PS1456

FAVORABLE OVERALL SURVIVAL OF IMETELSTAT-TREATED RELAPSED/REFRACTORY MYELOFIBROSIS PATIENTS COMPARED WITH CLOSELY MATCHED REAL WORLD DATA

Andrew Kuykendall*[1], Ying Wan[2], John Mascarenhas[3], Jean-Jacques Kiladjian[4], Alessandro Vannucchi[5], Julia Wang[6], Qi Xia[6], Eugene Zhu[6], Faye Feller[2], Aleksandra Rizo[2], Jacqueline Bussolari[6], Rami Komrokji[1]

[1]Moffitt Cancer Center, Tampa, FL, United States, [2]Geron Corporation, Menlo Park, CA, United States, [3]Icahn School of Medicine at Mount Sinai, New York, NY, United States, [4]Hôpital Saint-Louis, Paris, France, [5]AOU Careggi, University of Florence, Florence, Italy, [6]Janssen Research & Development, Raritan, NJ, United States

*Presenting Author

BACKGROUND

- Myelofibrosis (MF), particularly Intermediate-2 (Int-2) or High-Risk disease per Dynamic International Prognostic Scoring System (DIPSS) criteria, is a life-threatening disease for which the Janus Kinase inhibitor (JAKi), ruxolitinib, is currently the only approved therapy.
 - o Currently, there is no approved or effective therapy for patients who lack or lose response to ruxolitinib.
