PII-050

Clinical Pharmacology of Imetelstat, A First-in-Class Oligonucleotide Telomerase Inhibitor

Ashley L Lennox[1], Fei Huang[1], Melissa Kelly Behrs[1], Chandra Pamulapati[1], Mario González-Sales[2], Ying Wan[1], Libo Sun[1], Tymara Berry[1], Faye Feller[1], Peter N Morcos[3]

[1]Geron Corporation, Parsippany, New Jersey, USA; [2]Modeling Great Solutions Pharmaceutical Research & Studies, FZE, Dubai, UAE;
[3]Morcos Pharmaceutical Consulting, LLC, Marlboro, New Jersey, USA

INTRODUCTION

- Imetelstat is a first-in-class telomerase inhibitor in development for the treatment of myeloid malignancies, including treatment of transfusion-dependent anemia in lower-risk myelodysplastic syndromes (LR-MDS).
- Imetelstat is a 13-mer N3'→P5' thio-phosphoramidate oligonucleotide with a lipid tail to enhance cellular uptake[1-2]. It has a sequence complementary to and specifically binds with high affinity to the template region of the RNA component of human telomerase **(Figure 1)**.
- Imetelstat and other oligonucleotide classes have structural similarities; however, the mechanism of action for imetelstat is not antisense based, as it does not target mRNA of any gene. Instead, it acts as a classical active site enzyme inhibitor.
- Telomerase inhibition by imetelstat leads to loss of a malignant cell's ability to maintain telomere length, resulting in inhibition of cell proliferation and death of malignant cells[3-11].
- Nonclinical proof-of-concept studies for imetelstat correlated pharmacokinetic (PK) exposure, pharmacodynamic effect (target engagement by inhibition of telomerase activity) and tumor growth inhibition *in vivo* in xenograft mouse models and indicated that higher imetelstat doses were associated with greater plasma exposure and target engagement[12-13].
- The IMerge Phase 3 study (NCT02598661) established the benefit/risk of imetelstat 7.5 mg/kg q4w in LR-MDS, demonstrating a statistically significant and clinically meaningful improvement in red blood cell transfusion independence over placebo and a manageable safety profile[14].
- The clinical benefit of imetelstat in myelofibrosis (MF) was demonstrated in the IMbark Phase 2 randomized dose-finding study (NCT02426086)[15]. The confirmatory IMpactMF Phase 3 study (NCT04576156) is ongoing to evaluate a potential improvement in overall survival for imetelstat compared to best available therapy in patients with MF[16].

