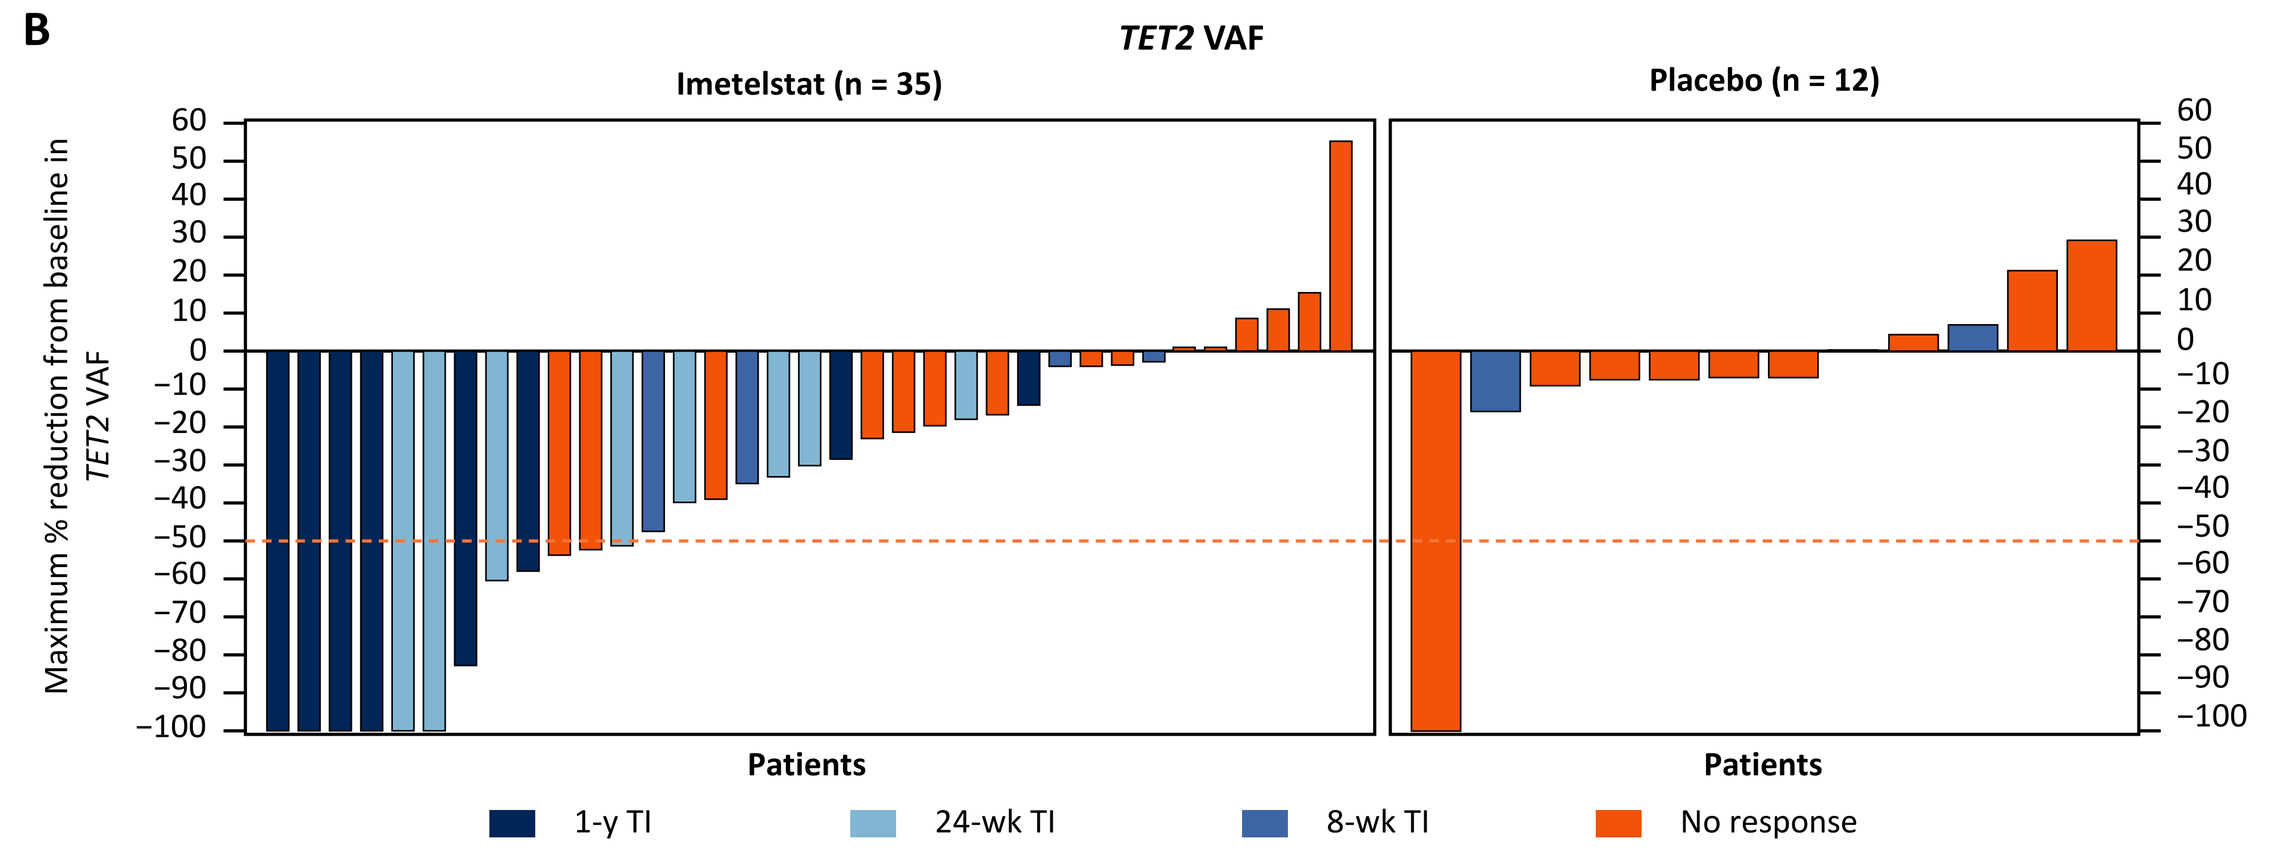
B
TET2 VAF
Imetelstat (n = 35)
Placebo (n = 12)
Maximum % reduction from baseline in TET2 VAF
Patients
1-y TI
24-wk TI
8-wk TI
No response