 - o These relapsed/refractory (R/R) patients have a notably poor prognosis, with median overall survival (OS) of 12-14 months.[1,2]
- Imetelstat, a 13-mer oligonucleotide that specifically targets the RNA template of human telomerase, is a potent competitive inhibitor of telomerase enzymatic activity (**Figure 1**).
- A Phase 2 study, MYF2001 (NCT02426086), evaluated 2 doses of imetelstat in JAKi-R/R Int-2 or High-Risk MF.[3]
 - o Encouraging total symptom score improvements were observed in the 9.4 mg/kg arm (no appreciable efficacy signal was observed in the 4.7 mg/kg arm).
 - o Median OS was 29.9 months (95% confidence interval [CI] 22.8, NE) in the 9.4 mg/kg arm, with clinical cut-off of 22 Oct 2018.

Goal of the Study

- To further assess the potential OS benefit of imetelstat, R/R MF patients treated with 9.4 mg/kg in MYF2001 were compared to a closely matched patient population from real-world data (RWD), treated with best available therapy (BAT).

Figure 1. Imetelstat binds to RNA template, preventing maintenance of telomeres

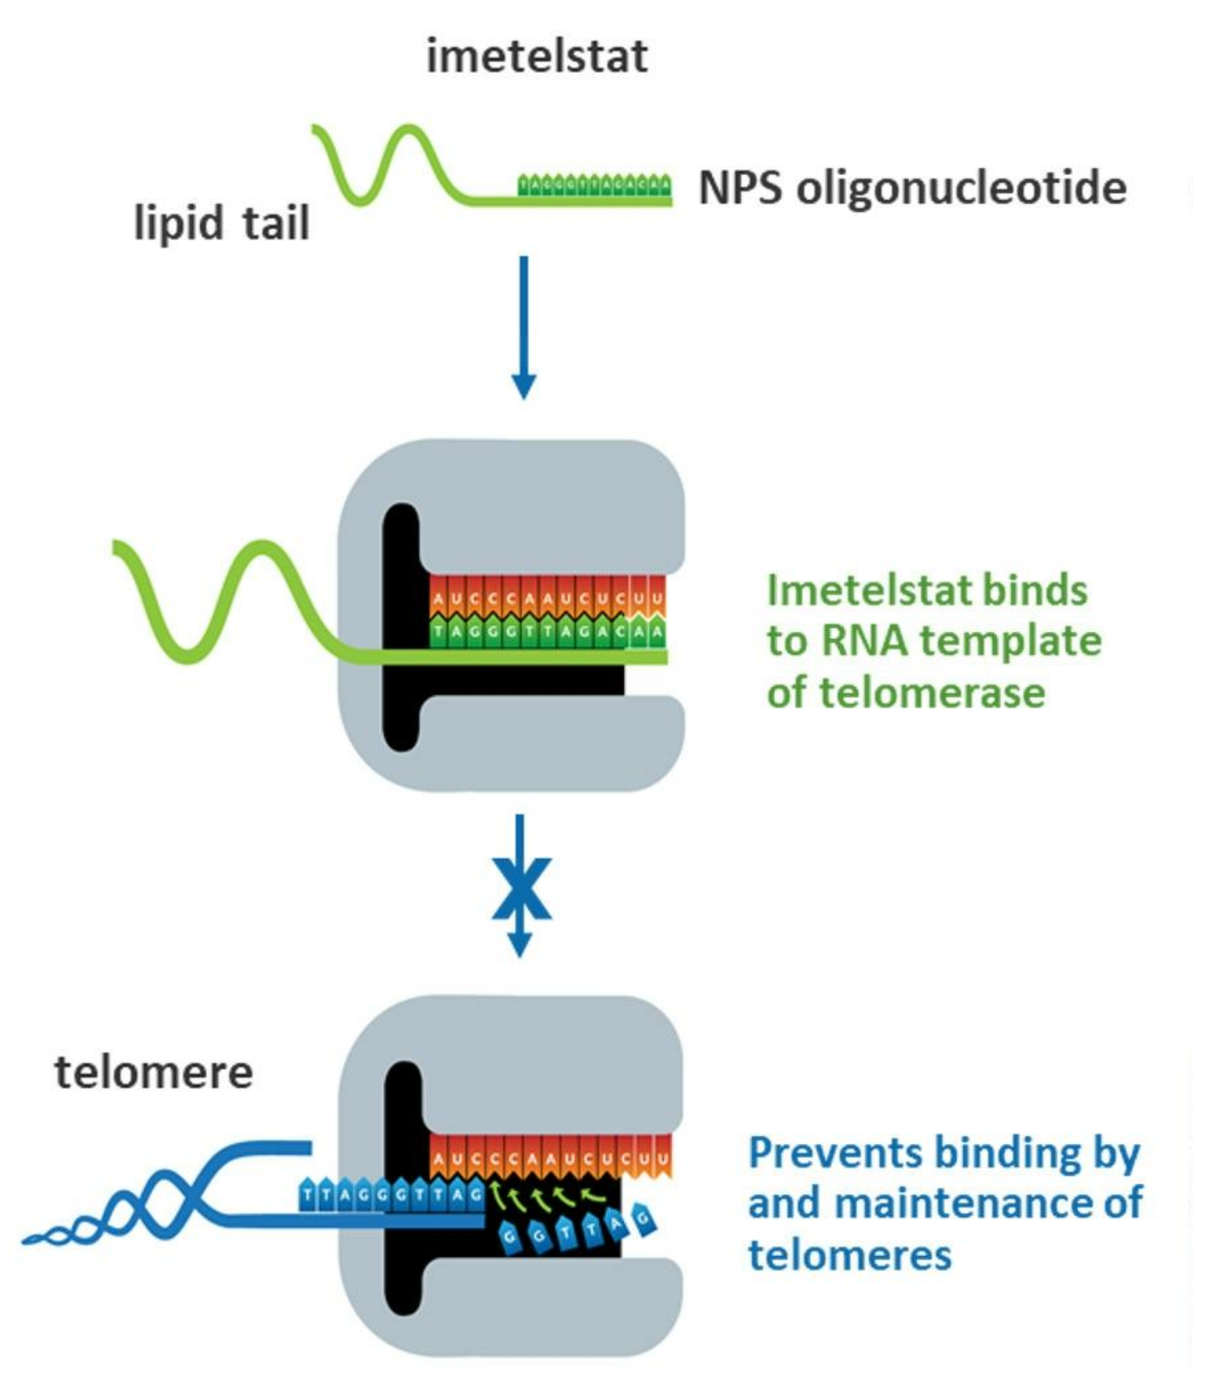
imetelstat
lipid tail
NPS oligonucleotide
Imetelstat binds to RNA template of telomerase
telomere
Prevents binding by and maintenance of telomeres

