Results From the Phase 3 Trial of Imetelstat, a First-in-Class Telomerase Inhibitor, in Patients With Red Blood Cell Transfusion-Dependent Non-del(5q) Lower-Risk Myelodysplastic Syndromes Relapsed/Refractory to/Ineligible for Erythropoiesis Stimulating Agents

Yazan F. Madanat,[1] Michael R. Savona,[2] Mikkael A. Sekeres,[3] Uwe Platzbecker,[4] Valeria Santini,[5] Pierre Fenaux,[6] Maria Diez-Campelo,[7] David Valcárcel,[8] Tymara Berry,[9] Souria Dougherty,[9] Sheetal Shah,[9] Libo Sun,[9] Ying Wan,[9] Fei Huang,[9] Shyamala Navada,[9] Rami S. Komrokji,[10] Amer M. Zeidan[11]

[1]Simmons Comprehensive Cancer Center, UT Southwestern Medical Center, Dallas, TX, USA; [2]Vanderbilt-Ingram Cancer Center, Vanderbilt University Medical Center, Nashville, TN, USA; [3]Division of Hematology, Sylvester Comprehensive Cancer Center, University of Miami, Miami, FL, USA; [4]Department of Hematology, Cellular Therapy and Hemostaseology, Leipzig University Hospital, Leipzig, Germany; [5]MDS Unit, Hematology, AOUC, University of Florence, Florence, Italy; [6]Hôpital Saint-Louis, Université de Paris 7, Paris, France; [7]Hematology Department, The University Hospital of Salamanca, Salamanca, Spain; [8]Hematology Department, Hospital Universitari Vall d'Hebron, Barcelona, Spain; [9]Geron Corporation, Parsippany, NJ, USA; [10]Moffitt Cancer Center, Tampa, FL, USA; [11]Department of Internal Medicine, Yale School of Medicine and Yale Cancer Center, Yale University, New Haven, CT, USA

Introduction

- An unmet need exists for patients with LR-MDS who are RBC transfusion dependent and relapsed/refractory to or ineligible for ESAs
- Imetelstat is a first-in-class direct and competitive inhibitor of telomerase activity that specifically targets dysplastic clones with abnormally high telomerase activity, enabling recovery of effective hematopoiesis (**Fig. 1**)[1-4]

Figure 1. Mechanism of Action of Imetelstat

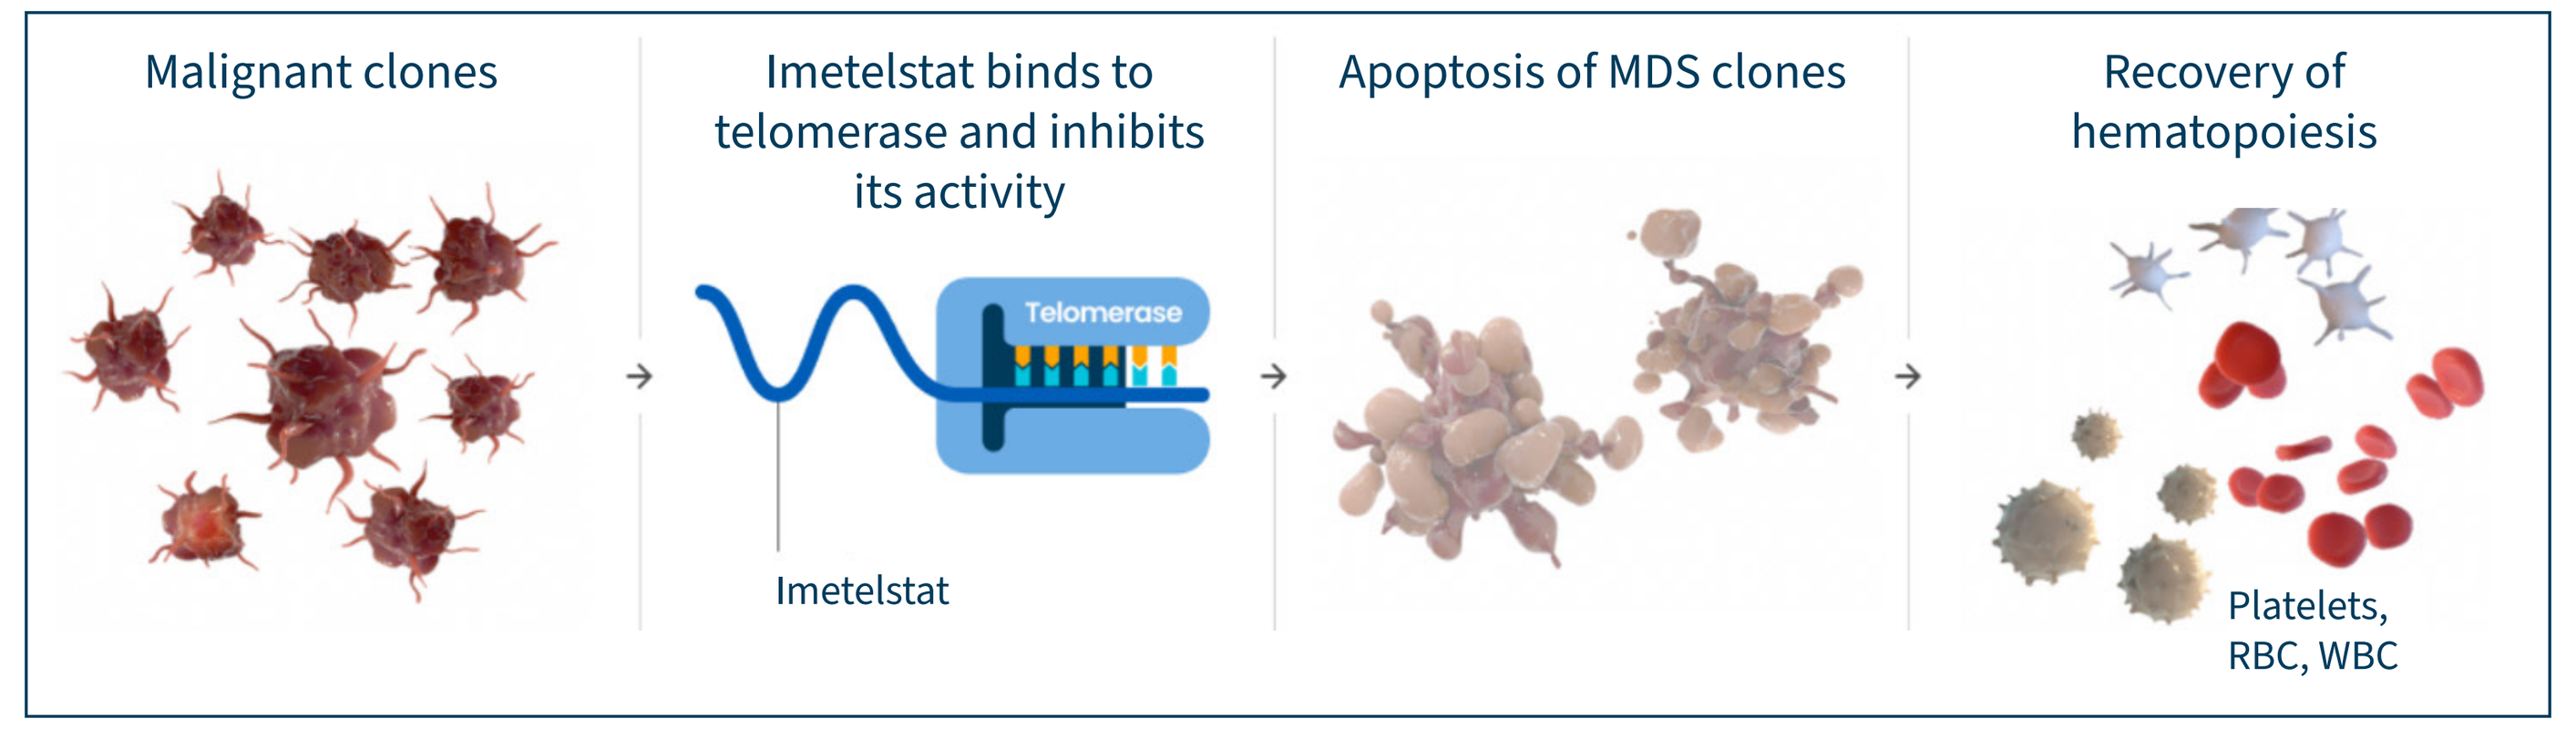
Malignant clones
Imetelstat binds to telomerase and inhibits its activity
Telomerase
Imetelstat
Apoptosis of MDS clones
Recovery of hematopoiesis
Platelets, RBC, WBC

Methods

Study Design

- IMerge phase 3 is a double-blind, randomized (2:1), placebo-controlled, phase 3 trial conducted at 118 sites[5] (**Fig. 2**)
- Patients with heavily RBC-TD, non-del(5q) LR-MDS who were relapsed-refractory to ESAs or ineligible for ESAs and naive to lenalidomide/HMA were randomized to receive imetelstat 7.5 mg/kg IV (n = 118) or placebo (n = 60) every 4 weeks until disease progression, unacceptable toxicity, withdrawal of consent, or lack of response
- The primary endpoint was 8-week TI rate; secondary endpoints included safety, 24-week TI, duration of response, and HI-E
- Exploratory analyses included assessment of cytogenetic response and mutational status with clinical response