Patients, n (%)	*TET2* VAF ≥50% reduction with imetelstat			
	Yes (n = 12)	No (n = 23)	Total (N = 35)	*P* Value (Fisher exact test)
8-wk RBC-TI				
Yes	10 (83.3)	10 (43.5)	20 (57.1)	.034
No	2 (16.7)	13 (56.5)	15 (42.9)	
24-wk RBC-TI				
Yes	10 (83.3)	6 (26.1)	16 (45.7)	.003
No	2 (16.7)	17 (73.9)	19 (54.3)	
1-y RBC-TI				
Yes	6 (50.0)	2 (8.7)	8 (22.9)	.011
No	6 (50.0)	21 (91.3)	27 (77.1)	

Analyses included patients in the ITT population with a detectable SF3B1 mutant allele (≥5%) before treatment and ≥1 postbaseline mutation assessment.

Figure 6. VAF Reduction Correlated With Longer RBC-TI Duration (A) and Increased Hb Levels (B), in Patients Treated With Imetelstat

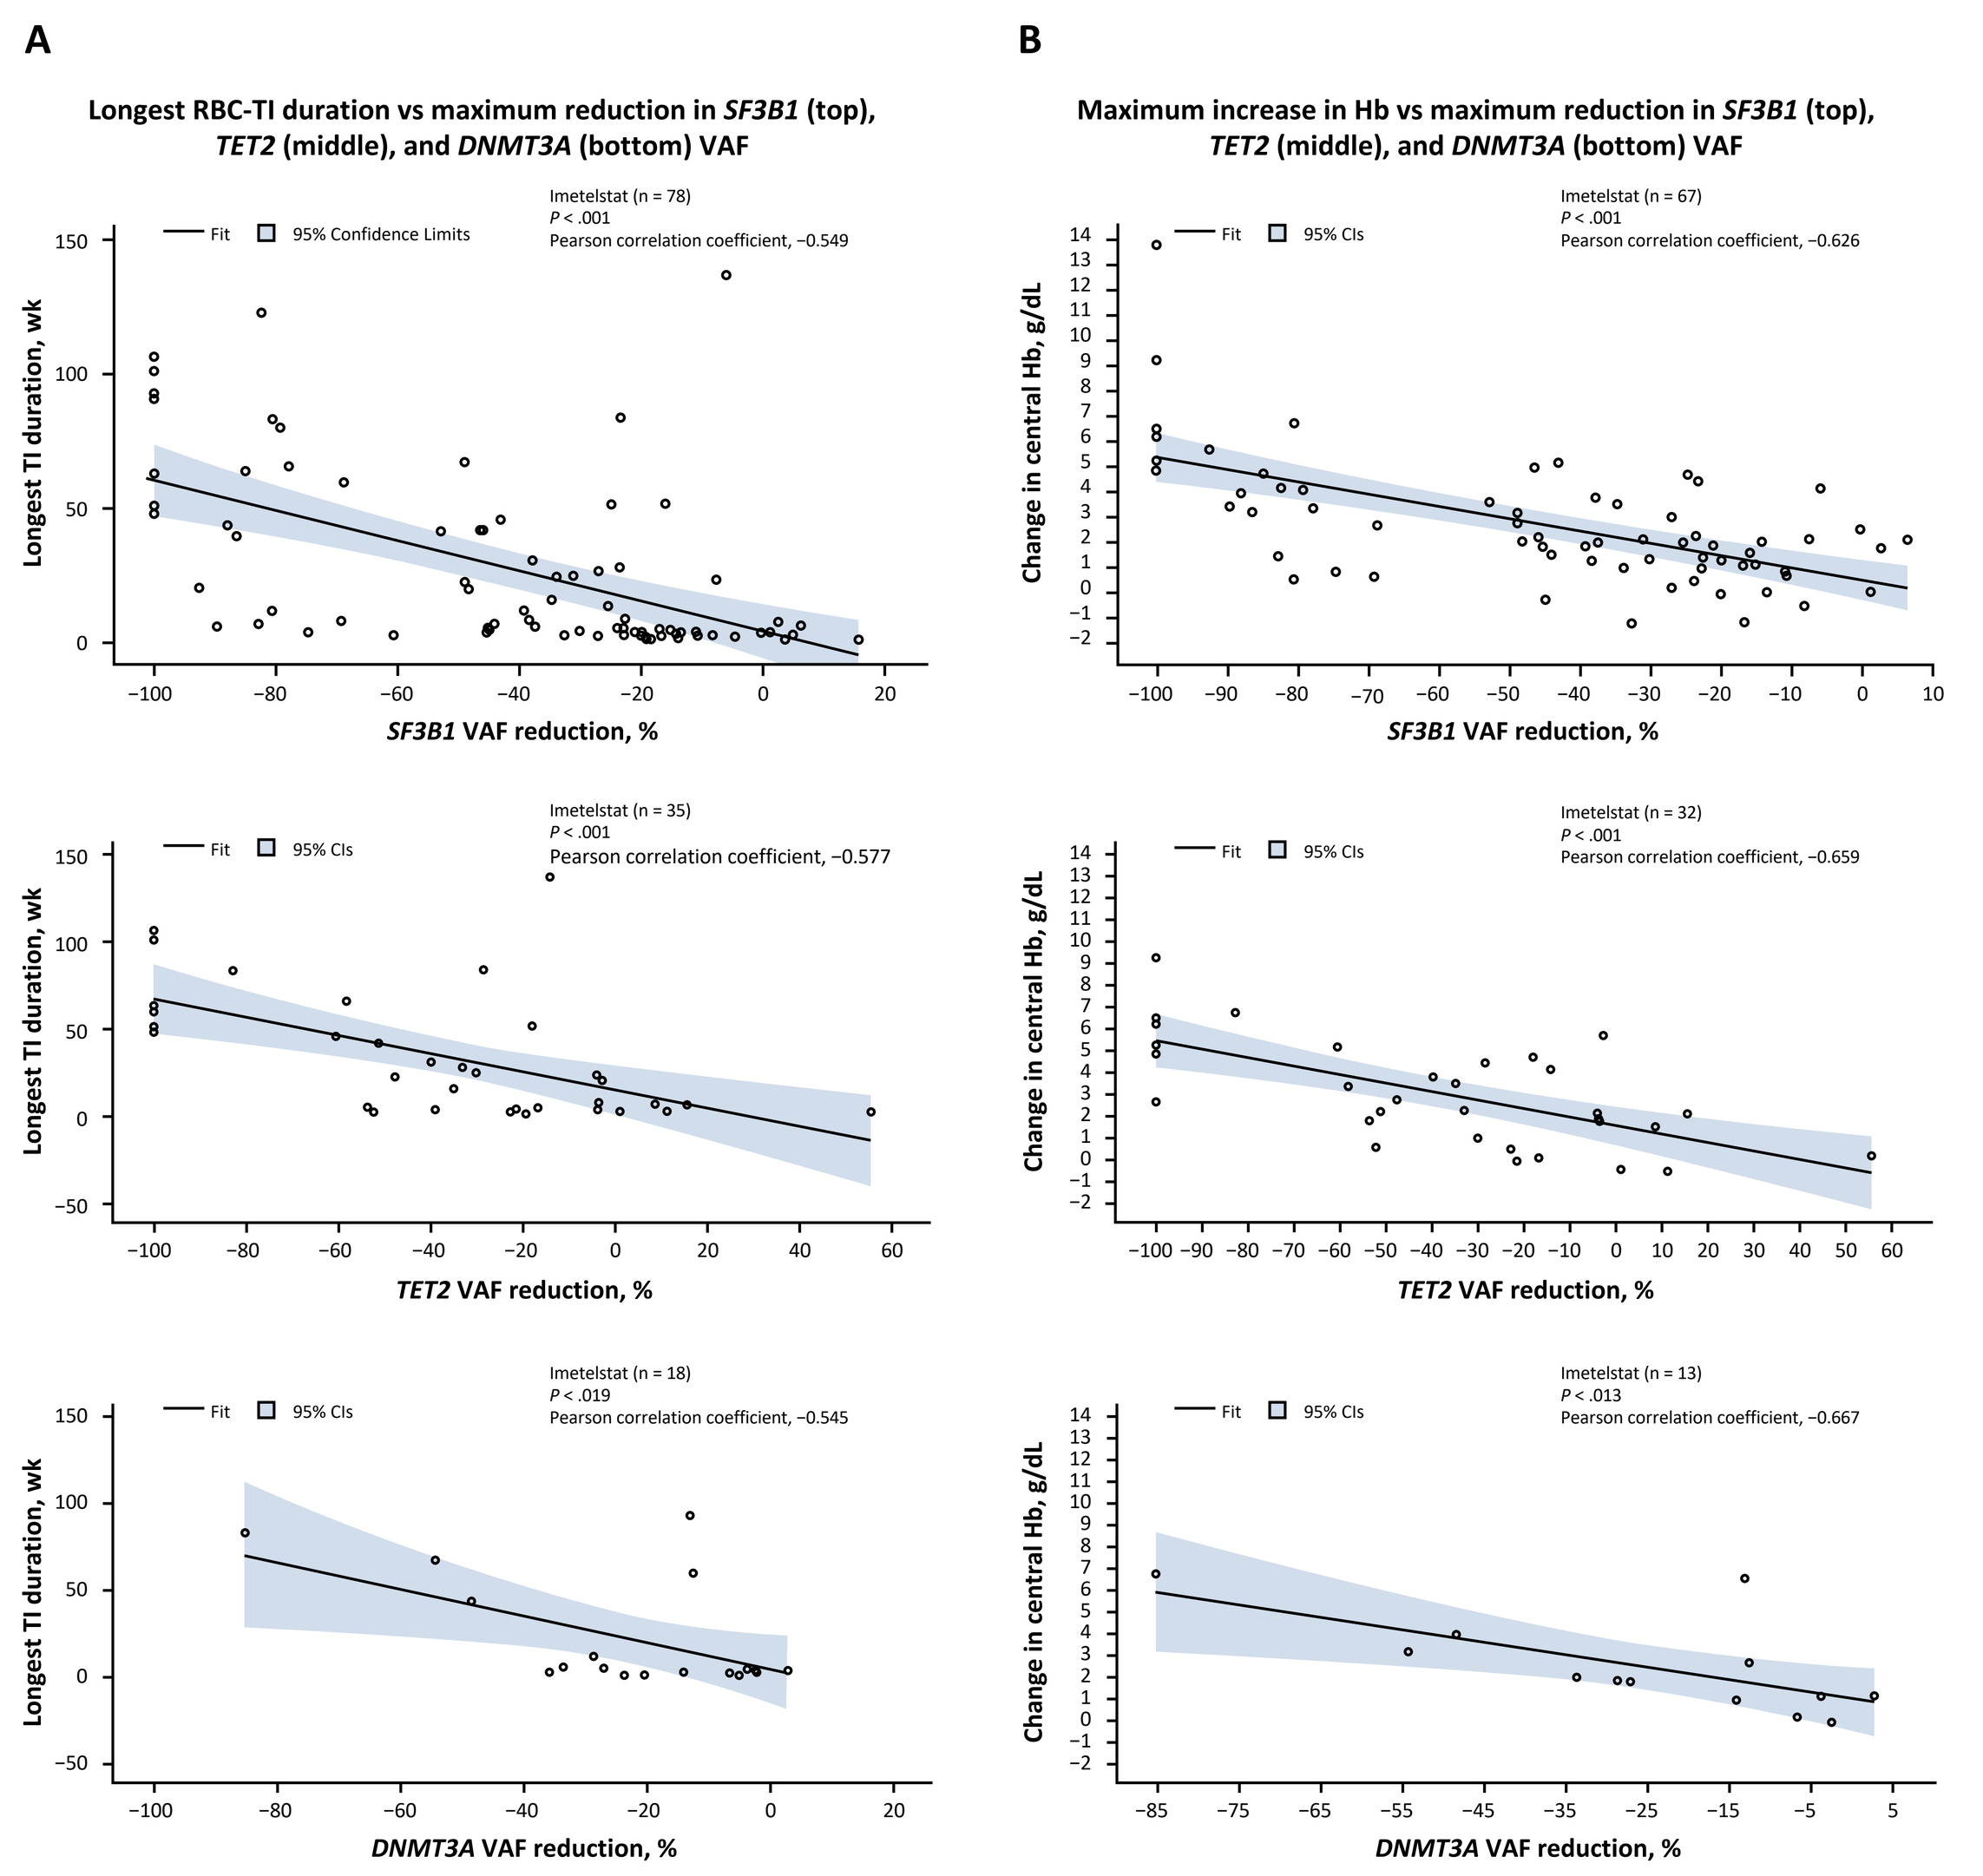
A
Longest RBC-TI duration vs maximum reduction in SF3B1 (top), TET2 (middle), and DNMT3A (bottom) VAF
Imetelstat (n = 78)
P < .001
Pearson correlation coefficient, −0.549
Imetelstat (n = 35)
P < .001
Pearson correlation coefficient, −0.577
Imetelstat (n = 18)
P < .019
Pearson correlation coefficient, −0.545
Longest TI duration, wk
SF3B1 VAF reduction, %
TET2 VAF reduction, %
DNMT3A VAF reduction, %
B
Maximum increase in Hb vs maximum reduction in SF3B1 (top), TET2 (middle), and DNMT3A (bottom) VAF
Imetelstat (n = 67)
P < .001
Pearson correlation coefficient, −0.626
Imetelstat (n = 32)
P < .001
Pearson correlation coefficient, −0.659
Imetelstat (n = 13)
P < .013
Pearson correlation coefficient, −0.667
Change in central Hb, g/dL
Fit
95% Confidence Limits
95% CIs