Figure 1. Imetelstat Mechanism of Action

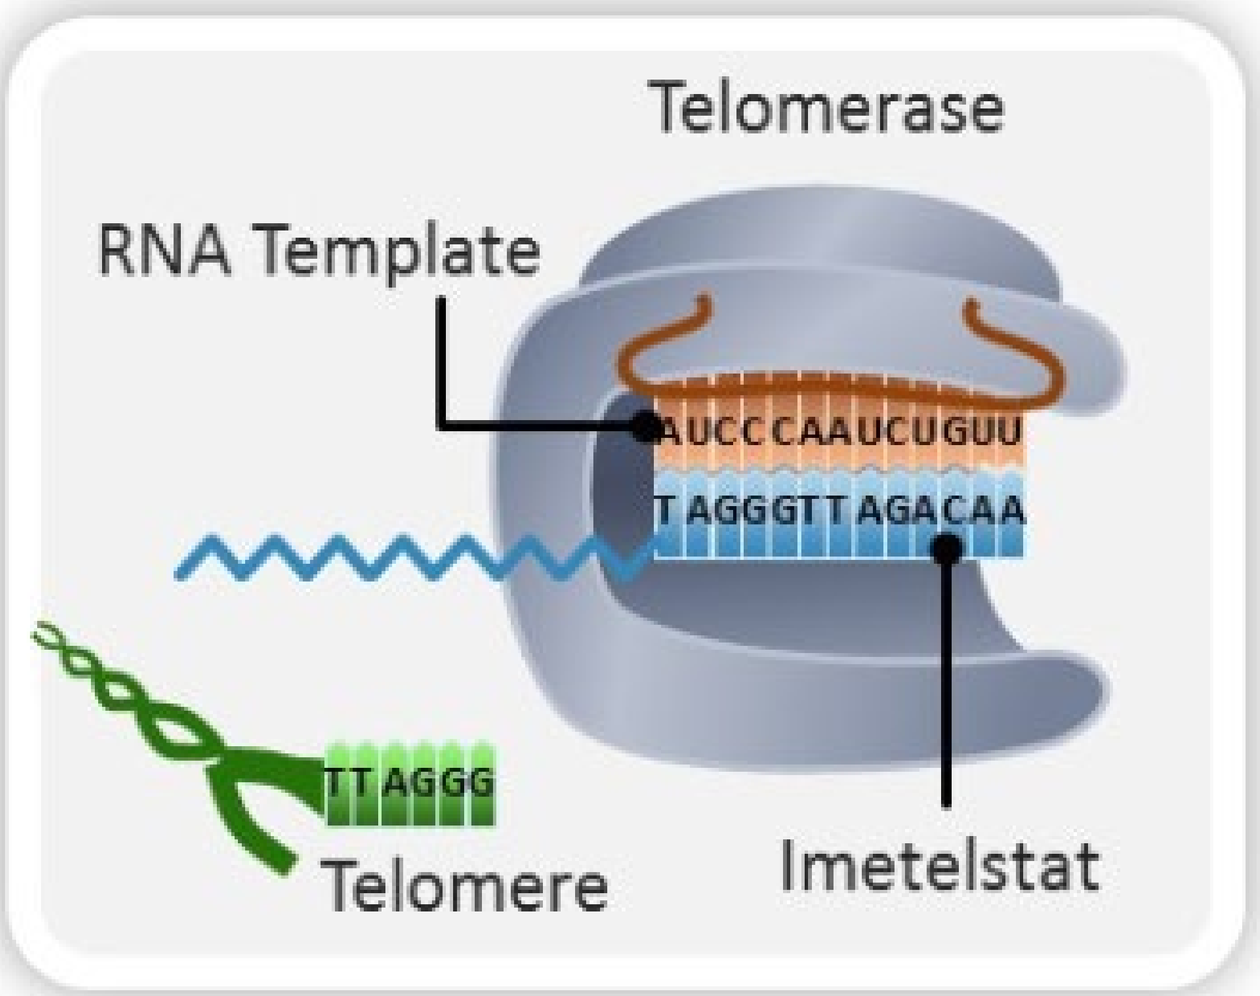
Telomerase
RNA Template
AUCCCAAUCUGUU
TAGGGTTAGACAA
TTAGGG
Telomere
Imetelstat

OBJECTIVE

Evaluate the clinical pharmacology properties of imetelstat to support its optimal use to treat transfusion-dependent anemia in patients with LR-MDS at the proposed clinical dose of 7.5 mg/kg* every-4 weeks (q4w) administered over 2 h IV infusion.

METHODS

- The imetelstat clinical pharmacology program consists of clinical trials in patients with solid tumor or hematologic malignancies across a wide range of doses (0.4 to 11.7 mg/kg) and schedules (qw to q4w interval) and supportive nonclinical studies in human biomaterials and animals.
- Evaluations included characterization of single and multiple-dose PK, ADME properties, effect of intrinsic factors on imetelstat PK, drug-drug interaction (DDI) liability, proarrhythmic potential, and immunogenicity.
- Imetelstat PK properties and sources of PK variability were evaluated with noncompartmental and population PK (popPK) analysis from 7 pooled studies **(González Sales ASCPT 2024, PII-128)**.

RESULTS

ADME Properties

- AUC increases in a more than dose proportional manner over the 0.4-11.7 mg/kg range.
- Little to no accumulation is observed following repeat doses **(Figure 2)**.
- Imetelstat has moderate variability, with geometric mean (CV%) C_{max} of 89.2 µg/mL (27.2%) and $AUC_{0\text{-}28d}$ of 554 h·µg/mL (43.4%) in MDS following a 7.5 mg/kg dose.

Absorption

- Imetelstat is administered via IV infusion, and T_{max} is observed at the end of infusion **(Figure 2)**.

Distribution

- Human plasma protein binding is 94% *in vitro*.
- Nonclinical ADME studies demonstrate rapid distribution of imetelstat from plasma to tissue, with highest concentrations in bone marrow (site of action for myeloid diseases), and liver, kidney, and spleen.

Metabolism

- Like other oligonucleotides, imetelstat is likely metabolized into smaller fragments by nucleases in tissues and the component fragments are excreted in urine[17-21].
- Hepatic cytochrome p450 (CYP) enzymes are not expected to metabolize imetelstat, as reported for other oligonucleotides[19, 22].