Figure 2. IMerge Phase 3 Study Design

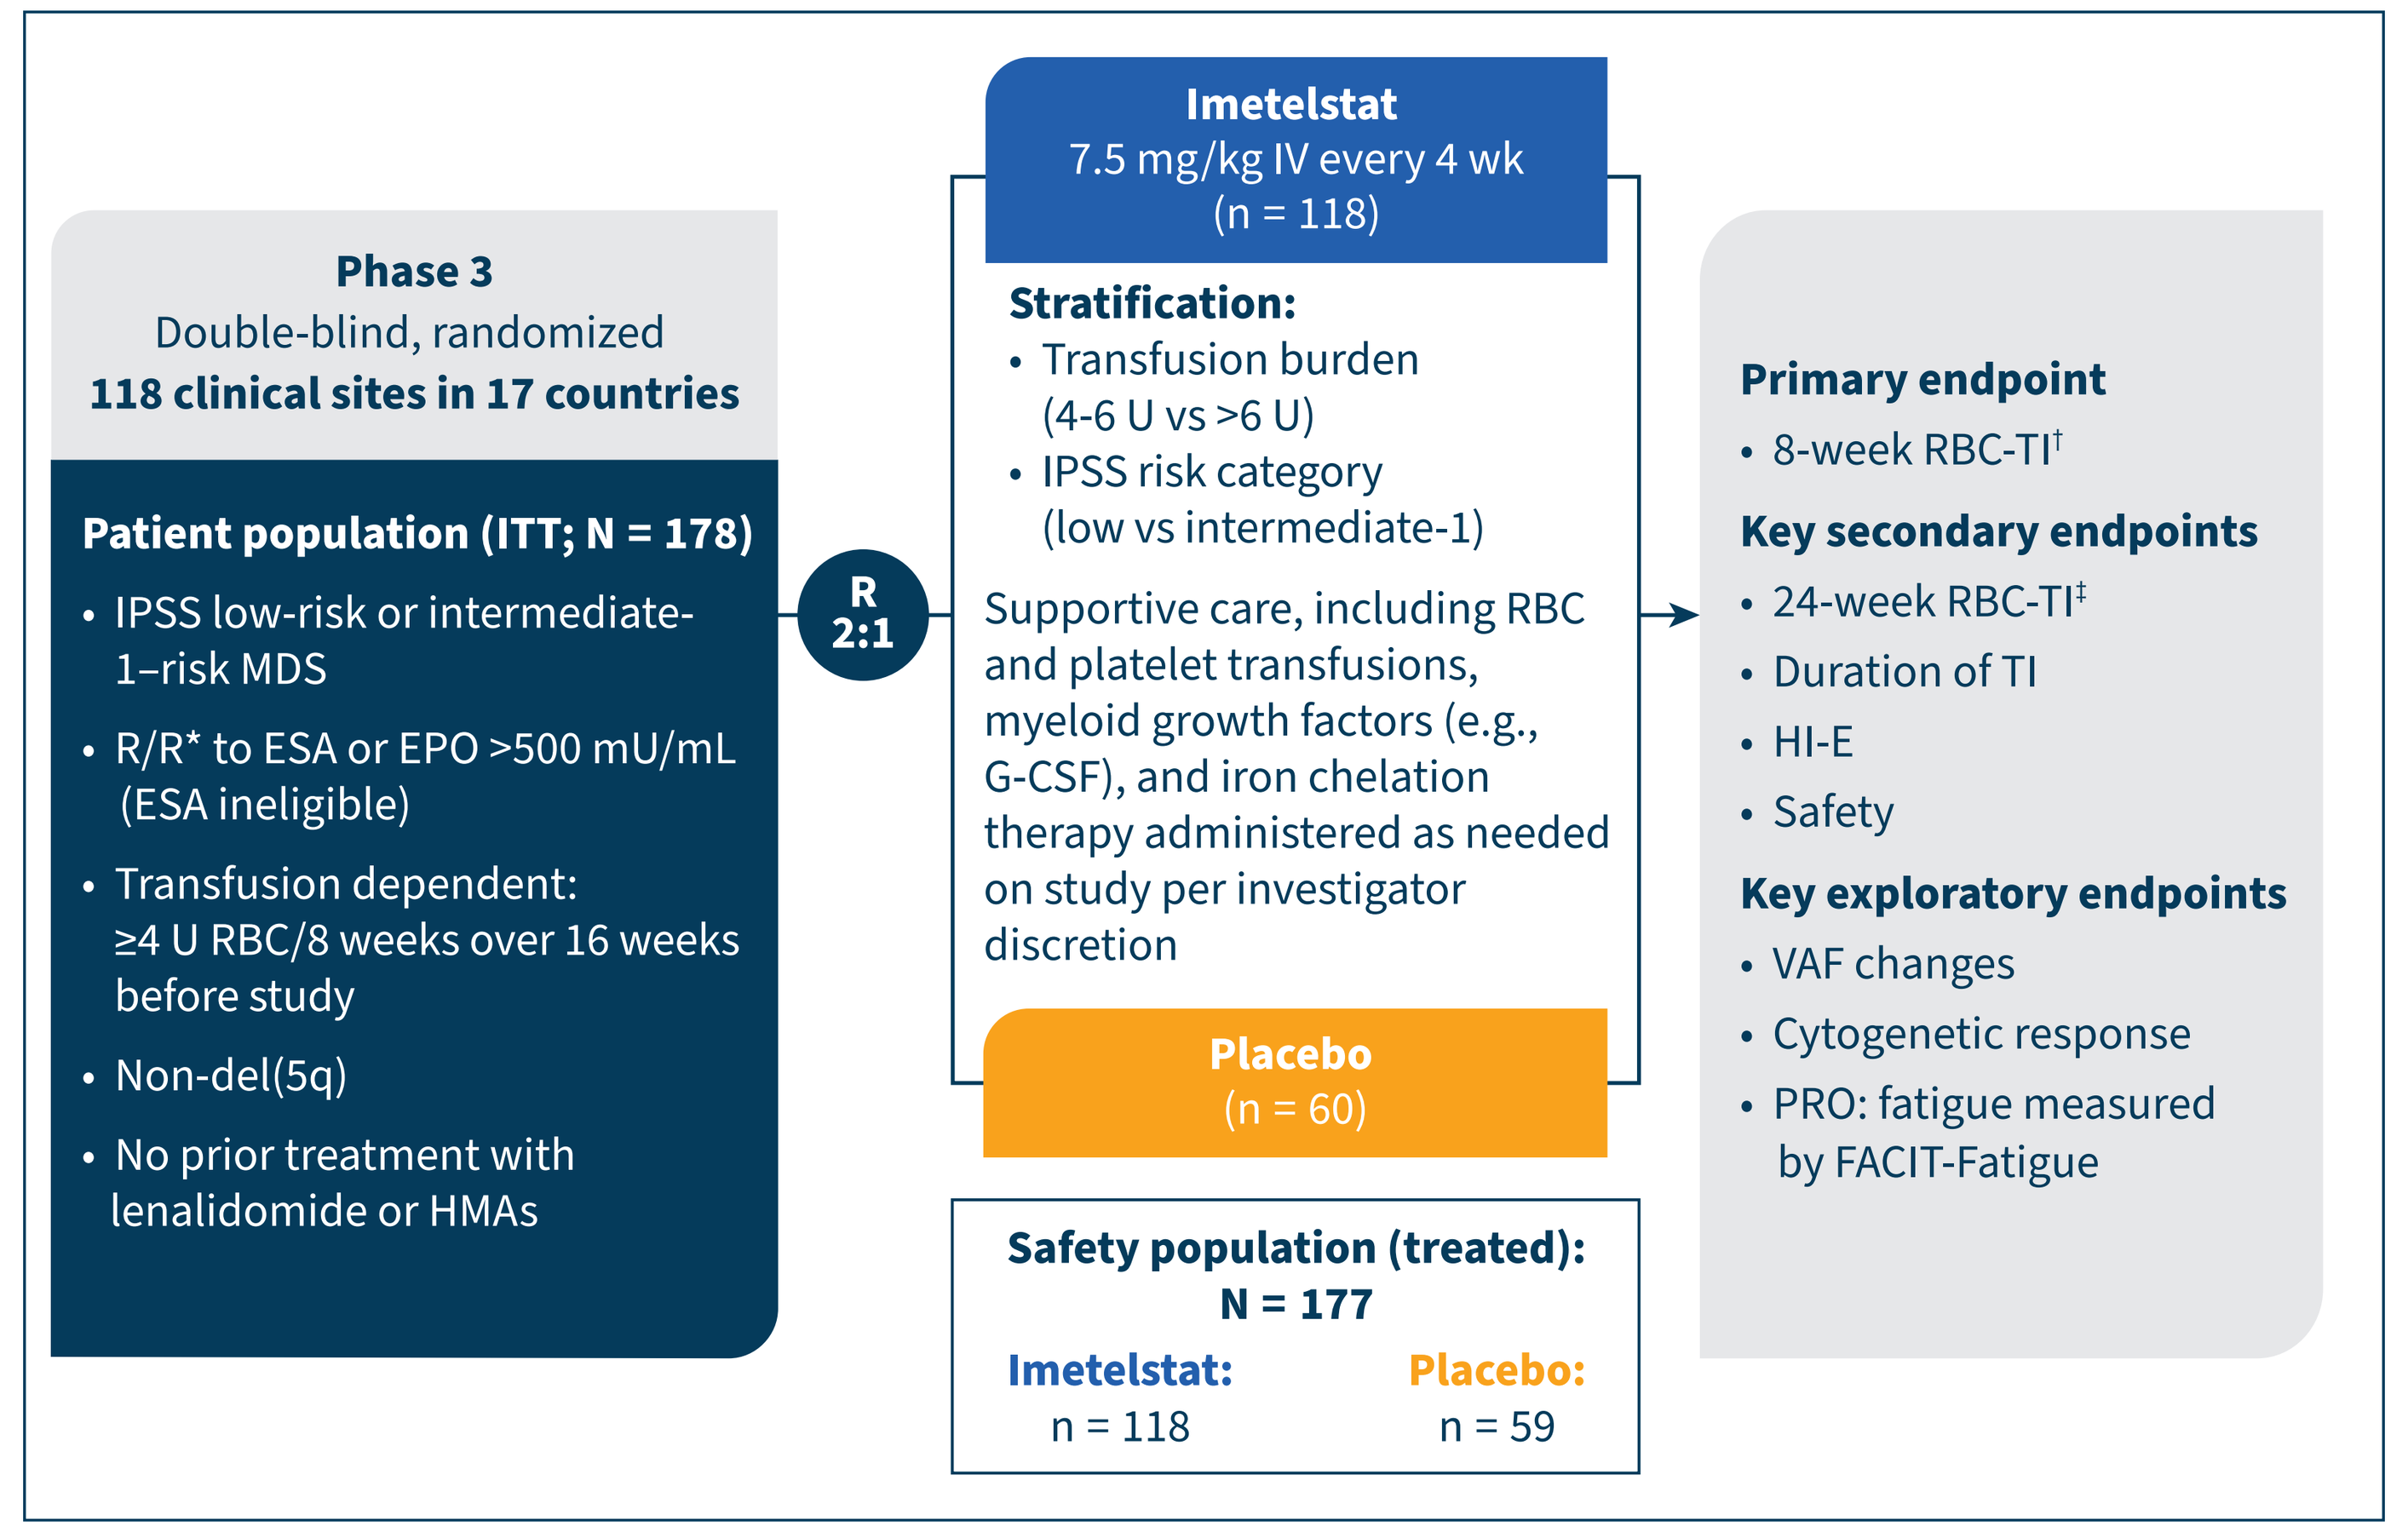
Phase 3
Double-blind, randomized
118 clinical sites in 17 countries
Patient population (ITT; N = 178)
• IPSS low-risk or intermediate-1–risk MDS
• R/R* to ESA or EPO >500 mU/mL (ESA ineligible)
• Transfusion dependent: ≥4 U RBC/8 weeks over 16 weeks before study
• Non-del(5q)
• No prior treatment with lenalidomide or HMAs
R 2:1
Imetelstat
7.5 mg/kg IV every 4 wk
(n = 118)
Stratification:
• Transfusion burden (4-6 U vs >6 U)
• IPSS risk category (low vs intermediate-1)
Supportive care, including RBC and platelet transfusions, myeloid growth factors (e.g., G-CSF), and iron chelation therapy administered as needed on study per investigator discretion
Placebo
(n = 60)
Safety population (treated): N = 177
Imetelstat: n = 118
Placebo: n = 59
Primary endpoint
• 8-week RBC-TI†
Key secondary endpoints
• 24-week RBC-TI‡
• Duration of TI
• HI-E
• Safety
Key exploratory endpoints
• VAF changes
• Cytogenetic response
• PRO: fatigue measured by FACIT-Fatigue

*Received ≥8 weeks of ESA treatment (epoetin alfa ≥40,000 U, epoetin beta ≥30,000 U, darbepoetin alfa 150 µg, or equivalent per week) without Hb rise ≥1.5 g/dL or decreased RBC transfusion requirement ≥4 U/8 weeks or transfusion dependence or reduction in Hb by ≥1.5 g/dL after HI-E from ≥8 weeks of ESA treatment. †Percentage of patients without any RBC transfusion for ≥8 consecutive weeks since entry to the trial. ‡Percentage of patients without any RBC transfusion for ≥24 consecutive weeks since entry to the trial.