Analyses included patients in the ITT population with a detectable mutant allele for the indicated gene (≥5%) before treatment and ≥1 postbaseline mutation assessment (A, B) and had postbaseline Hb assessment, excluding assessments within 14 days post-RBC transfusion (B). Fitted lines and P value based on linear regression with maximum increase in RBC-TI duration (A) or maximum increase in Hb from pretreatment (B) as the dependent variable and the maximum percentage reduction from baseline in each gene VAF as independent variable. P Value calculated using Fisher exact test between "yes" vs "no" in each outcome.

Figure 7. Correlation of 8-Week and 24-Week RBC-TI With Reduction in RS+ Cells, Cytogenetic Responses, and VAF Reduction in Patients Treated With Imetelstat

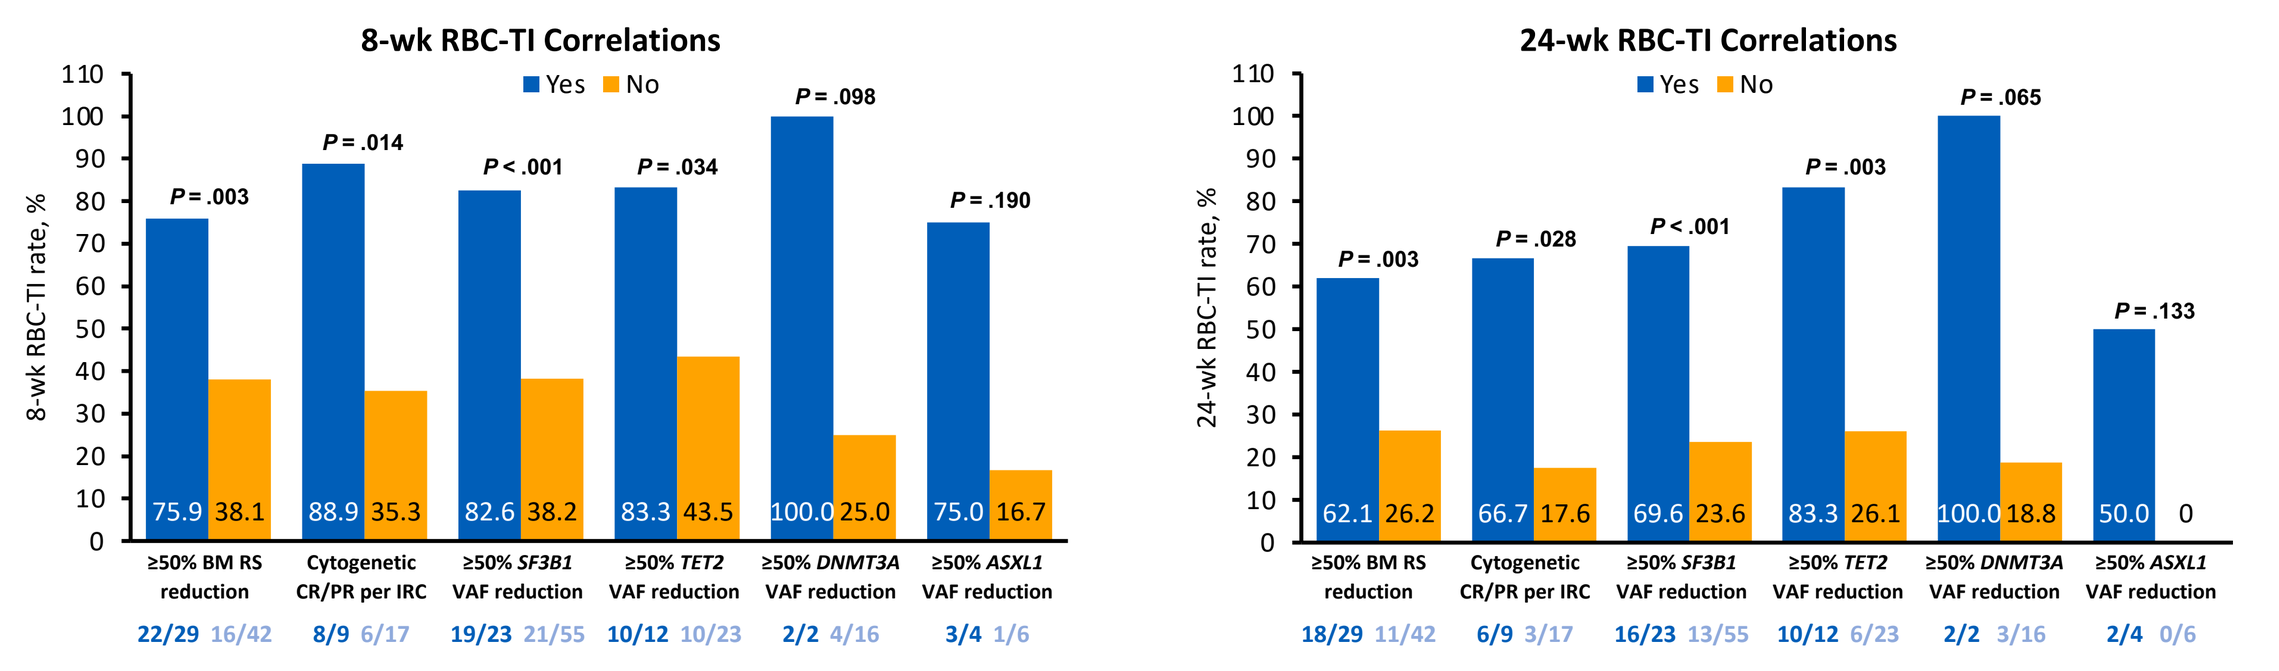
8-wk RBC-TI Correlations
Yes
No
8-wk RBC-TI rate, %
P = .003
P = .014
P < .001
P = .034
P = .098
P = .190
75.9 38.1
88.9 35.3
82.6 38.2
83.3 43.5
100.0 25.0
75.0 16.7
≥50% BM RS reduction
Cytogenetic CR/PR per IRC
≥50% SF3B1 VAF reduction
≥50% TET2 VAF reduction
≥50% DNMT3A VAF reduction
≥50% ASXL1 VAF reduction
22/29 16/42
8/9 6/17
19/23 21/55
10/12 10/23
2/2 4/16
3/4 1/6
24-wk RBC-TI Correlations
24-wk RBC-TI rate, %
P = .003
P = .028
P < .001
P = .003
P = .065
P = .133
62.1 26.2
66.7 17.6
69.6 23.6
83.3 26.1
100.0 18.8
50.0 0
18/29 11/42
6/9 3/17
16/23 13/55
10/12 6/23
2/2 3/16
2/4 0/6