Patient Population

- Patients who participated in MYF2001 had MF that was R/R to JAKi, defined as documented progressive disease (PD) during or after JAKi treatment (**Table 1**).

Table 1. MYF2001: Patient Eligibility

- Int-2/high-risk MF per DIPSS criteria
- Relapsed or refractory to JAKi defined as documented progressive disease during or after JAKi:
 - o Subjects must have worsening of splenomegaly-related abdominal pain at any time after the start of JAKi therapy and EITHER:
 - – No reduction in spleen volume or size after 12 weeks of JAKi therapy, or
 - – Worsening splenomegaly* at any time after the start of JAKi therapy documented by:
 - o Increase in spleen volume from nadir by 25% measured by MRI or CT, or
 - o Increase in spleen size by palpation, CT, or ultrasound
- Active symptoms of MF
- Baseline measurable splenomegaly (palpable spleen ≥5 cm below LCM or ≥450 cm^3 by MRI)

*Adapted from IWG-MRT response criteria definition of progressive disease.
CT, computed tomography; JAKi, Janus Kinase inhibitor; LCM, left costal margin; MF, myelofibrosis; MRI, magnetic resonance imaging.

- For the historical control, external RWD were collected from a single-center study at the Moffitt Cancer Center which included 96 patients who had discontinued ruxolitinib.
 - o A closely matched cohort was identified using the guidelines for inclusion and exclusion criteria as defined in MYF2001 protocol.
 - o The cohort consisted of patients with MF who had discontinued JAKi due to lack or loss of response and were subsequently treated with BAT at the Moffitt Cancer Center from January 1998 to August 2018.

Statistical Methods

- To mimic the effect of randomization and improve comparability, a propensity score analysis approach, using average treatment effect for overlap population (ATO) and stabilized inverse probability treatment weighting (sIPTW), was conducted to balance the populations with respect to important baseline covariates and prognostic factors that may impact OS outcomes. The following critical baseline covariates were adjusted:
 - o Age
 - o Platelet count
 - o Time from diagnosis to JAKi discontinuation
 - o JAKi duration
 - o Spleen size
 - o JAK2 mutation
 - o Sex
 - o DIPSS score
 - o ECOG performance status
 - o MF type
 - o Transfusion status
- Primary analysis: OS was measured from the time of JAKi discontinuation to death or censored at last follow-up. All patients from the analysis populations of the MYF2001 and the RWD were included.
- Sensitivity analysis was performed to address the immortal time bias introduced by early deaths post JAKi discontinuation observed in the RWD, that is to align more closely with the MYF2001 clinical trial experience, where such patients would not have completed the screening phase:
 - o The RWD set excluded 2 patients who died within 1 month post JAKi discontinuation, with OS measured from 1 month post JAKi discontinuation.
 - o For MYF2001, OS was measured from the randomization date.
- A second sensitivity analysis was conducted to assess the impact of subsequent hematopoietic stem cell transplant on OS.

RESULTS

Patients

- The analysis population included (**Figure 2**):
 - o 57 patients treated with imetelstat 9.4 mg/kg from MYF2001
 - – Median follow-up, 23 months
 - o 38 patients treated with BAT from RWD
 - – Median follow-up, 43 months
- Baseline covariates had improved balance after propensity score adjustment (**Table 2**).
 - o Without adjustment, baseline covariates were generally imbalanced between the MYF2001 and RWD groups. A statistically significant standardized mean difference for the baseline spleen size, JAKi duration and time from JAKi discontinuation was observed (p<0.01).
 - o After adjustment for baseline covariate imbalances using the ATO method, standardized mean differences were minimized (all were <0.001). The sIPTW method also offered an improved balance between the groups in terms of the standardized mean differences (all were <0.3).