Results

Baseline Characteristics

- Baseline characteristics were balanced between study arms[5,6] (**Table 1**)

Table 1. Baseline Characteristics of IMerge Phase 3 Trial

Characteristic	Imetelstat (n = 118)	Placebo (n = 60)
Median age, years (range)	72 (44–87)	73 (39–85)
Male, n (%)	71 (60)	40 (67)
Median time since diagnosis, years (range)	3.5 (0.1–26.7)	2.8 (0.2–25.7)
WHO classification, n (%)		
RS+	73 (62)	37 (62)
RS−	44 (37)	23 (38)
IPSS risk category, n (%)		
Low	80 (68)	39 (65)
Intermediate-1	38 (32)	21 (35)
Median pretreatment Hb, g/dL (range)*	7.9 (5.3–10.1)	7.8 (6.1–9.2)
Median prior RBC transfusion burden, RBC U/8 weeks (range)	6 (4–33)	6 (4–13)
Prior RBC transfusion burden, n (%)		
≥4 to ≤6 U/8 weeks	62 (53)	33 (55)
>6 U/8 weeks	56 (48)	27 (45)
Median sEPO, mU/mL (range)	174.9 (6.0–4460.0)	277.0 (16.9–5514.0)
sEPO level, n (%)†		
≤500 mU/mL	87 (74)	36 (60)
>500 mU/mL	26 (22)	22 (37)
Prior ESA, n (%)	108 (92)	52 (87)
Prior luspatercept, n (%)‡	7 (6)	4 (7)

*Average of all Hb values in the 8 weeks prior to the first dose date, excluding values within 14 days after a transfusion, which was considered to be influenced by transfusion. †Data missing for 5 patients in the imetelstat group and 2 in the placebo group. ‡Insufficient number of patients previously treated with luspatercept to draw conclusions about the effect of imetelstat treatment in such patients.

Overall RBC-TI Rates

- Imetelstat showed significant efficacy vs placebo for 8-week, 24-week, and 1-year RBC-TI[5,6] (**Fig. 3**)
- Among imetelstat responders, median duration of TI was 52 weeks, 80 weeks, and 132 weeks for 8-week, 24-week, and 1-year RBC-TI, respectively

Figure 3. RBC-TI Rates in the ITT Population

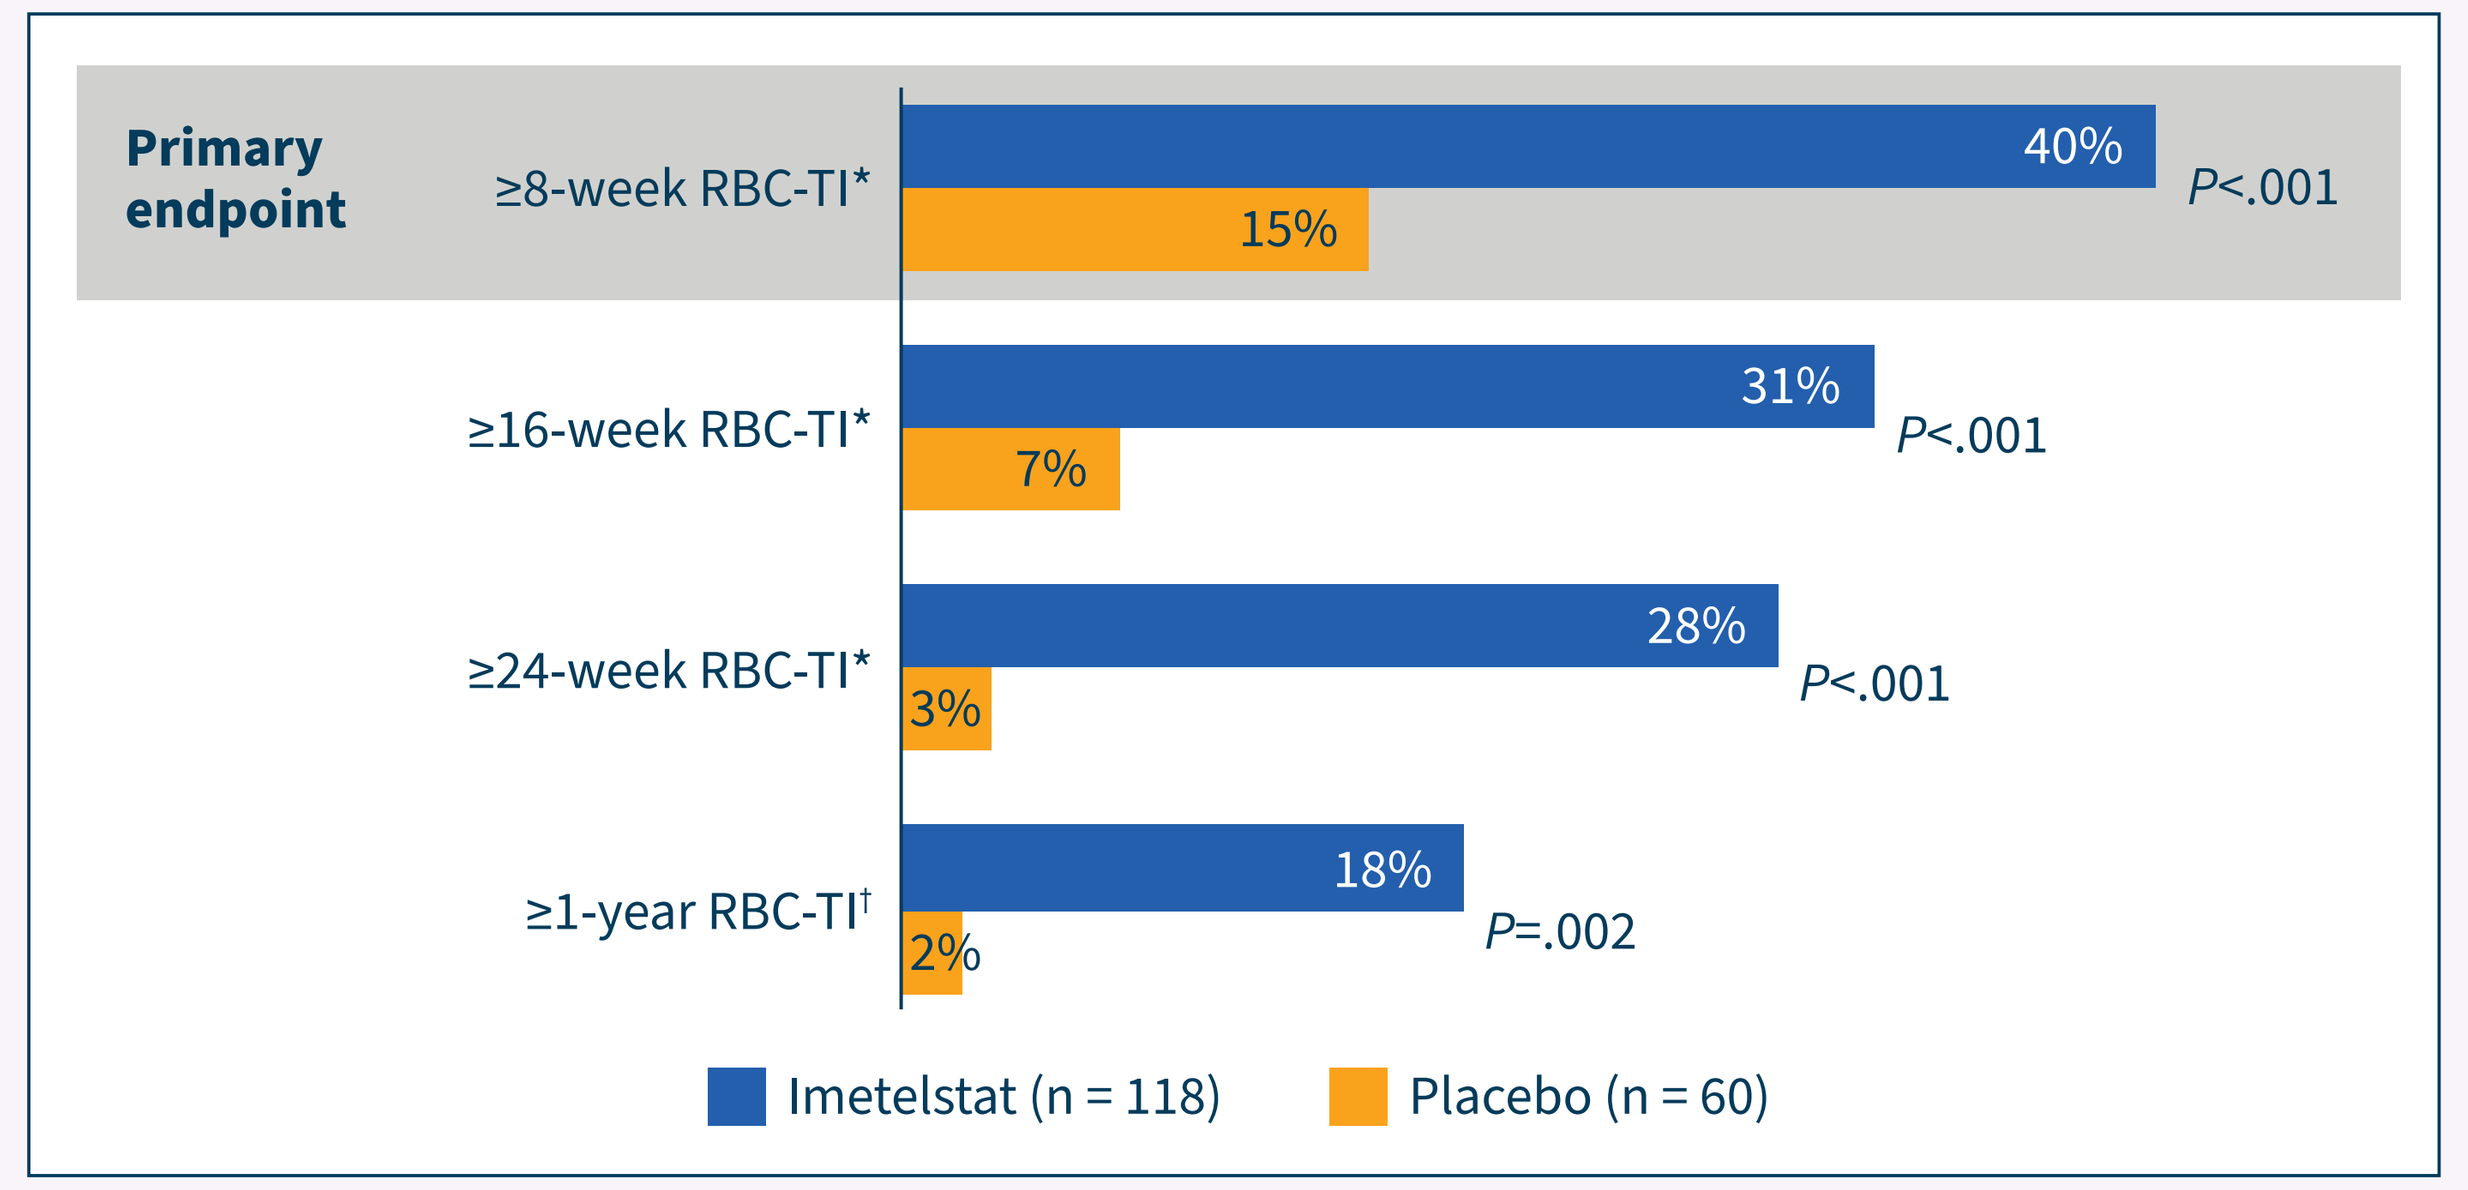
Primary endpoint
≥8-week RBC-TI*
40%
15%
P<.001
≥16-week RBC-TI*
31%
7%
P<.001
≥24-week RBC-TI*
28%
3%
P<.001
≥1-year RBC-TI†
18%
2%
P=.002
Imetelstat (n = 118)
Placebo (n = 60)