Elimination

- The apparent t½ of imetelstat in plasma following a 7.5 mg/kg dose in LR-MDS is 4.9 h, which likely reflects a distributional t½.
- In nonclinical ADME studies, urine is the primary route of elimination of radioactive imetelstat, with ~62 to 82% recovery in urine and ~12 to 21% recovery in feces after 168 h. Unchanged imetelstat is not detected in urine.

Figure 2. Imetelstat Plasma PK Profile and EOI Concentrations

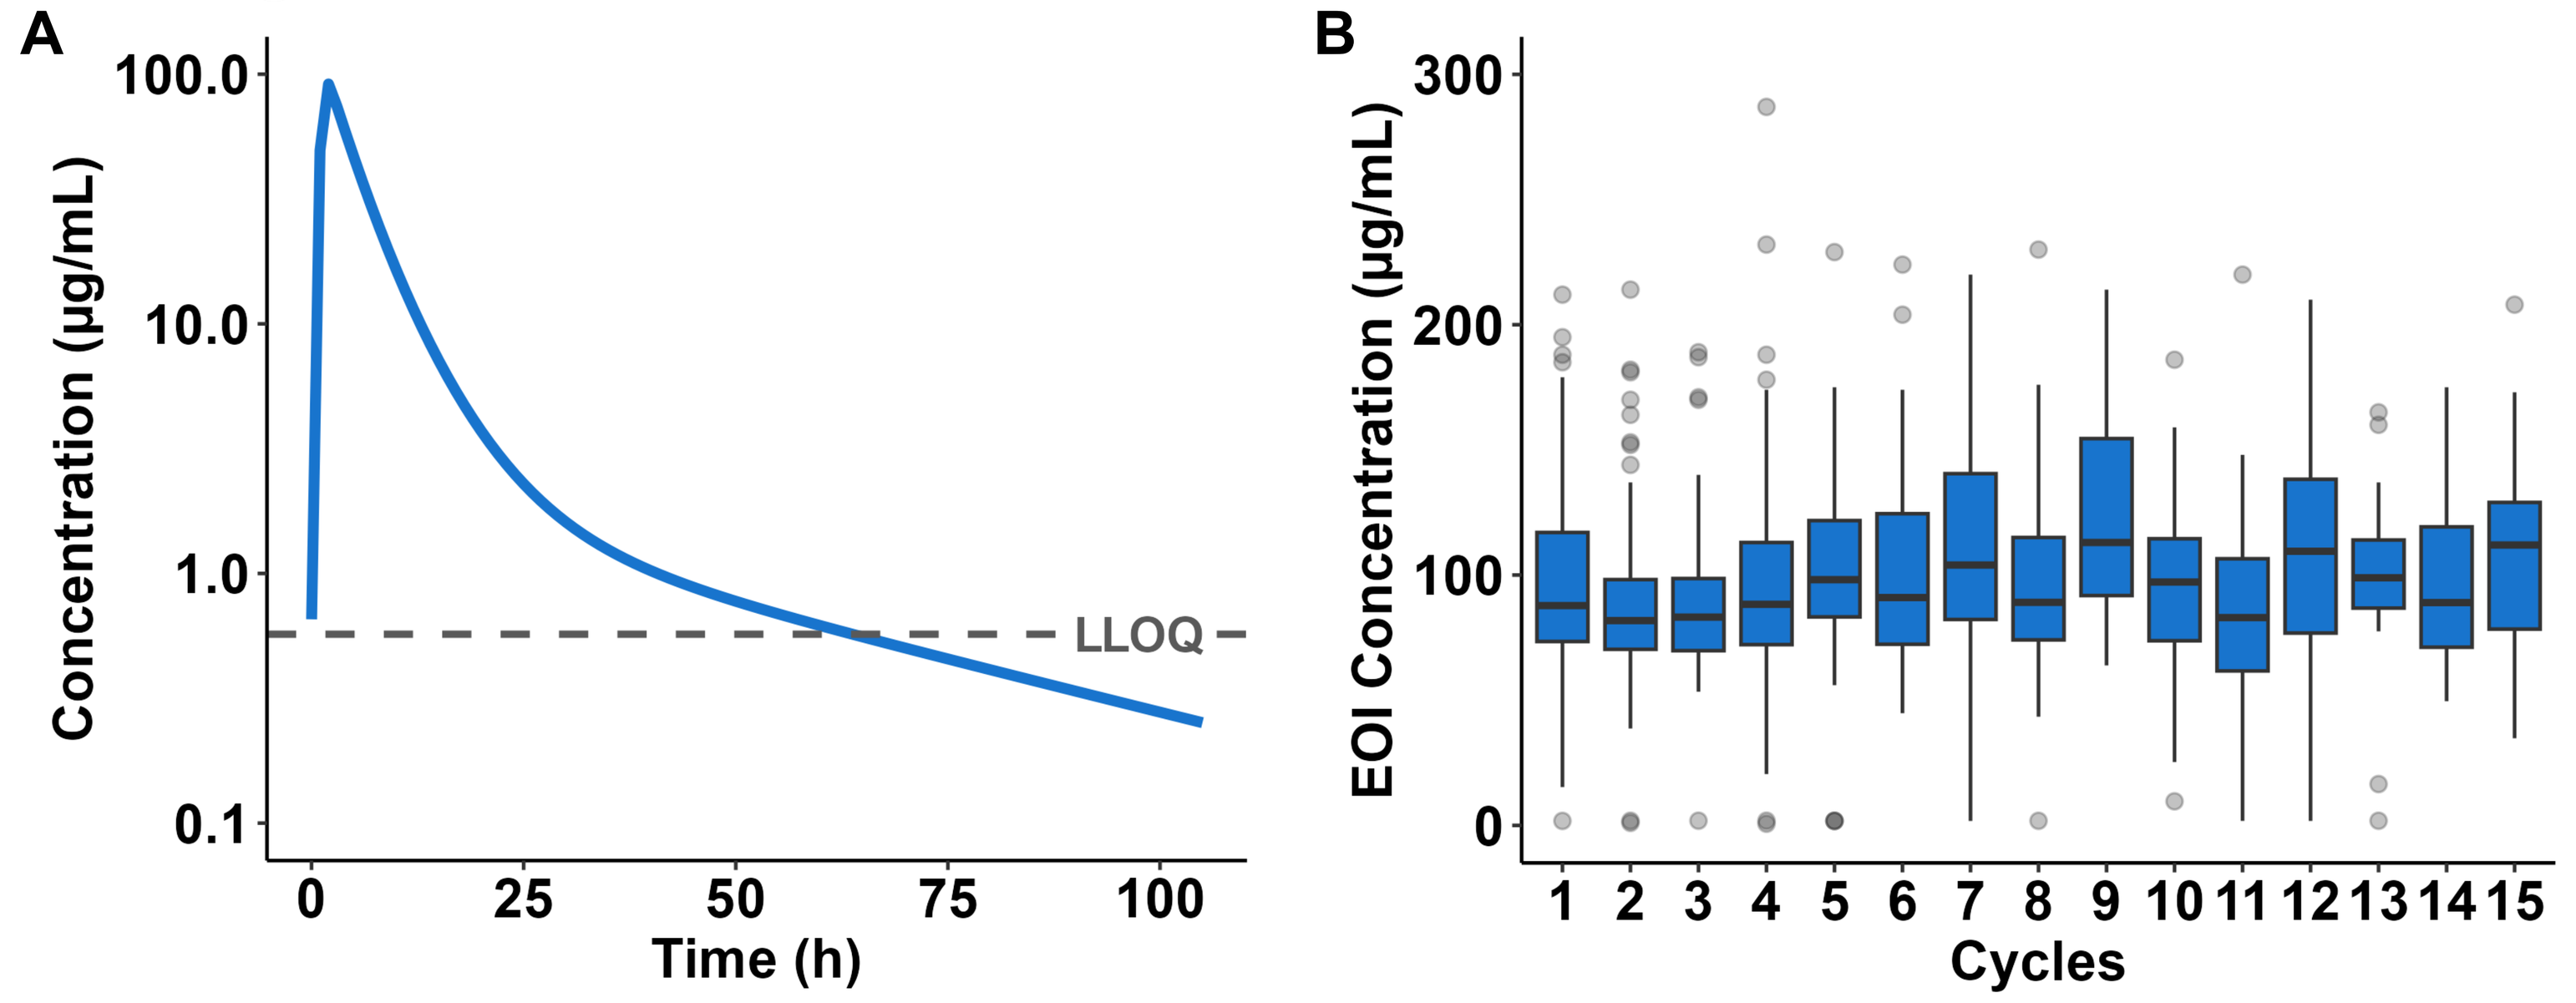
A
Concentration (µg/mL)
100.0
10.0
1.0
0.1
LLOQ
0
25
50
75
100
Time (h)
B
EOI Concentration (µg/mL)
300
200
100
0
1 2 3 4 5 6 7 8 9 10 11 12 13 14 15
Cycles

A) Model-predicted Cycle 1 concentration-time profile (N=170) and **B)** observed end of infusion concentrations (N=171) across cycles for patients with LR-MDS following 7.5 mg/kg q4w dosing.