Figure 2. Analysis Population

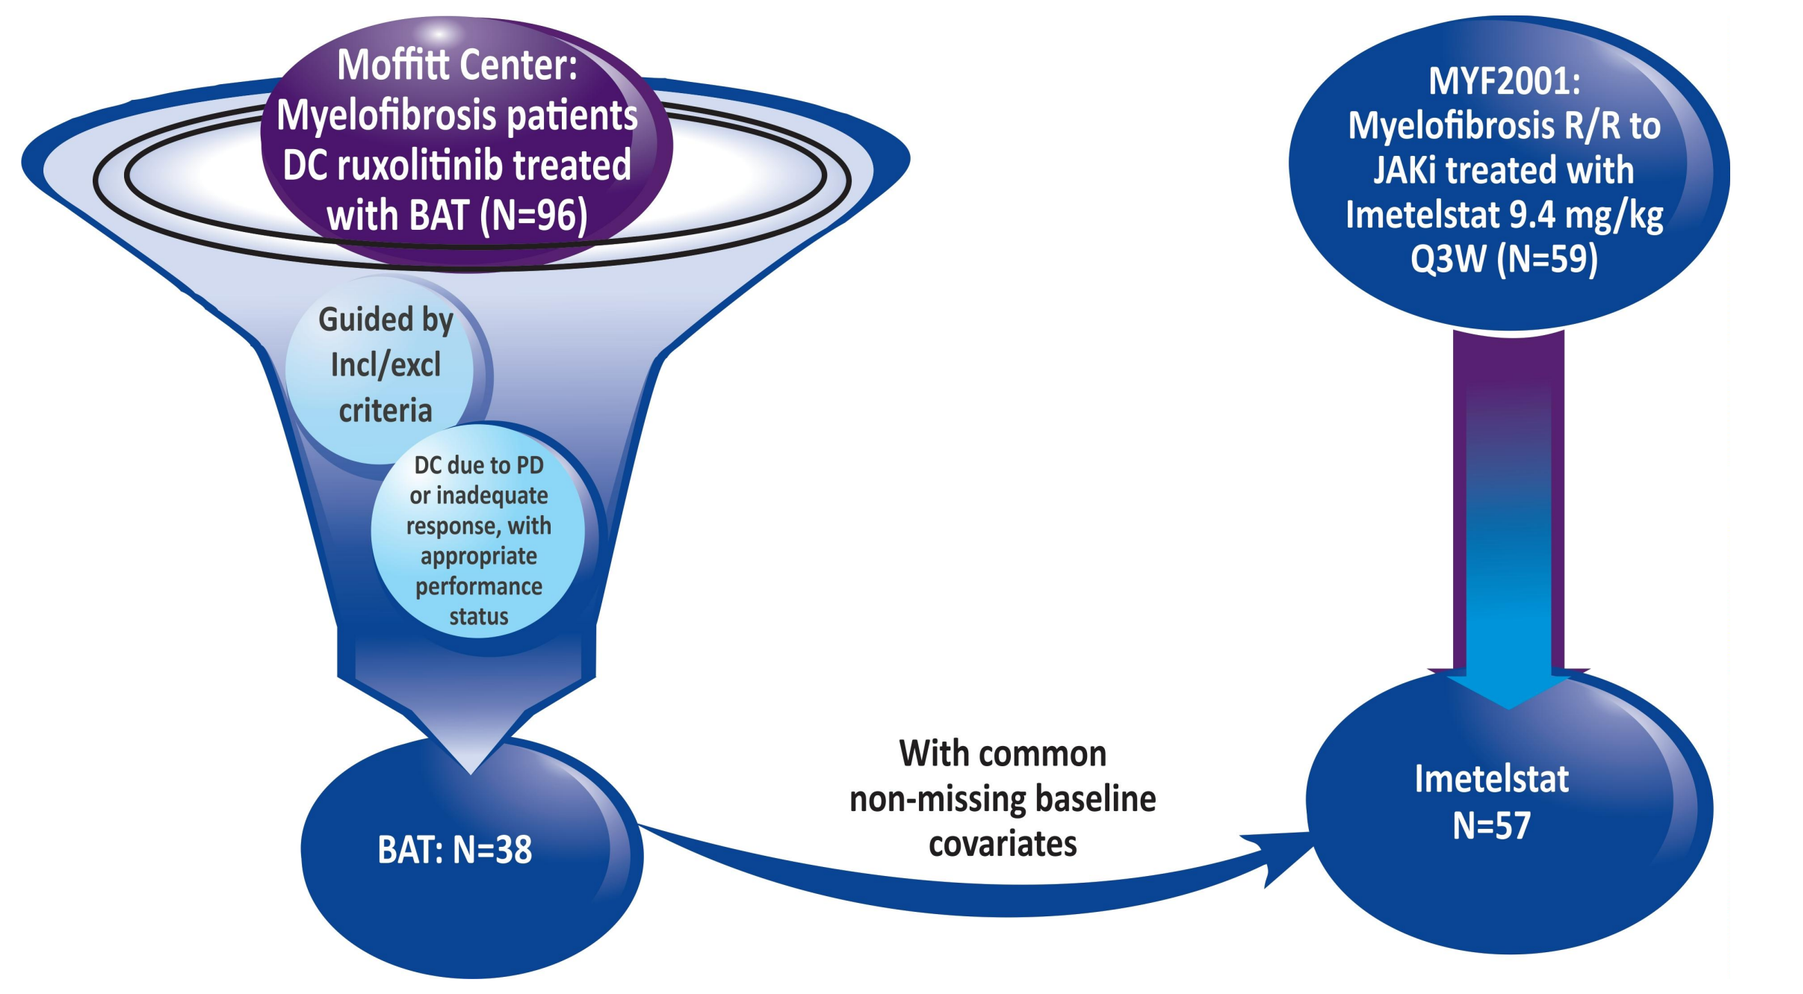
Moffitt Center: Myelofibrosis patients DC ruxolitinib treated with BAT (N=96)
Guided by Incl/excl criteria
DC due to PD or inadequate response, with appropriate performance status
BAT: N=38
With common non-missing baseline covariates
MYF2001: Myelofibrosis R/R to JAKi treated with Imetelstat 9.4 mg/kg Q3W (N=59)
Imetelstat N=57