*Data cutoff date: October 13, 2022. †Data cutoff date: October 13, 2023.

RBC-TI Rates in Subgroups

- 8-week and 24-week RBC-TI favored imetelstat in various subgroups[5] (**Fig. 4**)

Figure 4. RBC-TI Rates in Subgroups

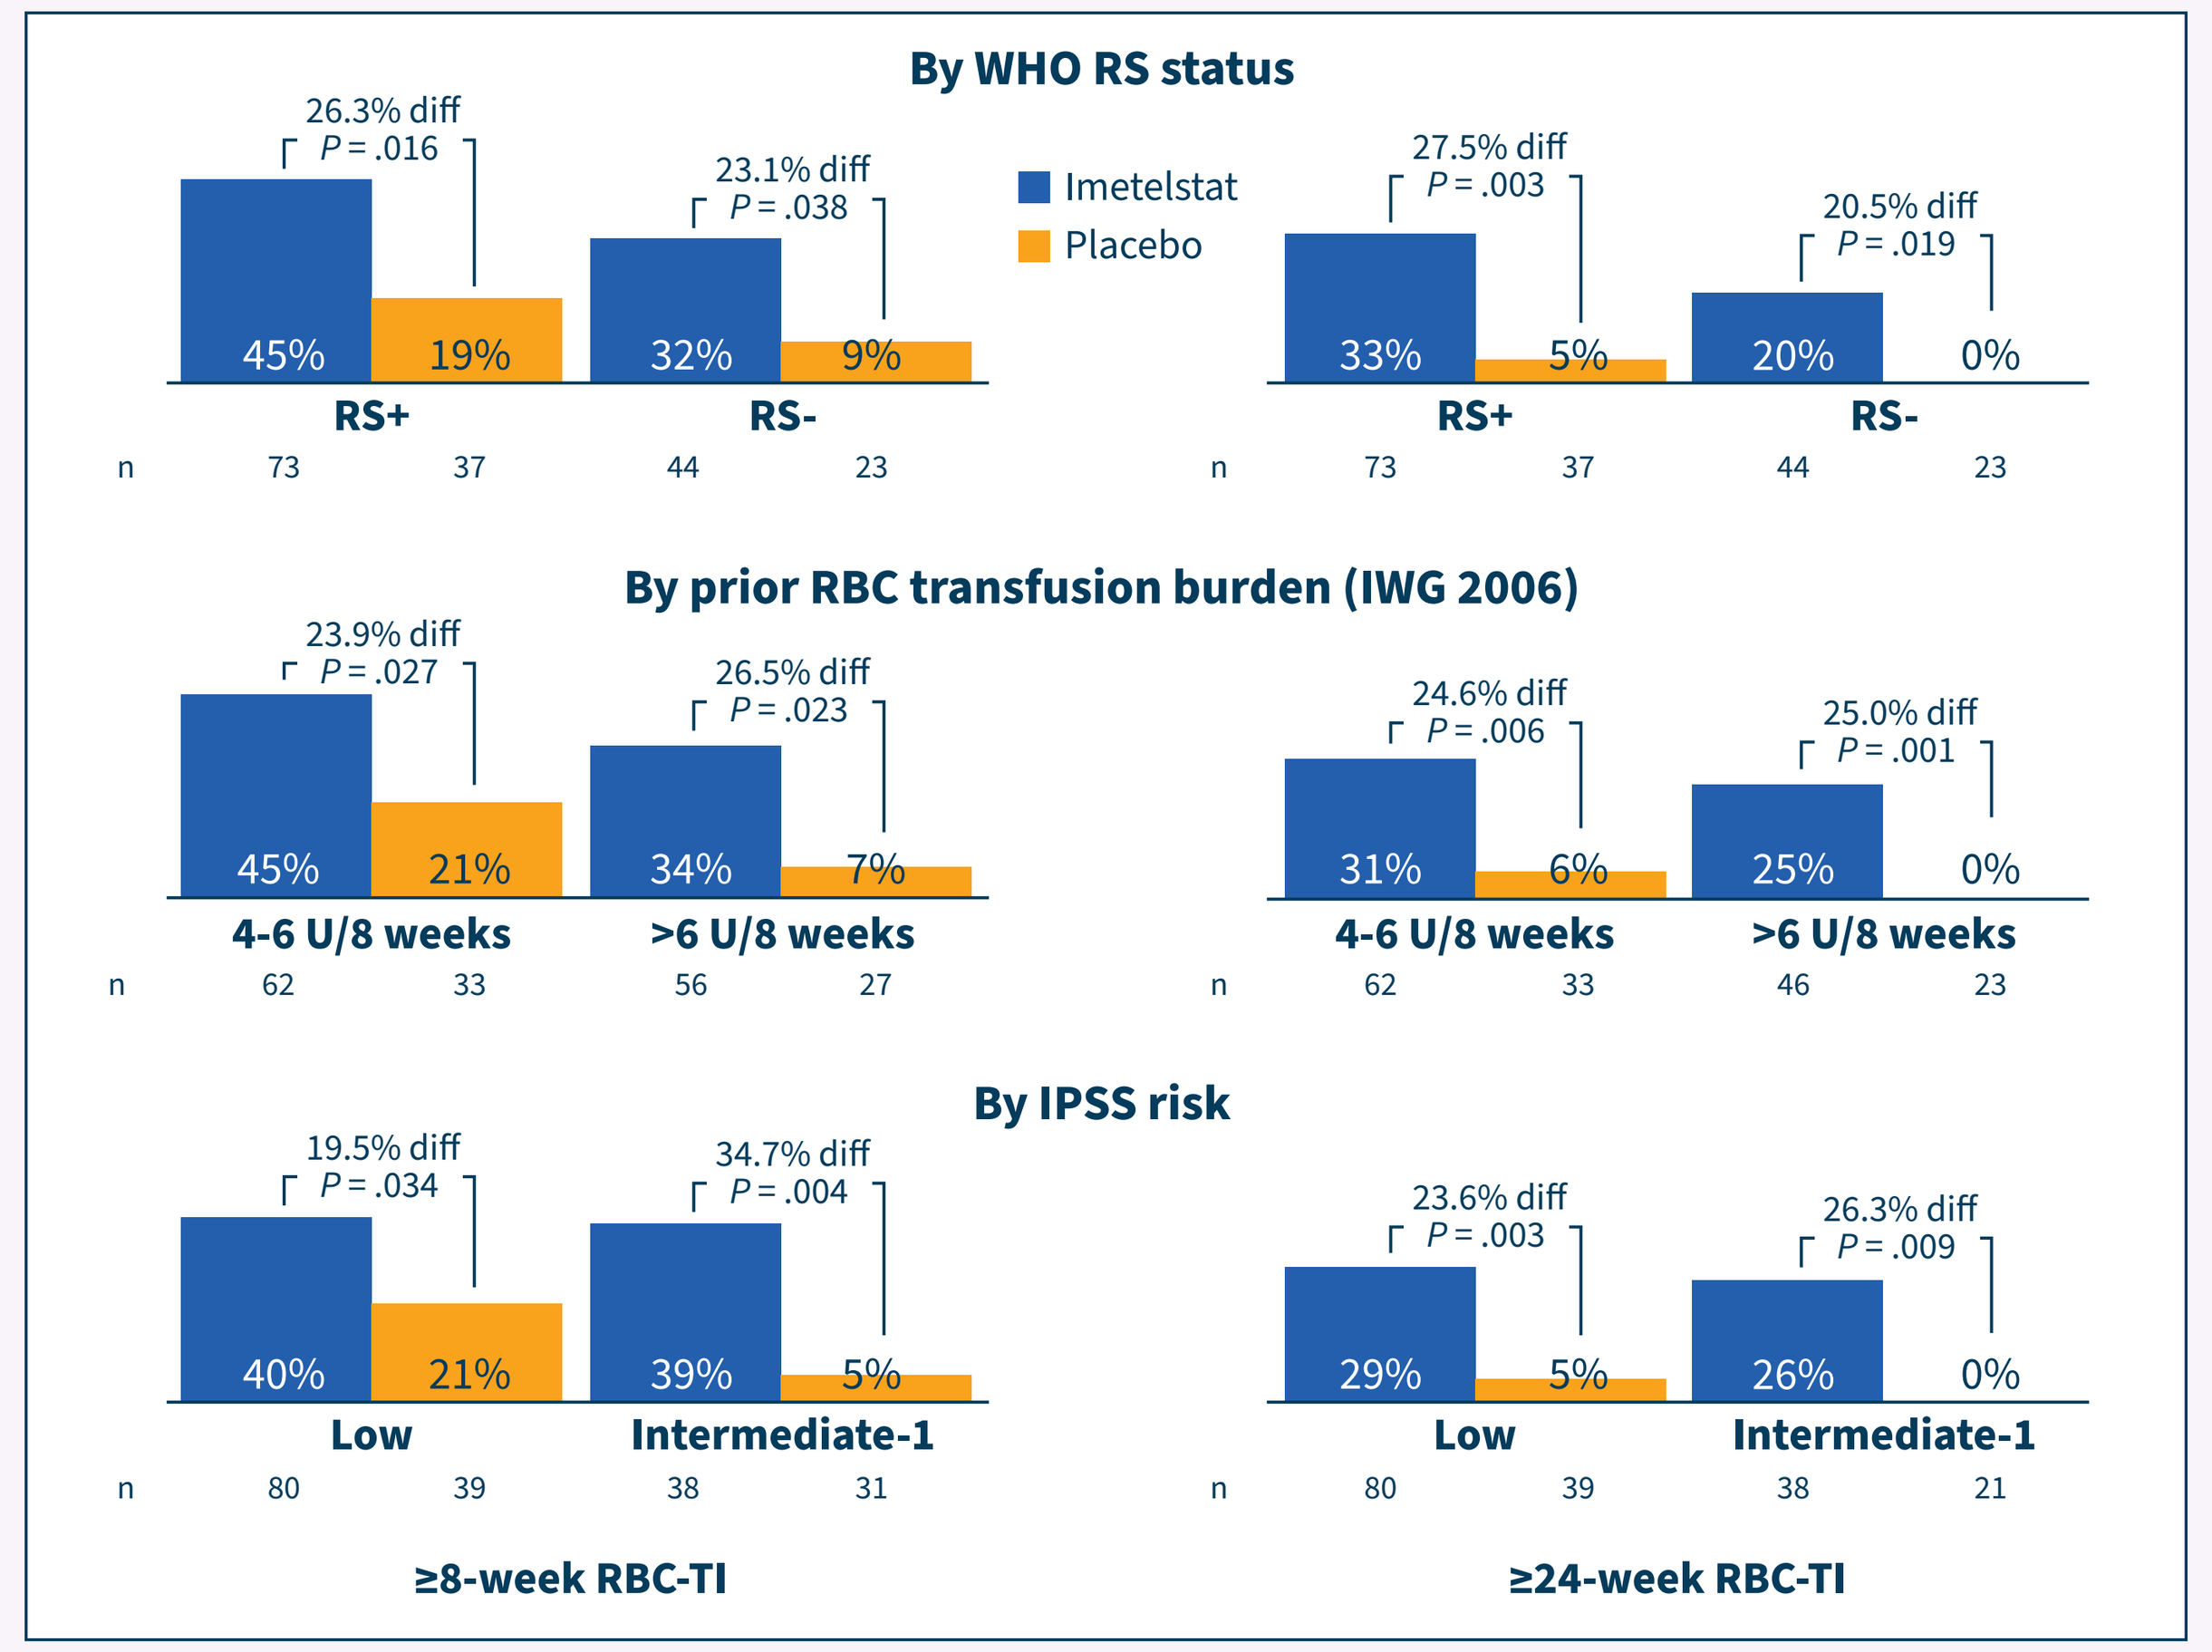
By WHO RS status
Imetelstat
Placebo
26.3% diff P = .016
45% 19% RS+ n 73 37
23.1% diff P = .038
32% 9% RS- 44 23
27.5% diff P = .003
33% 5% RS+ n 73 37
20.5% diff P = .019
20% 0% RS- 44 23
By prior RBC transfusion burden (IWG 2006)
23.9% diff P = .027
45% 21% 4-6 U/8 weeks n 62 33
26.5% diff P = .023
34% 7% >6 U/8 weeks 56 27
24.6% diff P = .006
31% 6% 4-6 U/8 weeks n 62 33
25.0% diff P = .001
25% 0% >6 U/8 weeks 46 23
By IPSS risk
19.5% diff P = .034
40% 21% Low n 80 39
34.7% diff P = .004
39% 5% Intermediate-1 38 31
23.6% diff P = .003
29% 5% Low n 80 39
26.3% diff P = .009
26% 0% Intermediate-1 38 21
≥8-week RBC-TI
≥24-week RBC-TI