Intrinsic Factors Influencing PK

- **Sex**: Females have lower clearance; given the limited impact on exposure (<30% increase) that is within the observed variation, dose adjustments are not warranted **(Figure 3)**.
- **Body Weight:** Theory-based allometric exponents for body weight were included on disposition parameters in popPK, supporting use of the body-weight based dosing regimen **(Figure 3).**
- **Organ Impairment:** No effect of mild to moderate hepatic or renal impairment, and limited data available on severe impairment **(Figure 3)**.
- **Age and Race:** No effect of age or race.

Figure 3. Box Plots of AUC by Intrinsic Factors in MDS

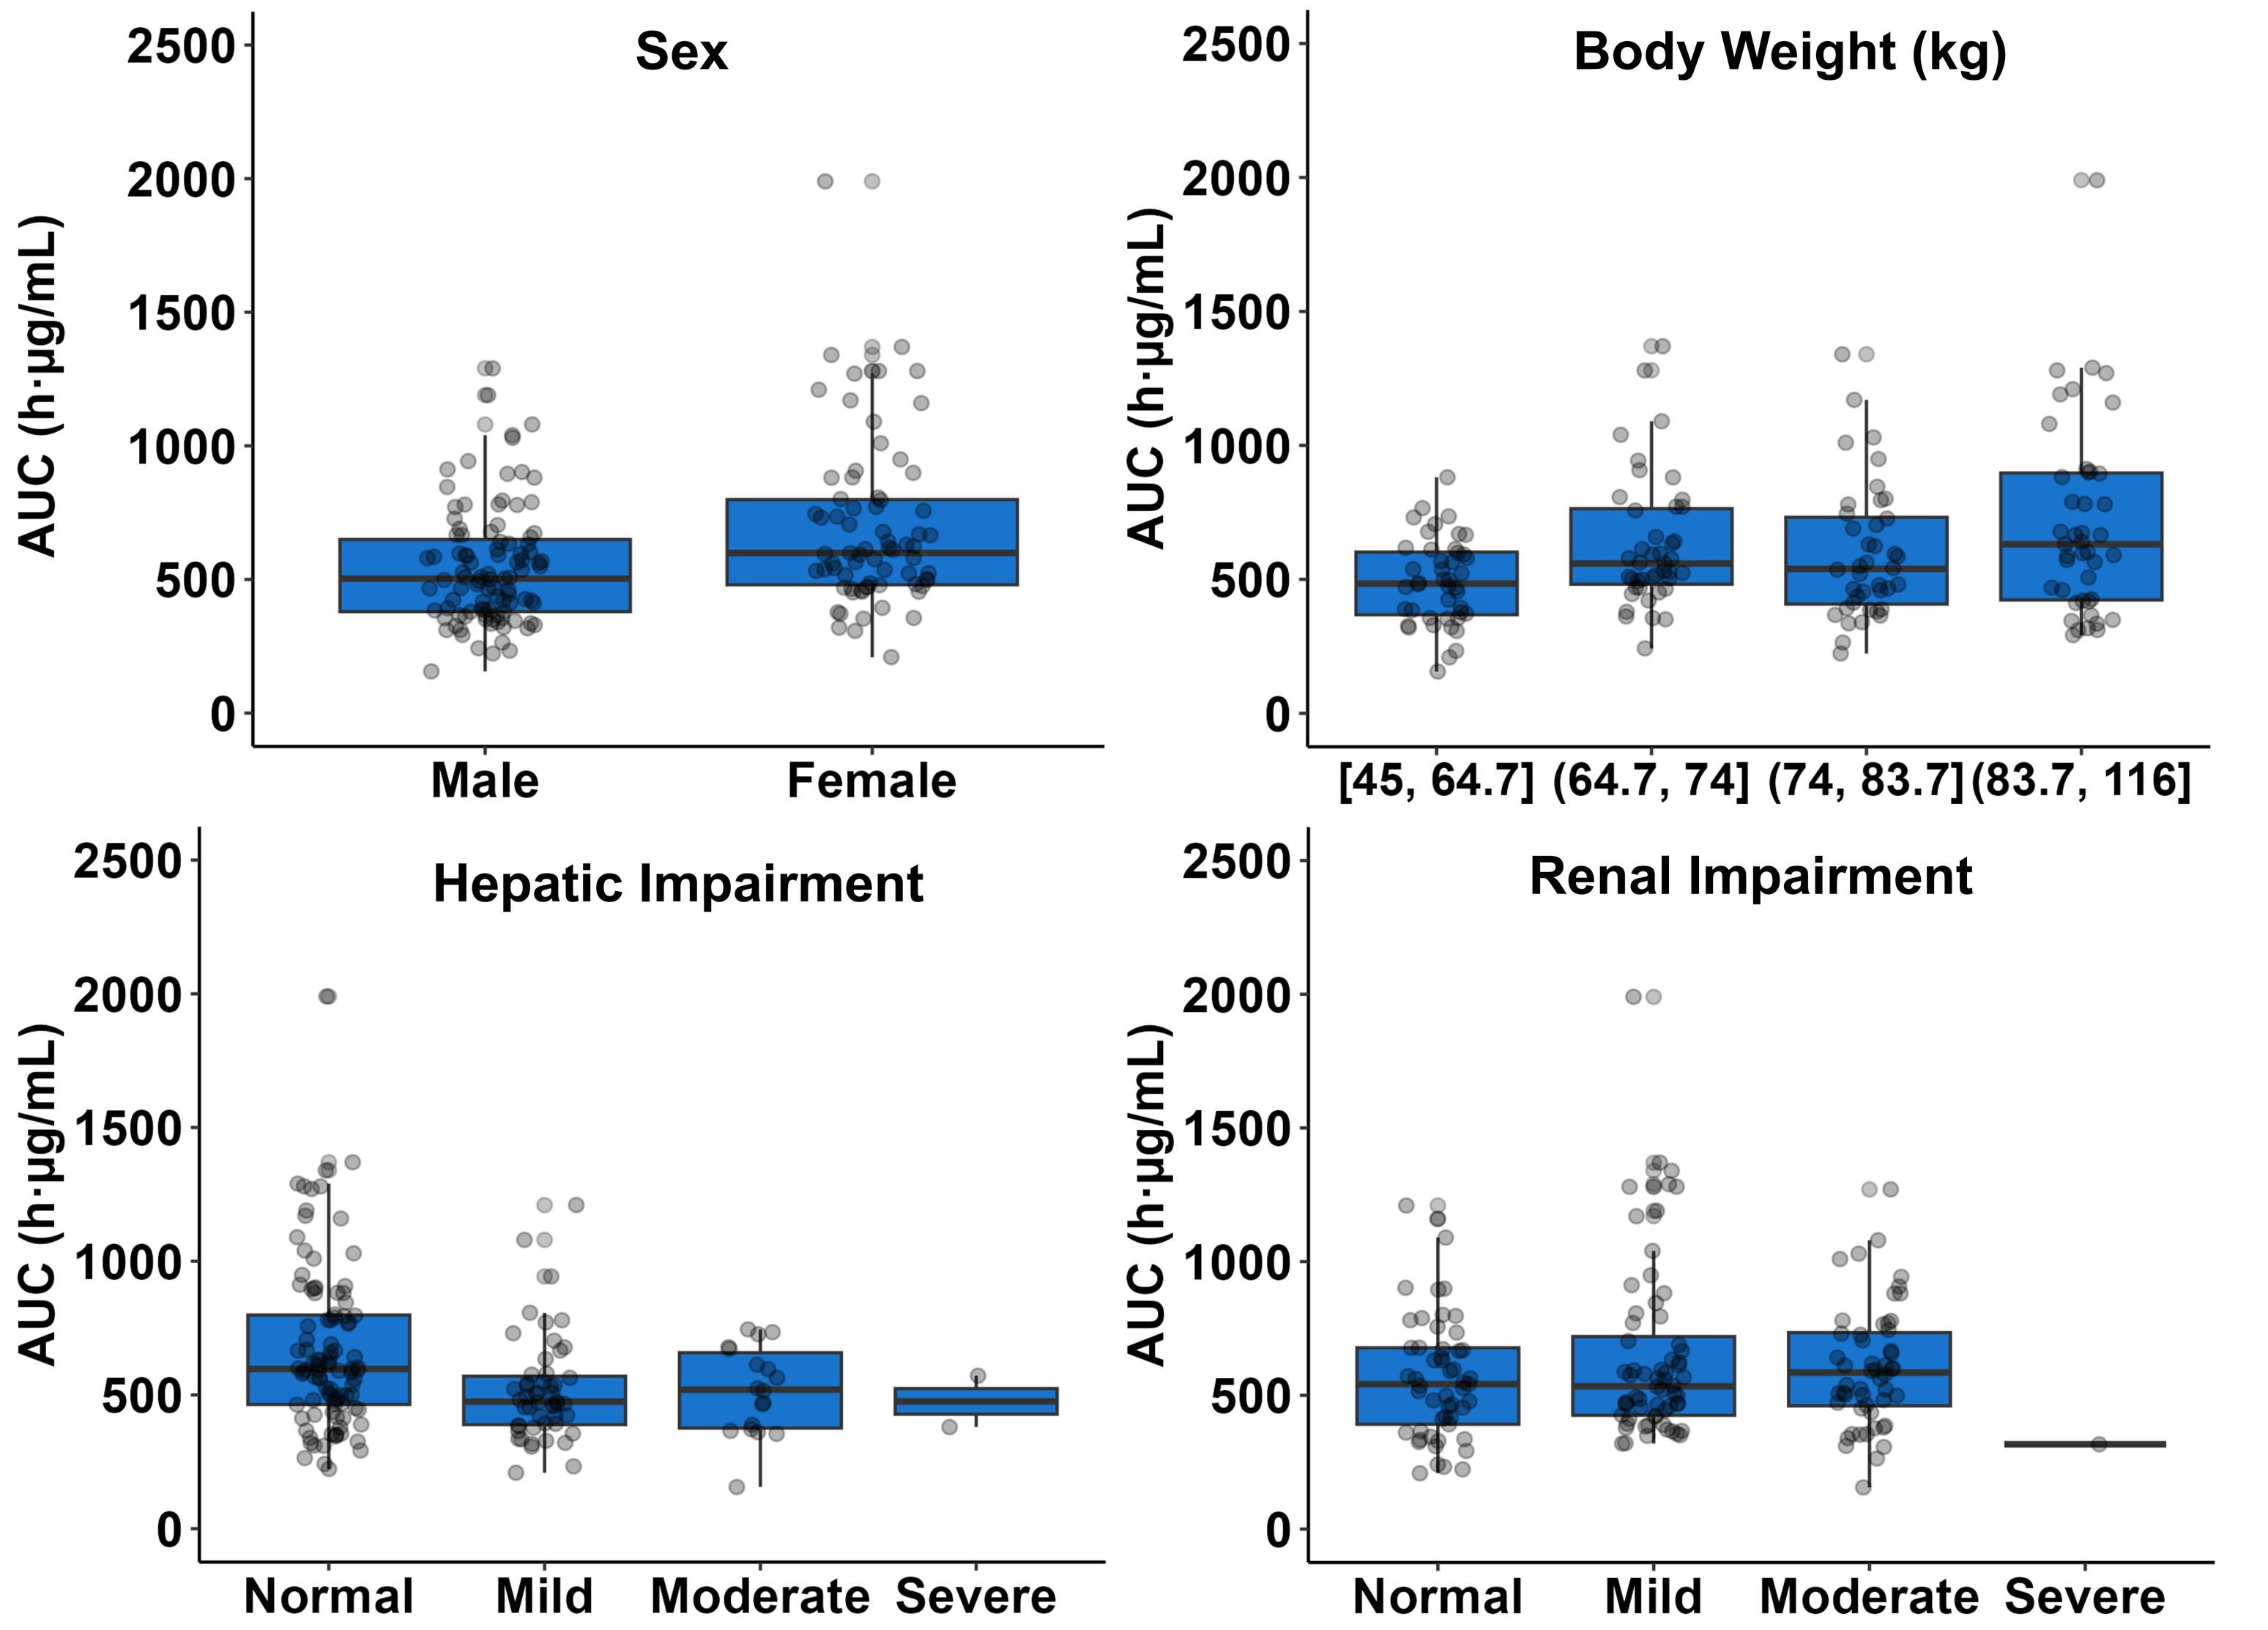
Sex
AUC (h·µg/mL)
2500
2000
1500
1000
500
0
Male
Female
Body Weight (kg)
[45, 64.7]
(64.7, 74]
(74, 83.7]
(83.7, 116]
Hepatic Impairment
Normal
Mild
Moderate
Severe
Renal Impairment
Normal
Mild
Moderate
Severe