BAT, best available therapy; DC, discontinuation; JAKi, Janus Kinase inhibitor; PD, progressive disease.

Table 2. Baseline Characteristics

Without Adjustment

	Levels	IMETELSTAT 9.4 MG/KG	BAT (Moffitt)	p-value	Standardized Mean Difference
N		57	38		Unweighted
AGE at Rux DC (years) (mean (sd))		66.39 (9.48)	67.84 (10.74)	0.488	0.144
SEX (%)	F	23 (40.4)	13 (34.2)	0.698	0.127
	M	34 (59.6)	25 (65.8)		
Subpopulation of MF (%)	Primary	34 (59.6)	26 (68.4)	0.515	0.184
	Post-ET/Post-PV	23 (40.4)	12 (31.6)		
DIPSS Risk Status (%)	Intermediate-1/2	33 (57.9)	27 (71.1)	0.278	0.278
	High-Risk	24 (42.1)	11 (28.9)		
Baseline Spleen Size (mean (sd))		17.18 (7.41)	13.05 (7.34)	0.009	0.559
Baseline Platelet Count (x10E9/L) (mean (sd))		212.11 (161.15)	169.53 (104.69)	0.154	0.313
ECOG 0 (%)	0/1	46 (80.7)	24 (63.2)	0.096	0.398
	2/3	11 (19.3)	14 (36.8)		
Transfusion Dependent (%)	No	45 (78.9)	22 (57.9)	0.048	0.465
	Yes	12 (21.1)	16 (42.1)		
Time from Diagnosis to JAKi DC (months) (mean (sd))		55.16 (45.82)	26.94 (29.36)	0.001	0.734
Duration of JAKi treatment (months) (mean (sd))		25.12 (17.98)	13.85 (14.06)	0.002	0.698
Biomarker JAK2n (%)	Negative	25 (43.9)	7 (18.4)	0.019	0.571
	Postive	32 (56.1)	31 (81.6)		

With Adjustment (Propensity Score With ATO)

	Levels	IMETELSTAT 9.4 MG/KG	BAT (Moffitt)	p-value	Standardized Mean Difference
N		47.5	47.5		Overlap
AGE at Rux DC (years) (mean (sd))		66.64 (9.97)	66.64 (11.82)	1.000	<0.001
SEX (%)	F	18.5 (39.0)	18.5 (39.0)	1.000	<0.001
	M	29.0 (61.0)	29.0 (61.0)		
Subpopulation of MF (%)	Primary	32.8 (69.0)	32.8 (69.0)	1.000	<0.001
	Post-ET/Post-PV	14.7 (31.0)	14.7 (31.0)		
DIPSS Risk Status (%)	Intermediate-1/2	30.4 (63.9)	30.4 (63.9)	1.000	<0.001
	High-Risk	17.1 (36.1)	17.1 (36.1)		
Baseline Spleen Size (mean (sd))		15.01 (6.96)	15.01 (6.94)	1.000	<0.001
Baseline Platelet Count (x10E9/L) (mean (sd))		178.54 (127.97)	178.54 (104.23)	1.000	<0.001
ECOG 0 (%)	0/1	34.7 (73.0)	34.7 (73.0)	1.000	<0.001
	2/3	12.8 (27.0)	12.8 (27.0)		
Transfusion Dependent (%)	No	32.2 (67.8)	32.2 (67.8)	1.000	<0.001
	Yes	15.3 (32.2)	15.3 (32.2)		
Time from Diagnosis to JAKi DC (months) (mean (sd))		34.73 (33.14)	34.73 (34.40)	1.000	<0.001
Duration of JAKi treatment (months) (mean (sd))		16.80 (13.25)	16.80 (15.80)	1.000	<0.001
Biomarker JAK2n (%)	Negative	12.2 (25.6)	12.2 (25.6)	1.000	<0.001
	Postive	35.3 (74.4)	35.3 (74.4)		

With Adjustment (Propensity Score With sIPTW)

	Levels	IMETELSTAT 9.4 MG/KG	BAT (Moffitt)	p-value	Standardized Mean Difference
N		59.17	32.49		Stabilized IPTW
AGE at Rux DC (years) (mean (sd))		66.70 (9.10)	67.01 (11.42)	0.900	0.031
SEX (%)	F	22.6 (38.3)	11.7 (36.1)	0.856	0.045
	M	36.4 (61.7)	20.7 (63.9)		
Subpopulation of MF (%)	Primary	40.1 (67.8)	22.4 (69.0)	0.915	0.026
	Post-ET/Post-PV	19.0 (32.2)	10.1 (31.0)		
DIPSS Risk Status (%)	Intermediate-1/2	36.1 (61.1)	20.7 (63.7)	0.837	0.053
	High-Risk	23.0 (38.9)	11.8 (36.3)		
Baseline Spleen Size (mean (sd))		15.71 (7.20)	14.60 (6.98)	0.491	0.156
Baseline Platelet Count (x10E9/L) (mean (sd))		192.93 (144.47)	178.56 (108.75)	0.633	0.112
ECOG 0 (%)	0/1	43.3 (73.2)	23.9 (73.5)	0.98	0.006
	2/3	15.8 (26.8)	8.6 (26.5)		
Transfusion Dependent (%)	No	40.5 (68.5)	22.0 (67.9)	0.957	0.014
	Yes	18.6 (31.5)	10.4 (32.1)		
Time from Diagnosis to JAKi DC (months) (mean (sd))		43.26 (41.34)	34.52 (33.62)	0.304	0.232
Duration of JAKi treatment (months) (mean (sd))		20.41 (16.26)	16.11 (15.69)	0.263	0.269
Biomarker JAK2n (%)	Negative	18.6 (31.4)	9.1 (28.1)	0.782	0.073
	Postive	40.5 (68.6)	23.4 (71.9)		