Hb Increase in Imetelstat RBC-TI Responders

- In the imetelstat group, median Hb increase from baseline was 3.6 g/dL, 4.2 g/dL, and 5.2 g/dL, for ≥8-week, ≥24-week, and ≥1-year RBC-TI responders, respectively
- Increase in Hb level was noted in imetelstat TI responders within ≤4 weeks of treatment (**Fig. 5**)

Figure 5. Hb Increase in Imetelstat RBC-TI Responders vs Nonresponders (n = 118)

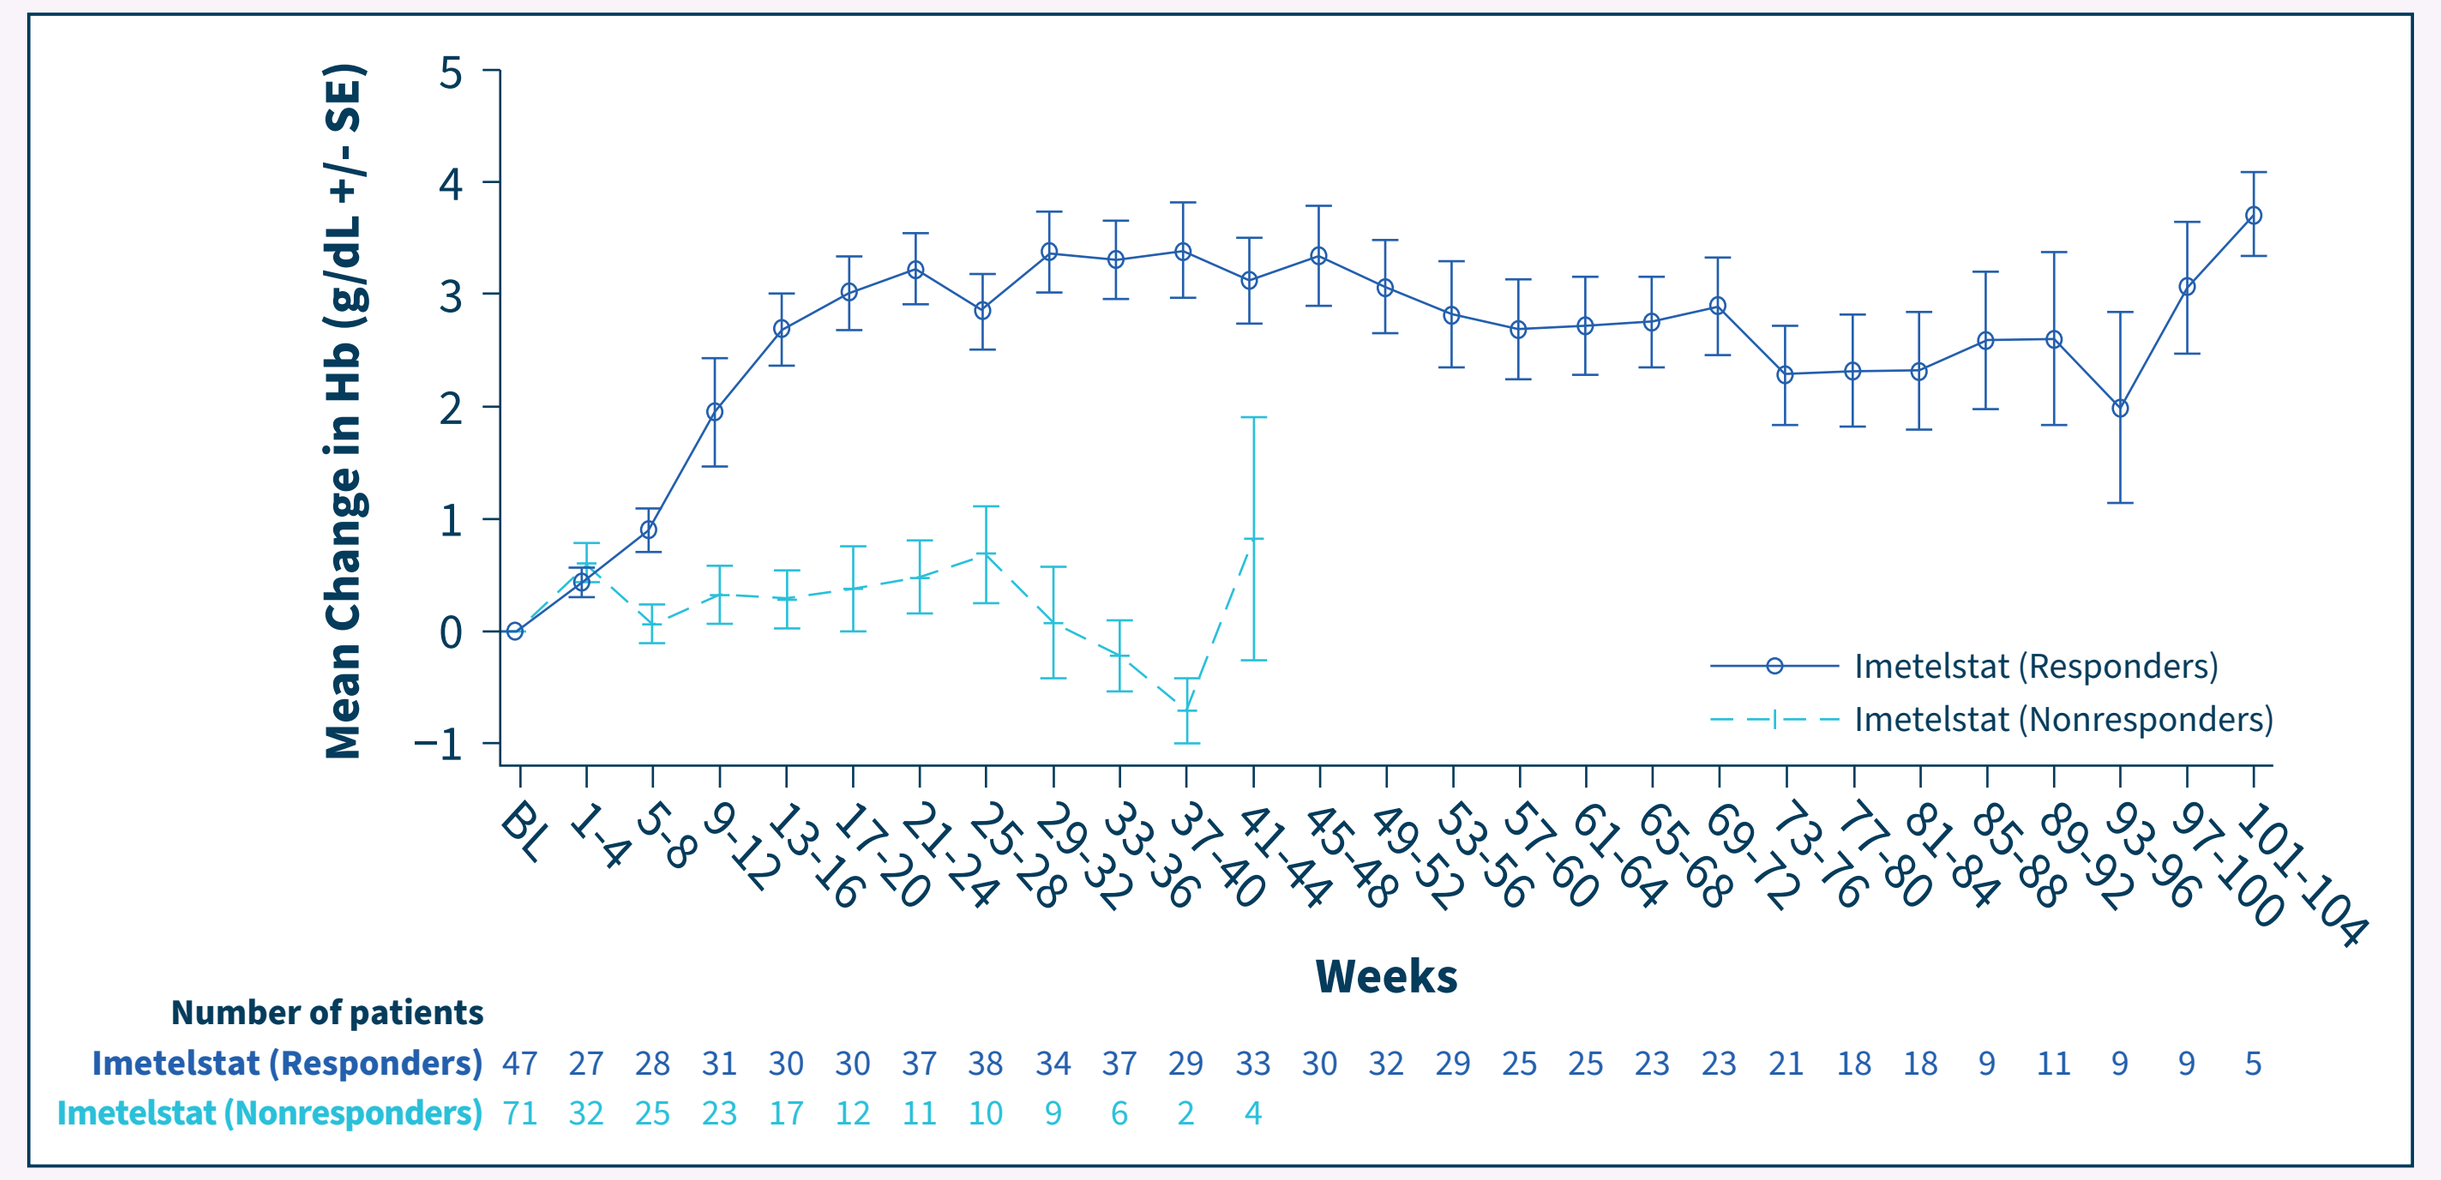
Mean Change in Hb (g/dL +/- SE)
5
4
3
2
1
0
−1
BL 1-4 5-8 9-12 13-16 17-20 21-24 25-28 29-32 33-36 37-40 41-44 45-48 49-52 53-56 57-60 61-64 65-68 69-72 73-76 77-80 81-84 85-88 89-92 93-96 97-100 101-104
Weeks
Imetelstat (Responders)
Imetelstat (Nonresponders)
Number of patients
Imetelstat (Responders) 47 27 28 31 30 30 37 38 34 37 29 33 30 32 29 25 25 23 23 21 18 18 9 11 9 9 5
Imetelstat (Nonresponders) 71 32 25 23 17 12 11 10 9 6 2 4