Model-predicted Cycle 1 AUC for patients with LR-MDS following a 7.5 mg/kg dose (N=170).

Drug-Drug Interactions

Effect of Other Drugs on Imetelstat: Like other oligonucleotides[19,22], imetelstat is unlikely to be the victim of DDIs mediated by CYP enzymes or drug transporters.

Effect of Imetelstat on Other Drugs:

- *In vitro* studies suggest that imetelstat is an inhibitor of OATP1B1, OATP1B3, OAT1, and UGT1A1.
- Considering imetelstat's short plasma t½, lack of accumulation, and q4w dosing, clinically relevant drug interactions with substrates of these transporters and enzyme are unlikely.

Proarrhythmic Potential

- Imetelstat does not inhibit the hERG channel at concentrations up to 140x unbound C_{max} *in vitro*.
- No treatment-related clinical signs or effects on CNS and cardiac parameters were found in monkeys at >2.6x human C_{max}.
- No indication of cardiac toxicity in imetelstat-treated patients.
- Concentration-QT analysis is ongoing with data from clinical QT substudy in LR-MDS patients.

Immunogenicity

- Anti-drug antibodies (ADA) developed at a low frequency in patients with LR-MDS (17%) in the IMerge study and MF (21%) in the IMbark study.
- No effect of ADA was found on imetelstat PK, safety, or efficacy in LR-MDS.

CONCLUSIONS

- Imetelstat disposition properties were well characterized in support of the 7.5 mg/kg q4w dosing regimen in LR-MDS.
- No dose adjustments are warranted for sex, mild to moderate renal or hepatic impairment, age, or race.
- Body-weight based dosing is supported by the popPK analysis.
- There are no clinically relevant drug-drug interactions for imetelstat.
- Based on integrated nonclinical and clinical assessments, an imetelstat effect on QT prolongation is unlikely.
- ADA develop at low frequency and do not affect the benefit/risk profile in LR-MDS.

REFERENCES

1. Asai A et al. *Cancer Res.* 2003 63(14), 3931-3939.
2. Herbert B et al. *Oncogene.* 2005 24(33), 5262-5268.
3. Baerlocher G et al. *Blood Adv.* 2019 3(22), 3724-3728.
4. Brennan S et al. *PLoS One.* 2010 5(9).
5. Bruedigam C et al. *Cell Stem Cell.* 2014 15(6), 775-790.
6. Dikmen Z et al. *Cancer Res.* 2005 65(17), 7866-7873.
7. Ma W et al. *Blood.* 2017 130, 2860.
8. Marian C et al. *Clin Cancer Res.* 2010 16(1), 154-163.
9. Mosoyan G et al. *Leukemia.* 2017 31(11), 2458-2467.
10. Shammas M et al. *Leukemia.* 2008 22(7), 1410-1418.
11. Wang X et al. *Blood Adv.* 2018 2(18), 2378-2388.
12. Bassett E et al. *AACR Annual Meeting.* 2008.
13. Go N et al. *AACR Annual Meeting.* 2010.
14. Platzbecker U et al. *Lancet.* 2024 Jan 20;403(10423):249-260
15. Mascarenhas J et al. *J Clin Oncol.* 2021 Sep 10; 39(26):2881-2892.
16. Mascarenhas J et al. *Blood.* 2022 140 (Supplement 1):6826-6829.
17. Crooke R et al. *J Pharmacol Exp Ther.* 2000. 292(1), 140-149.
18. Gaus H et al. *Anal Chem.* 1997. 69(3), 313-319.
19. Geary R et al. *Expert Opin Drug Metab Toxicol.* 2009. 5(4), 381-391
20. Griffey R et al. *J Mass Spectrom.* 1997. 32(3), 305-313.
21. Yu R et al. *Drug Metab Dispos.* 2007. 35(3), 460-468.
22. FDA. 2022. https://www.fda.gov/media/159414/download

*Imetelstat dose level expressed in terms of imetelstat sodium (7.5 mg/kg imetelstat sodium = 7.1 mg/kg imetelstat active moiety).

Presented at the American Society for Clinical Pharmacology and Therapeutics Annual Meeting, March 27-29, 2024, Colorado Springs, CO, USA

Presenting Author: Ashley L Lennox