ATO, average treatment effect for overlap; BAT, best available therapy; DC, discontinuation; DIPSS, Dynamic International Prognostic Scoring System; ECOG, Eastern Cooperative Oncology Group; JAKi, Janus Kinase inhibitor; MF, myelofibrosis; RUX, ruxolitinib; sIPTW, stabilized inverse probability treatment weighting.

OS

- Statistically significant reductions in death were observed with imetelstat treatment across all analyses:
 - o There was a 65% lower risk of death with imetelstat vs BAT per both the unweighted analysis (median OS, 33.77 vs 12.04 months) and overlap weighting with ATO (median OS, 30.69 vs 12.04 months) (**Figure 3A** and **3B**).
 - o Per sIPTW, median OS durations were the same as those per the ATO method and the corresponding reduction in the risk of death with imetelstat was 67% (**Figure 3C**).
- Sensitivity analysis results addressing the immortal time bias introduced by early deaths post JAKi discontinuation observed in the RWD (**Figure 4A-C**) were consistent with the primary findings.
 - o With the removal of early deaths observed in the RWD from these analysis, imetelstat conferred a 65% lower risk of death vs BAT in the unweighted analysis and similar reductions of 64% and 66% per ATO and sIPTW, respectively.
- Sensitivity analysis results addressing the impact of subsequent transplant on OS were also consistent with the primary findings.
 - o Unweighted analysis: median OS with imetelstat vs BAT of 30.69 vs 10.23 months, with a 69% reduction in the risk of death.
 - o ATO and sIPTW: median OS with imetelstat vs BAT of 30.69 vs 12.04 months, with 68% and 70% reductions in the risk of death per ATO and sIPTW, respectively.

Figure 3. Overall survival: primary analysis

A. Unadjusted analysis (unweighted)*

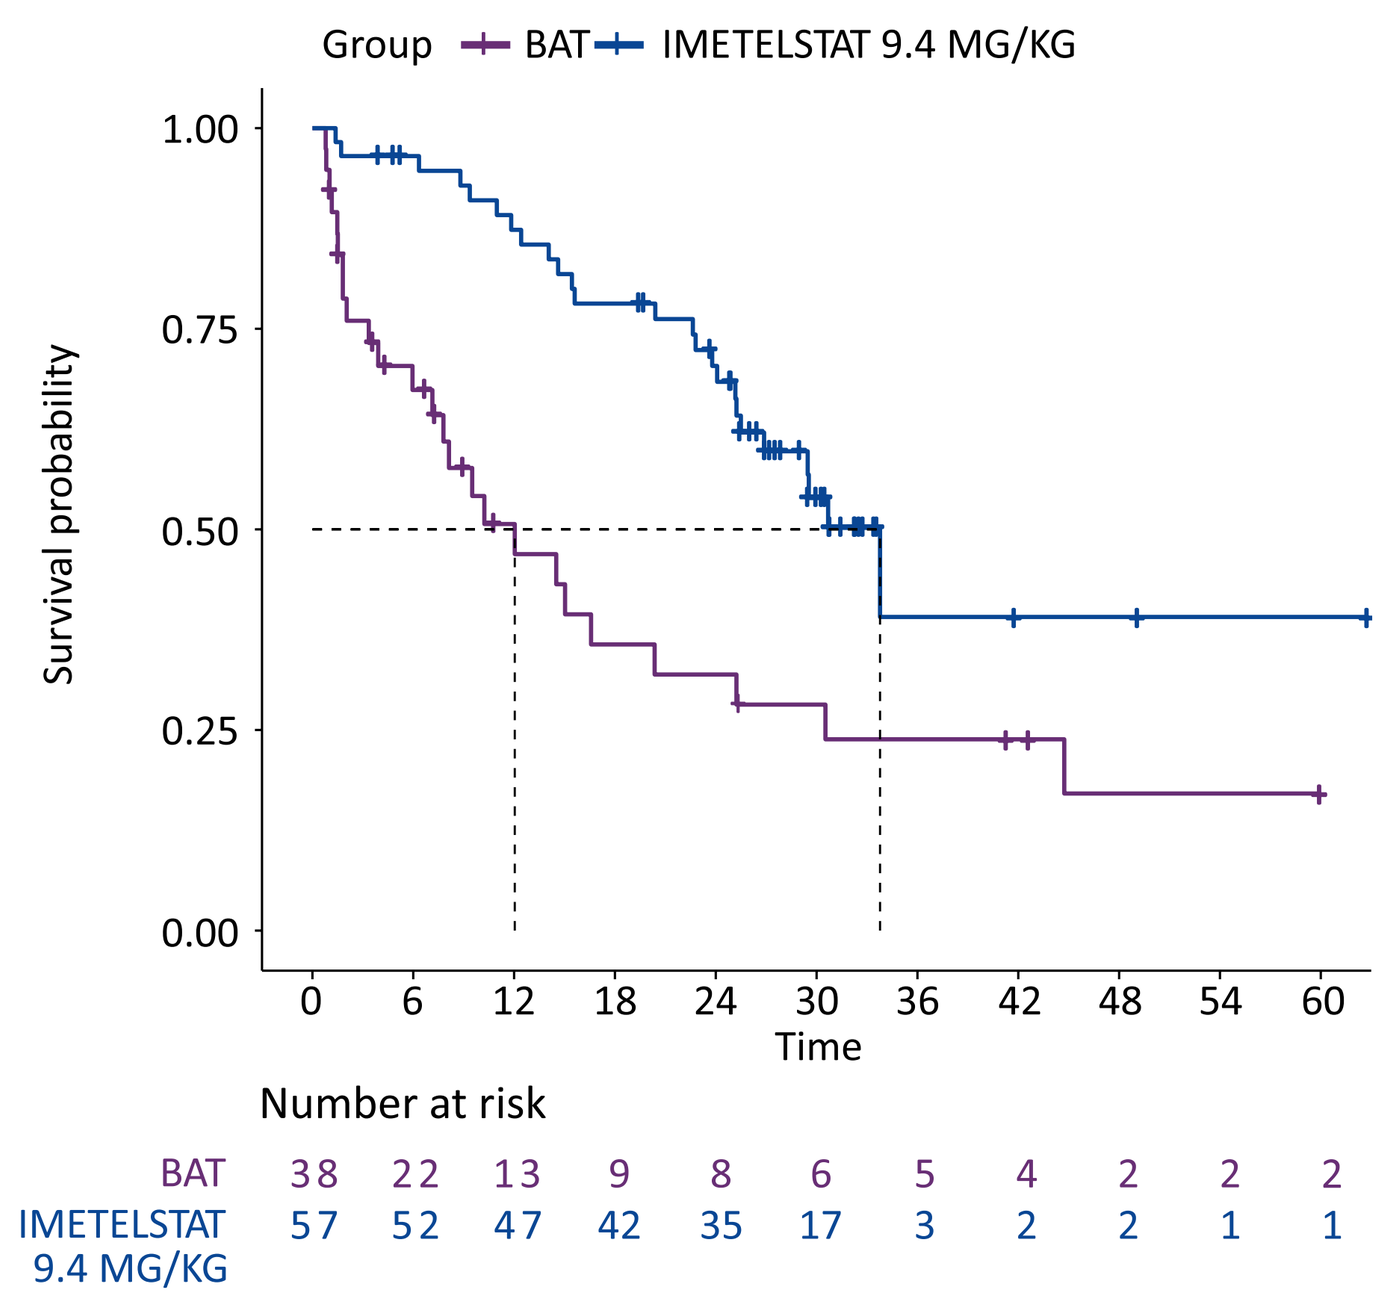
Kaplan–Meier Curves (Unweighted) for Naive Comparison
Group BAT IMETELSTAT 9.4 MG/KG
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 38 22 13 9 8 6 5 4 2 2 2
IMETELSTAT 9.4 MG/KG 57 52 47 42 35 17 3 2 2 1 1