Cytogenetic Response

- At baseline, 22% of patients had cytogenetic abnormalities, as measured by karyotyping[5] (**Fig. 6**)
- Complete or partial cytogenetic responses were observed in 35% (9/26) of patients in the imetelstat group and 15% (2/13) of patients in the placebo group, as assessed by an independent review committee[5]
- Among cytogenetic responders, 89% (8/9) of patients in the imetelstat group and 50% (1/2) in the placebo group also achieved ≥8-week RBC-TI[6]

Figure 6. Cytogenetic Response

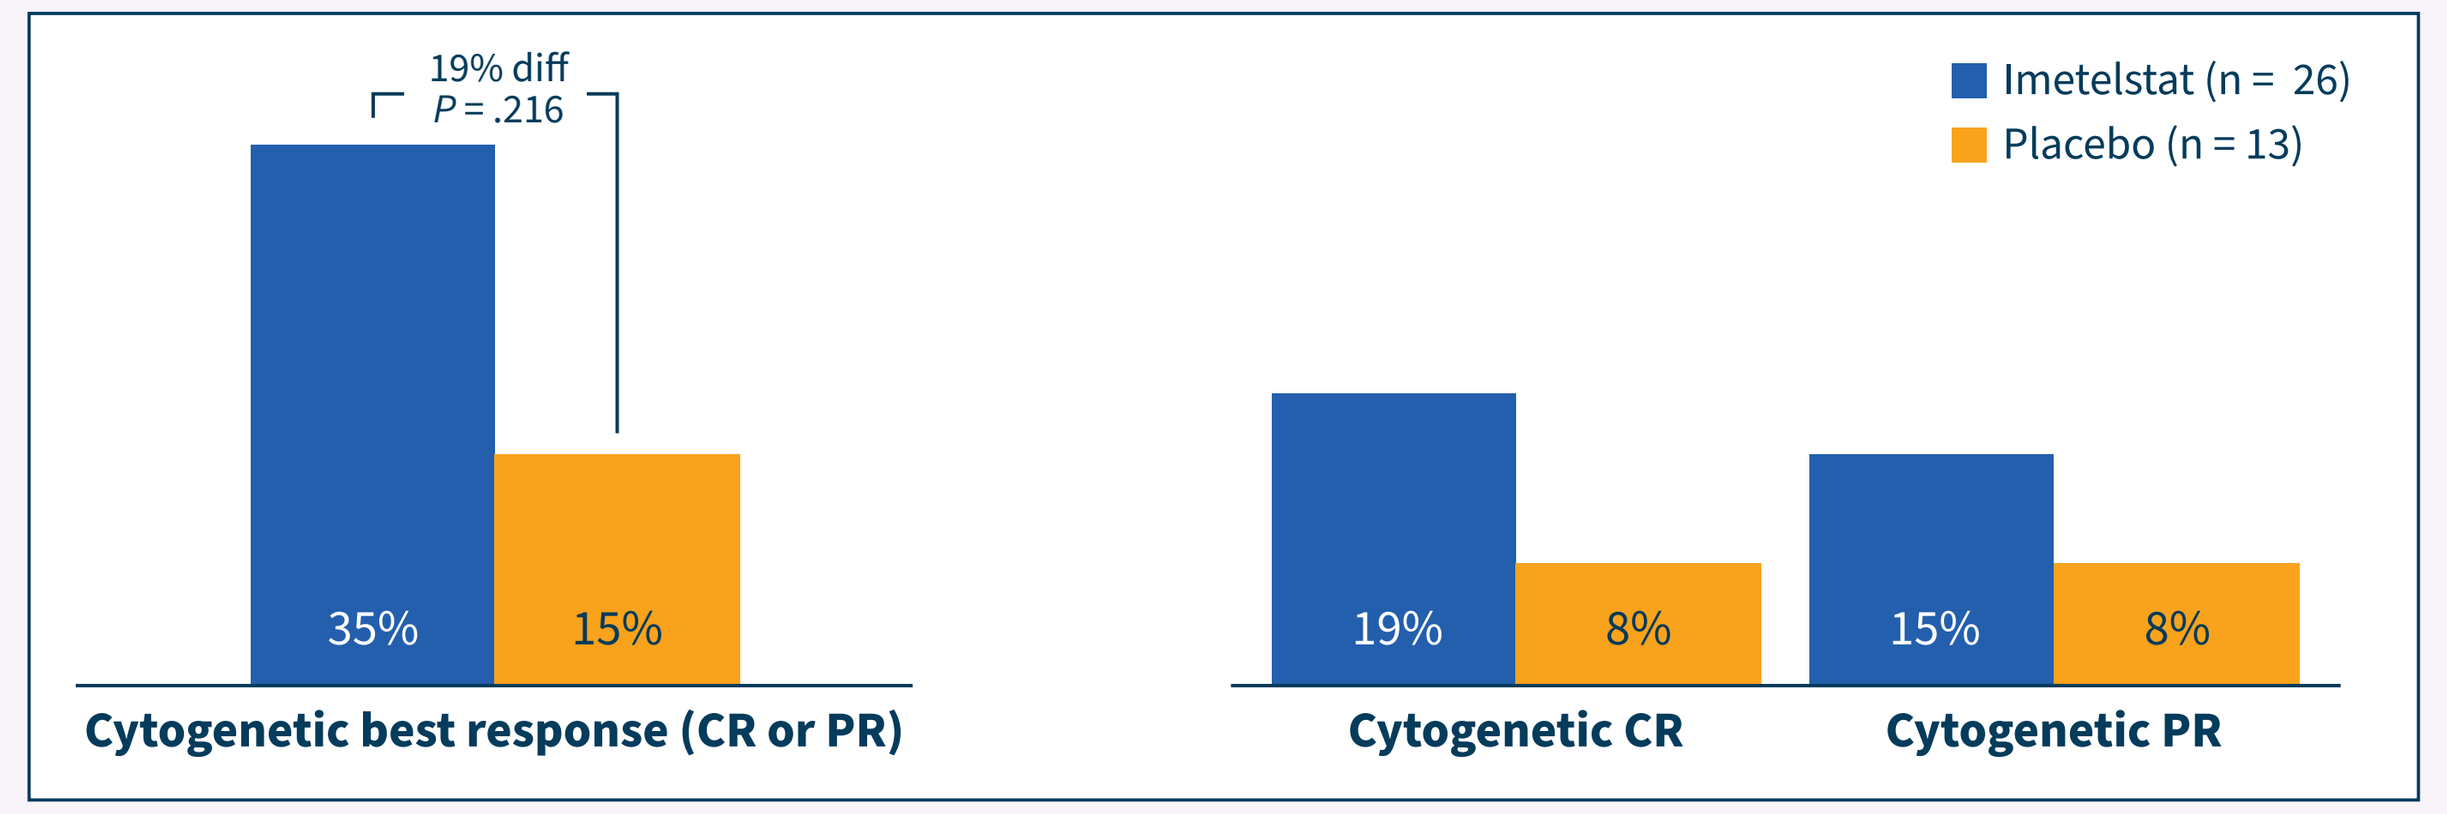
19% diff P = .216
35% 15%
Cytogenetic best response (CR or PR)
19% 8%
Cytogenetic CR
15% 8%
Cytogenetic PR
Imetelstat (n = 26)
Placebo (n = 13)

SF3B1 VAF Changes

- Mean percentage of *SF3B1* VAF reduction was −6% with placebo and −42% with imetelstat
- *SF3B1* VAF reduction significantly correlated with continuous TI response and Hb increase[5,6] (**Fig. 7**)
- Among 18 patients who achieved ≥1-year RBC-TI and had available mutational data, 13 (72%) had ≥50% *SF3B1* VAF reduction, including 7 patients with complete elimination of VAF[7]