CONCLUSIONS

- In this heavily transfusion dependent LR-MDS population treatment with imetelstat vs placebo led to:
 - Higher cytogenetic response rate
 - Higher percentage of patients achieving ≥50% reduction in BM RS cells (41% vs 10%)
 - Greater reduction of VAF in multiple genes, which correlated with clinical end points of TI response, longer RBC-TI duration, and increase in Hb levels
 - Cytogenetic response, RS BM reduction, and VAF reduction all correlated with 8-week and 24-week TI
- These data, along with the robust and sustained RBC-TI rates, suggest that imetelstat may alter the underlying biology of LR-MDS and can potentially modify the disease by reducing or eliminating malignant clones and improving ineffective erythropoiesis

ACKNOWLEDGMENTS

- Previously presented as an oral presentation at the European Hematology Association (EHA) 2023 congress
- The authors thank the patients and caregivers for their participation in this study and acknowledge the collaboration and commitment of the investigators and their research support staff
- All authors contributed to and approved the presentation
- Writing and editorial assistance was provided by Erin McMullin, PhD, Maleeha Fortuin-Seedat, PhD, and Mary C. Wiggin of Ashfield MedComms, an Inizio Company

DISCLOSURES

Valeria Santini served on advisory boards with AbbVie, BMS, CTI, Geron, Gilead, Novartis, Otsuka, Servier, and Syros and received a travel grant from Janssen.

CONTACT INFORMATION

IMerge (MDS3001): https://www.geron.com/patients/imerge-study
ClinicalTrials.gov: NCT02598661; email mds3001-info@geron.com

REFERENCES

1. Asai A, et al. *Cancer Res.* 2003;63(14):3931-3939.
2. Herbert B-S, et al. *Oncogene.* 2005;24(33):5262-5268.
3. Mosoyan G, et al. *Leukemia.* 2017;31(11):2458-2467.
4. Wang X, et al. *Blood Adv.* 2018;2(18):2378-2388.
5. Steensma DP, et al. *J Clin Oncol.* 2021;39(1):48-56.
6. Platzbecker U, et al. EHA 2023. Oral presentation S165.

ABBREVIATIONS

AE, adverse event; *ASXL1*, additional sex combs like-1; BM, bone marrow; CR, complete response; DCO, data cutoff; *DNMT3A*, DNA (cytosine-5)-methyltransferase 3A; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; FACIT, Functional Assessment of Chronic Illness Therapy; G-CSF, granulocyte-colony stimulating factor; Hb, hemoglobin; HMA, hypomethylating agent; IPSS, International Prognostic Scoring System; ITT, intent-to-treat; IV, intravenous; LR, lower risk; MDS, myelodysplastic neoplasms; NGS, next-generation sequencing; NS, not significant; PR, partial response; PRO, patient-reported outcome; R, randomization; RBC, red blood cell; R/R, relapsed/refractory; RS, ring sideroblast; sEPO, serum erythropoietin; *SF3B1*, splicing factor 3b subunit 1; *TET2*, Tet methylcytosine dioxygenase 2; TI, transfusion independence; VAF, variant allele frequency.