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	33.77 (26.87, NE)	12.04 (7.80, 30.53)
Q1, Q3	22.64, NE	3.36, 30.53
Range	(0.66, 17.41+)	(0.92+, 17.74)
Overall survival rates (95% CI)		
12-month	87% (79%, 97%)	51% (36%, 71%)
24-month	70% (59%, 84%)	32% (19%, 54%)
Hazard ratio (95% CI)	0.35 (0.20, 0.62)	
P-value	0.0003	

B. Propensity score analysis with ATO (overlap weighting)*†

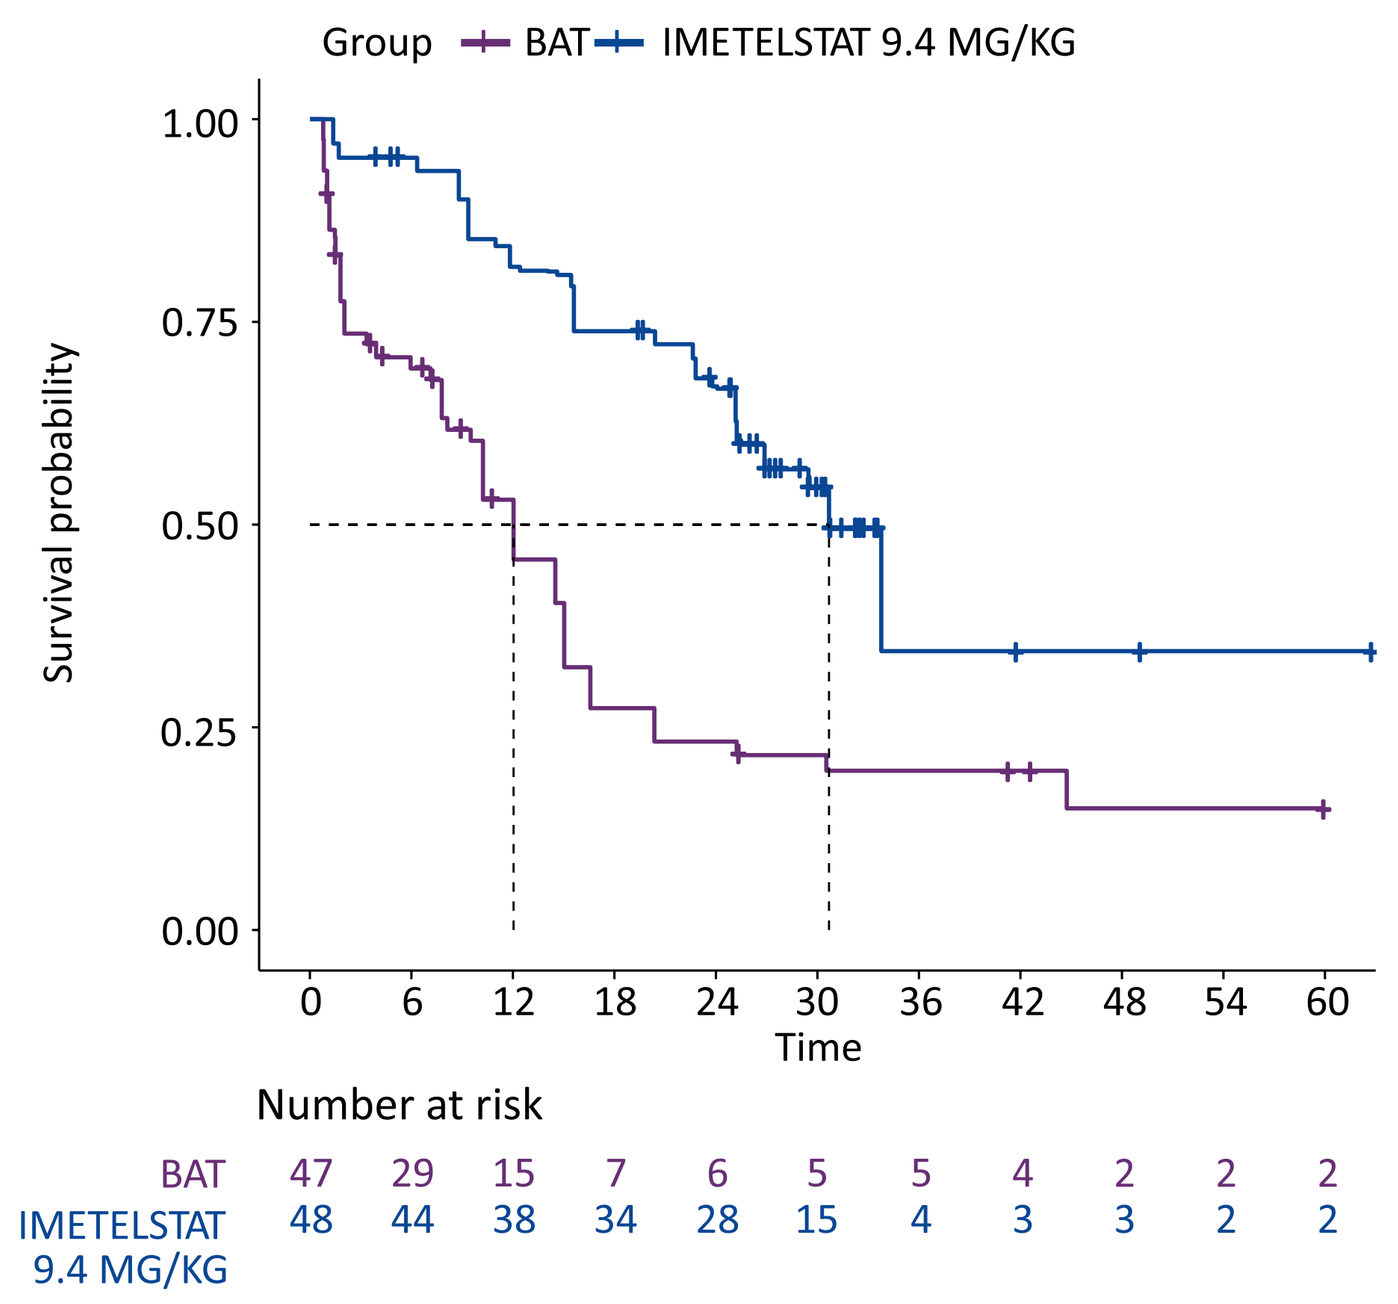
Kaplan–Meier Curves (Overlap)
Group BAT IMETELSTAT 9.4 MG/KG
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 47 29 15 7 6 5 5 4 2 2 2
IMETELSTAT 9.4 MG/KG 48 44 38 34 28 15 4 3 3 2 2

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	30.69 (25.17, NE)	12.04 (7.80, 16.58)
Q1, Q3	15.61, NE	2.04, 20.36
Range	(1.38, 62.82+)	(0.79, 60.00+)
Overall survival rates (95% CI)		
12-month	82% (70%, 96%)	53% (37%, 76%)
24-month	67% (53%, 85%)	23% (12%, 44%)
Hazard ratio (95% CI)	0.35 (0.18, 0.68)	
P-value	0.0019	