Figure 7. Correlation Between *SF3B1* VAF Reduction and (A) RBC-TI Duration and (B) Hb Increase

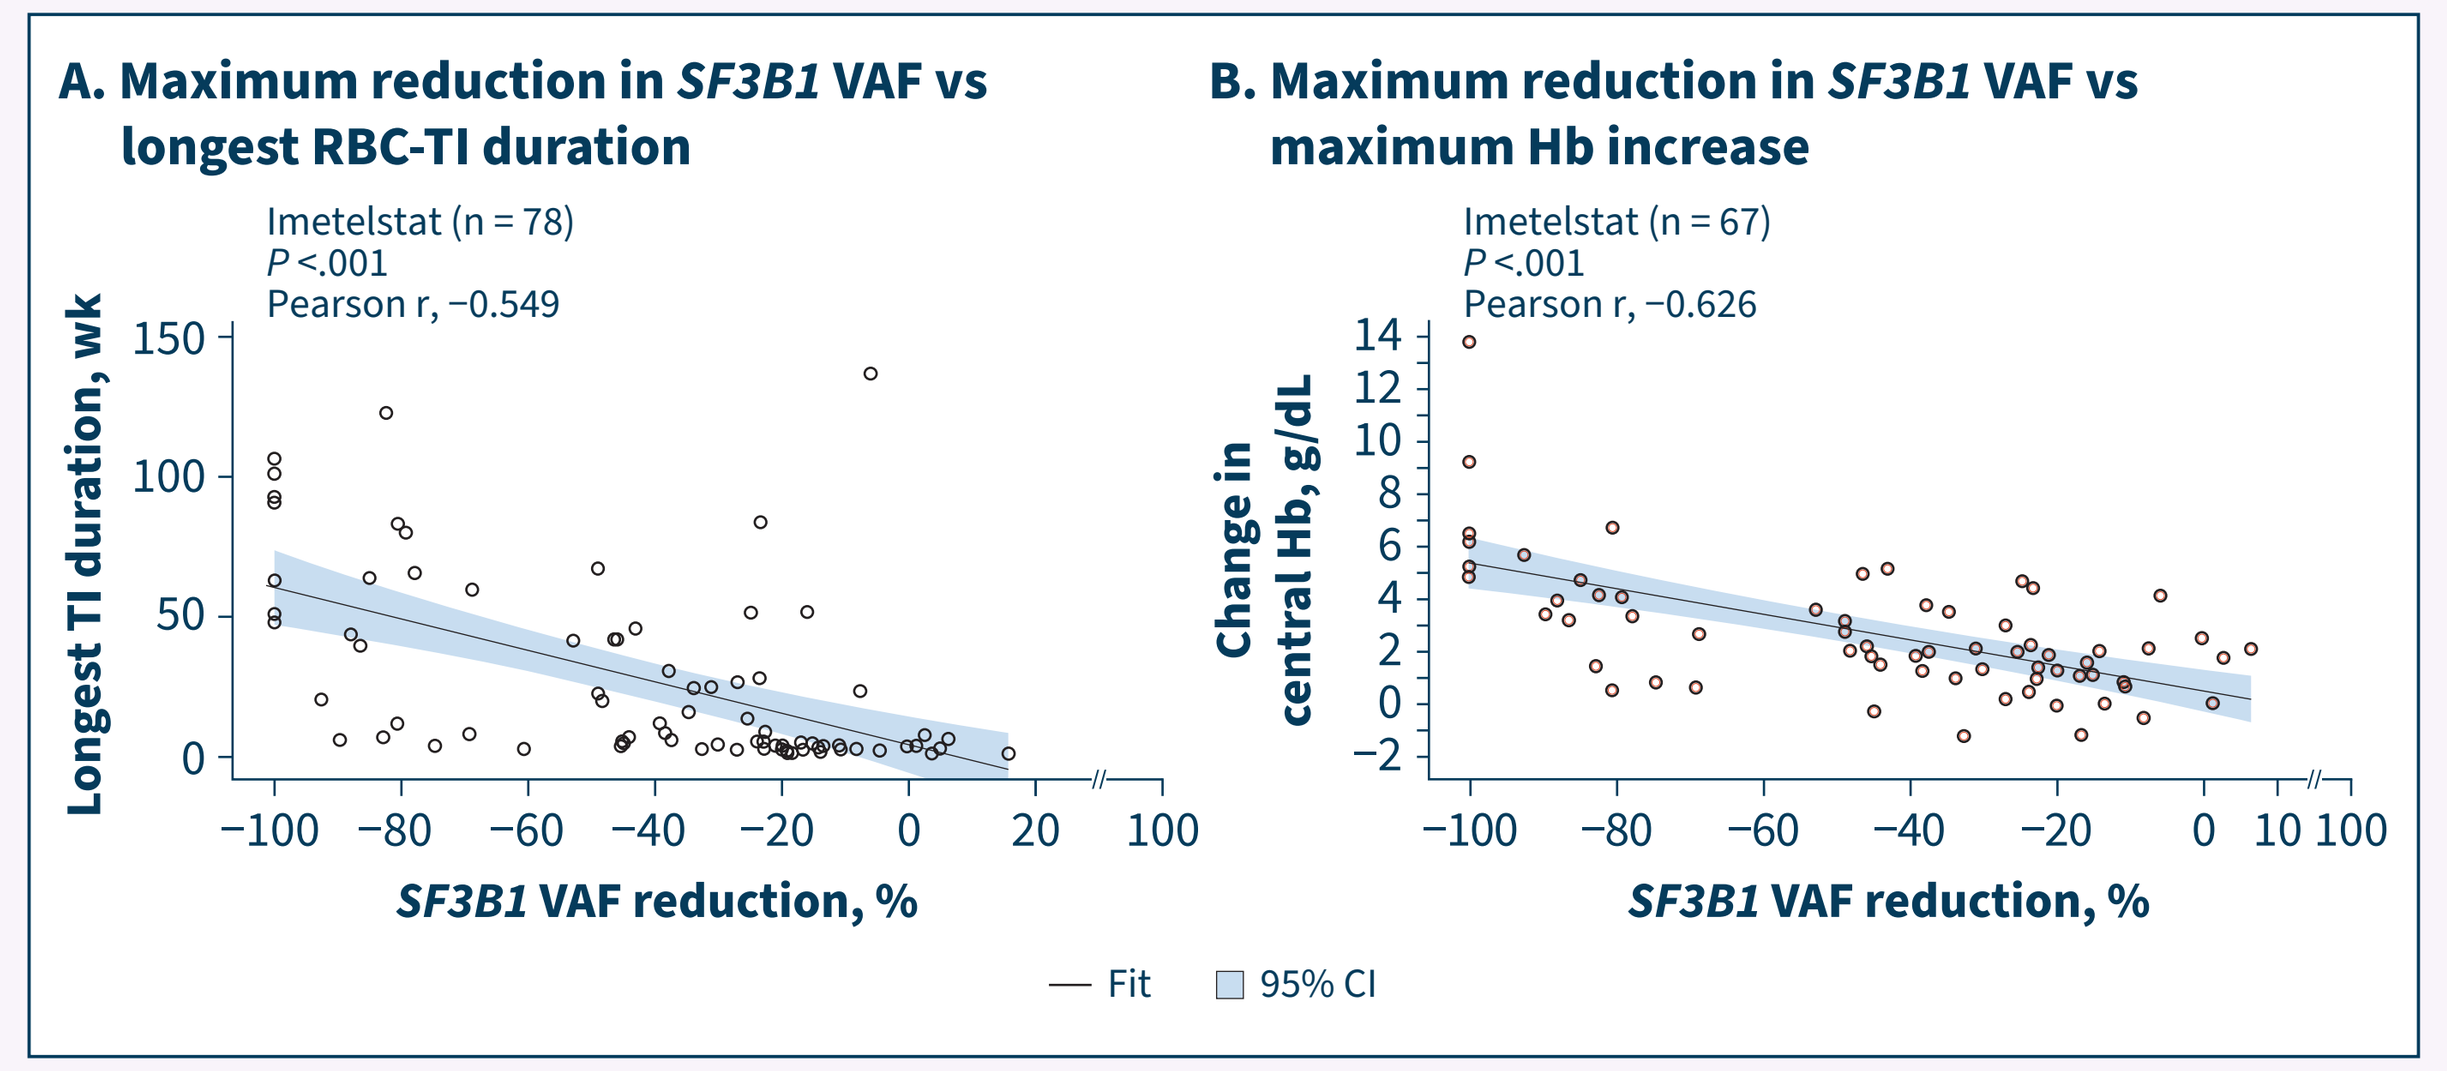
A. Maximum reduction in SF3B1 VAF vs longest RBC-TI duration
Imetelstat (n = 78)
P <.001
Pearson r, −0.549
Longest TI duration, wk
150
100
50
0
−100 −80 −60 −40 −20 0 20 100
SF3B1 VAF reduction, %
B. Maximum reduction in SF3B1 VAF vs maximum Hb increase
Imetelstat (n = 67)
P <.001
Pearson r, −0.626
Change in central Hb, g/dL
14
12
10
8
6
4
2
0
−2
−100 −80 −60 −40 −20 0 10 100
SF3B1 VAF reduction, %
Fit
95% CI

RBC-TI Correlations in Imetelstat Responders vs Nonresponders

- In patients treated with imetelstat, ≥8-week and ≥24-week RBC-TI significantly correlated with achievement of cytogenetic response, reduction in RS+ cells, and *SF3B1* VAF reduction[5,6] (**Table 2**)

Table 2. RBC-TI Correlations in Imetelstat-Treated Patients

	≥8 weeks			≥24 weeks		
Patients, n/n (%)	**Responders**	**Nonresponders**	***P* value**	**Responders**	**Nonresponders**	***P* value**
RBC-TI + cytogenetic CR/PR per IRC	8/9 (89)	6/17 (35)	.014	6/9 (67)	3/17 (18)	.028
RBC-TI + ≥50% bone marrow RS reduction	22/29 (76)	16/42 (38)	.003	18/29 (62)	11/42 (26)	.003
RBC-TI + ≥50% *SF3B1* VAF reduction	19/23 (83)	21/55 (38)	<.001	16/23 (70)	13/55 (24)	<.001

PRO Fatigue Measured by FACIT-Fatigue

- A higher proportion of patients treated with imetelstat vs placebo reported sustained meaningful improvement (increase of ≥3 points for ≥2 consecutive cycles) in fatigue score by FACIT-Fatigue scale and experienced a shorter median time to first sustained clinically meaningful improvement in fatigue[5] (**Fig. 8**)
- Imetelstat TI responders reported significantly sustained meaningful improvement in fatigue in comparison with nonresponders[5] (**Table 3**)

Figure 8. PRO Fatigue Measured by FACIT-Fatigue Scale

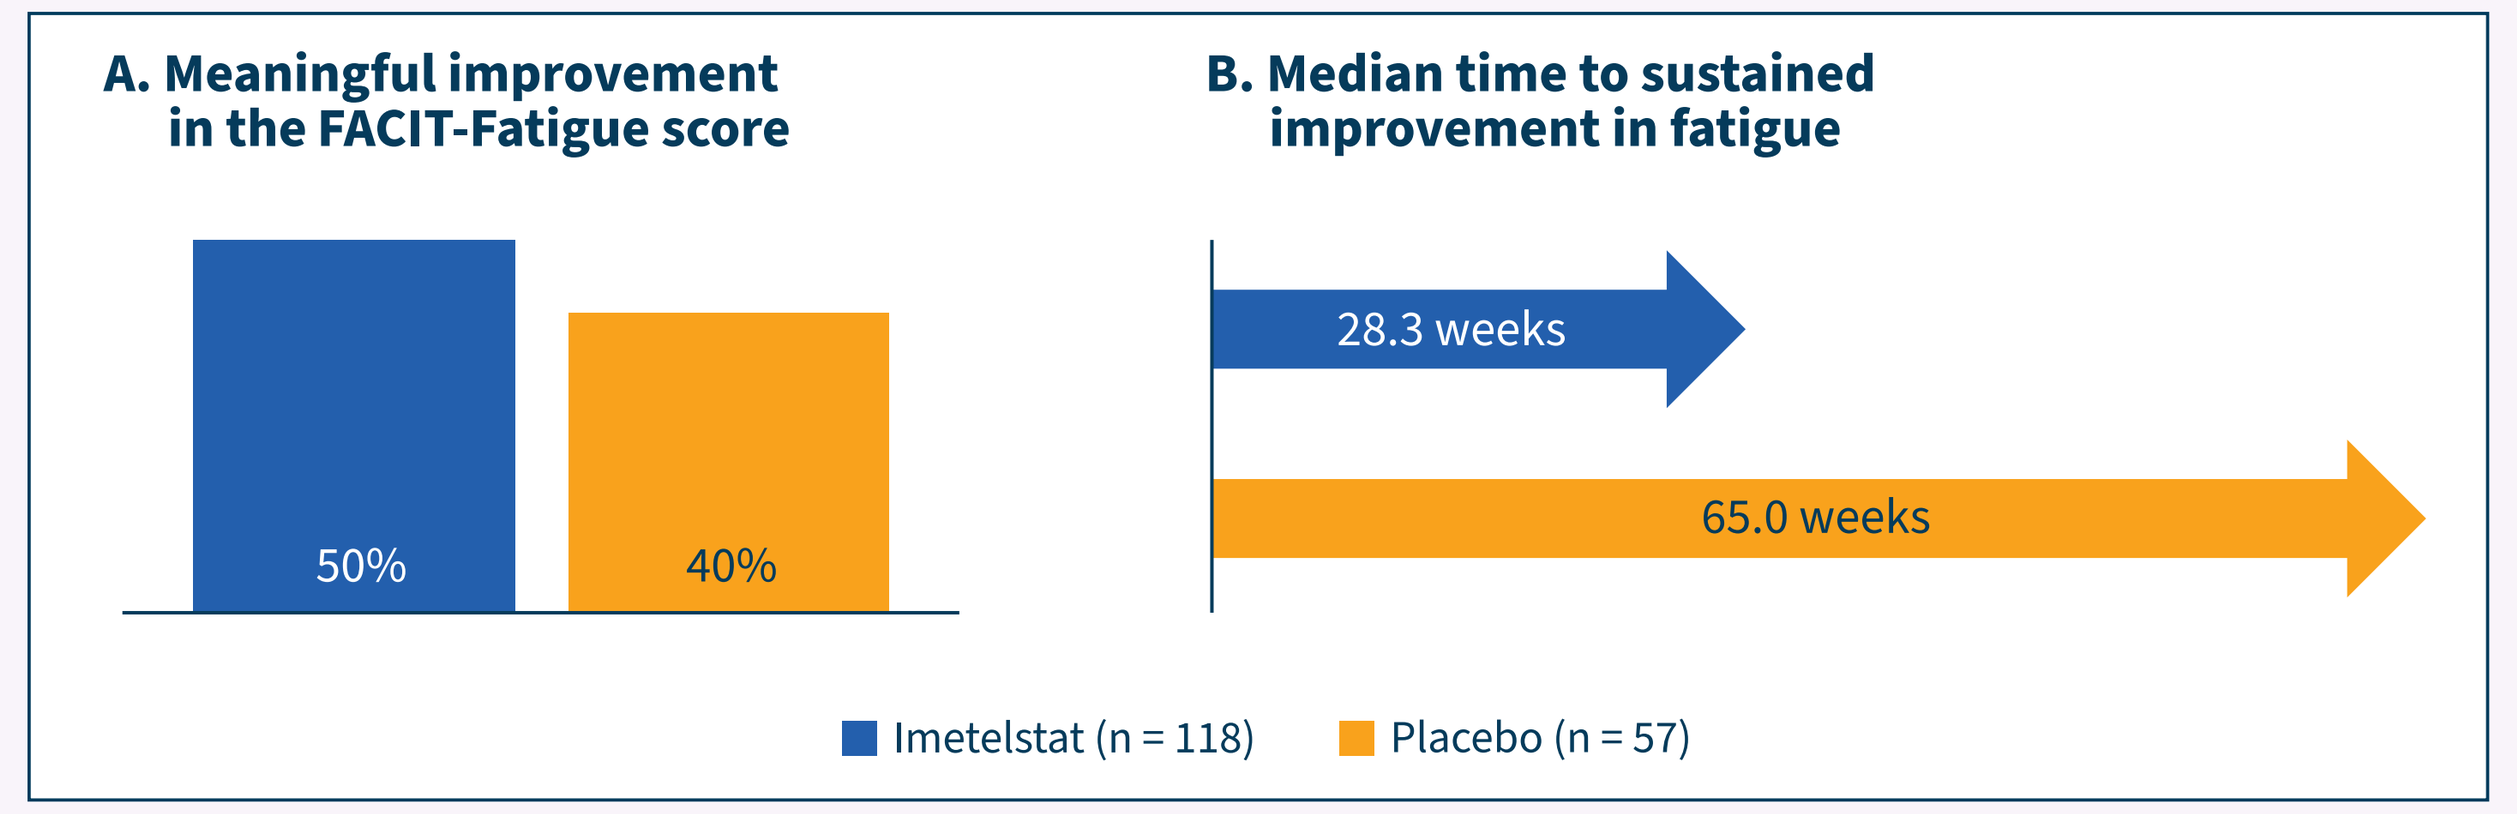
A. Meaningful improvement in the FACIT-Fatigue score
50%
40%
B. Median time to sustained improvement in fatigue
28.3 weeks
65.0 weeks
Imetelstat (n = 118)
Placebo (n = 57)

Table 3. Sustained Meaningful Improvement in Fatigue in RBC-TI Imetelstat Responders vs Nonresponders

Patients, n/n (%)	Responders	Nonresponders	*P* value
≥8-week RBC-TI	33/47 (70)	26/71 (37)	<.001
≥24-week RBC-TI	24/33 (73)	35/85 (41)	.004

Safety

- Most common grade 3-4 treatment-emergent AEs with imetelstat vs placebo were neutropenia (68% vs 3%) and thrombocytopenia (62% vs 8%)[5]
- >80% of grade 3-4 cytopenia events were reversible to grade ≤2 within 4 weeks[5]
- 72% and 60% of ≥8-week RBC-TI responders with imetelstat had grade 3-4 neutropenia and thrombocytopenia, respectively[8]
- Clinical consequences of infection and bleeding were low and similar for imetelstat and placebo

Conclusions

- **In IMerge, patients with LR-MDS derived clinical benefit with imetelstat vs placebo**
- **RBC-TI rates were consistently improved with imetelstat vs placebo across subgroups: RS status, prior RBC transfusion burden, or IPSS risk category**
- **Achievement of RBC-TI correlated with reduced mutational burden, Hb rise, and improvement in patient-reported fatigue**
- **Safety results were consistent with prior imetelstat clinical experience, with no new safety signals**
 - **>80% of grade 3-4 cytopenia events were reversible to grade ≤2 within 4 weeks**
 - **Clinical consequences from grade 3-4 neutropenia and thrombocytopenia were similar in patients treated with imetelstat and placebo**

ABBREVIATIONS

AE, adverse event; BL, baseline; CR, complete response; diff, difference; EMA, erythroid maturation agent; EPO, erythropoietin; ESA, erythropoiesis-stimulating agent; FACIT, Functional Assessment of Chronic Illness Therapy; G-CSF, granulocyte colony-stimulating factor; Hb, hemoglobin; HI-E, hematologic improvement-erythroid; HMA, hypomethylating agent; IPSS, International Prognostic Scoring System; IRC, independent review committee; ITT, intent-to-treat; IV, intravenous; IWG, International Working Group; LR-MDS, lower-risk myelodysplastic syndromes; MDS, myelodysplastic syndromes; mut, mutation; PR, partial response; PRO, patient-reported outcome; R, randomization; RBC, red blood cell; R/R, relapsed/refractory; RS, ring sideroblast; SE, standard error; sEPO, serum erythropoietin; SF3B1, splicing factor 3b subunit 1; TD, transfusion dependence; TI, transfusion independence; U, unit; VAF, variant allele frequency; WBC, white blood cell; WHO, World Health Organization.

REFERENCES

1. Wang X, et al. *Blood Adv*. 2018;2:2378–2388. **2.** Asai A, et al. *Cancer Res*. 2003;63:3931–3939. **3.** Herbert BS, et al. *Oncogene*. 2005;24:5262–5268. **4.** Mosoyan G, et al. *Leukemia*. 2017;31:2458–2467. **5.** Platzbecker U, Santini V, et al. *Lancet*. 2024;403:249–260. **6.** Santini V, et al. EHA 2023. Abstract S164. **7.** Platzbecker U, et al. *Blood*. 2023;142(Suppl 1):4605. **8.** Zeidan AM, et al. *Blood*. 2023;142(Suppl 1):6478.

ACKNOWLEDGMENTS

Portions of these data were published in *Lancet*. 2024;403:249–260 and presented at the European Hematology Association (EHA 2023) and at the American Society of Hematology (ASH 2023) congresses. All authors contributed to and approved the presentation. Writing and editorial assistance was provided by Ashfield MedComms, an Inizio Company, and was funded by Geron Corporation.

DISCLOSURES

The presenter, Dr Yazan F. Madanat, reports honoraria or consulting fees from Blueprint Medicines, Geron Corporation, and OncLive; participation in advisory boards and honoraria from Sierra Oncology, Stemline Therapeutics, Blueprint Medicines, Morphosys, Taiho Oncology, and Novartis; and travel reimbursement from Blueprint Medicines and Morphosys.

Presented at the NCCN Annual Conference, April 5-7, 2024; Orlando, FL, USA