C. Stabilized inverse probability treatment weighting (sIPTW)*†

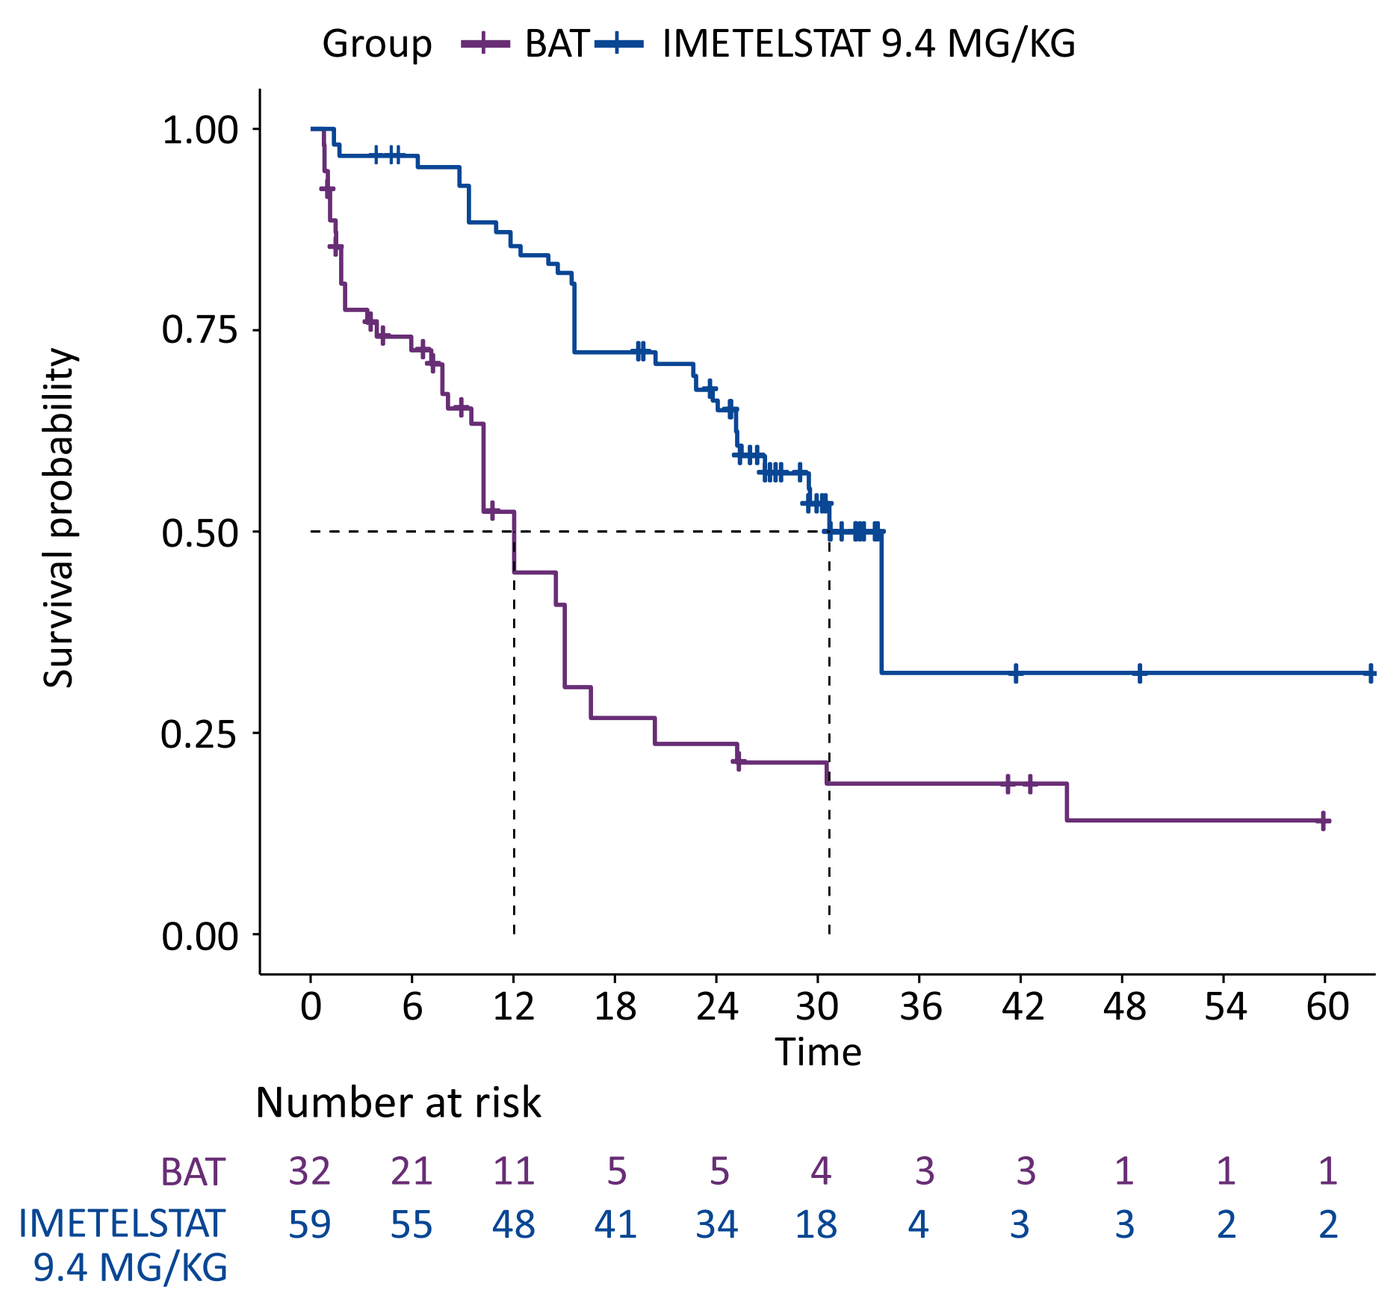
Kaplan–Meier Curves (Stabilized IPTW)
Group BAT IMETELSTAT 9.4 MG/KG
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 32 21 11 5 5 4 3 3 1 1 1
IMETELSTAT 9.4 MG/KG 59 55 48 41 34 18 4 3 3 2 2

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	30.69 (25.17, NE)	12.04 (9.51, 16.58)
Q1, Q3	15.61, NE	3.91, 20.36
Range	(1.38, 62.82+)	(0.79, 60.00+)
Overall survival rates (95% CI)		
12-month	85% (75%, 97%)	52% (36%, 76%)
24-month	66% (52%, 84%)	24% (13%, 42%)
Hazard ratio (95% CI)	0.33 (0.18, 0.61)	
P-value	0.0003	

BAT, best available therapy.
*Measured from the date of time of JAKi discontinuation to the date of death by any causes. †Adjusted for a continuous variables age, platelet count, time from diagnosis to JAKi discontinuation, duration of JAKi, and spleen size, and categorical variables sex, DIPSS score, ECOG, type of MF, transfusion status, and JAK2 mutation.

Figure 4. Overall survival: sensitivity analysis

A. Unadjusted analysis (unweighted)*

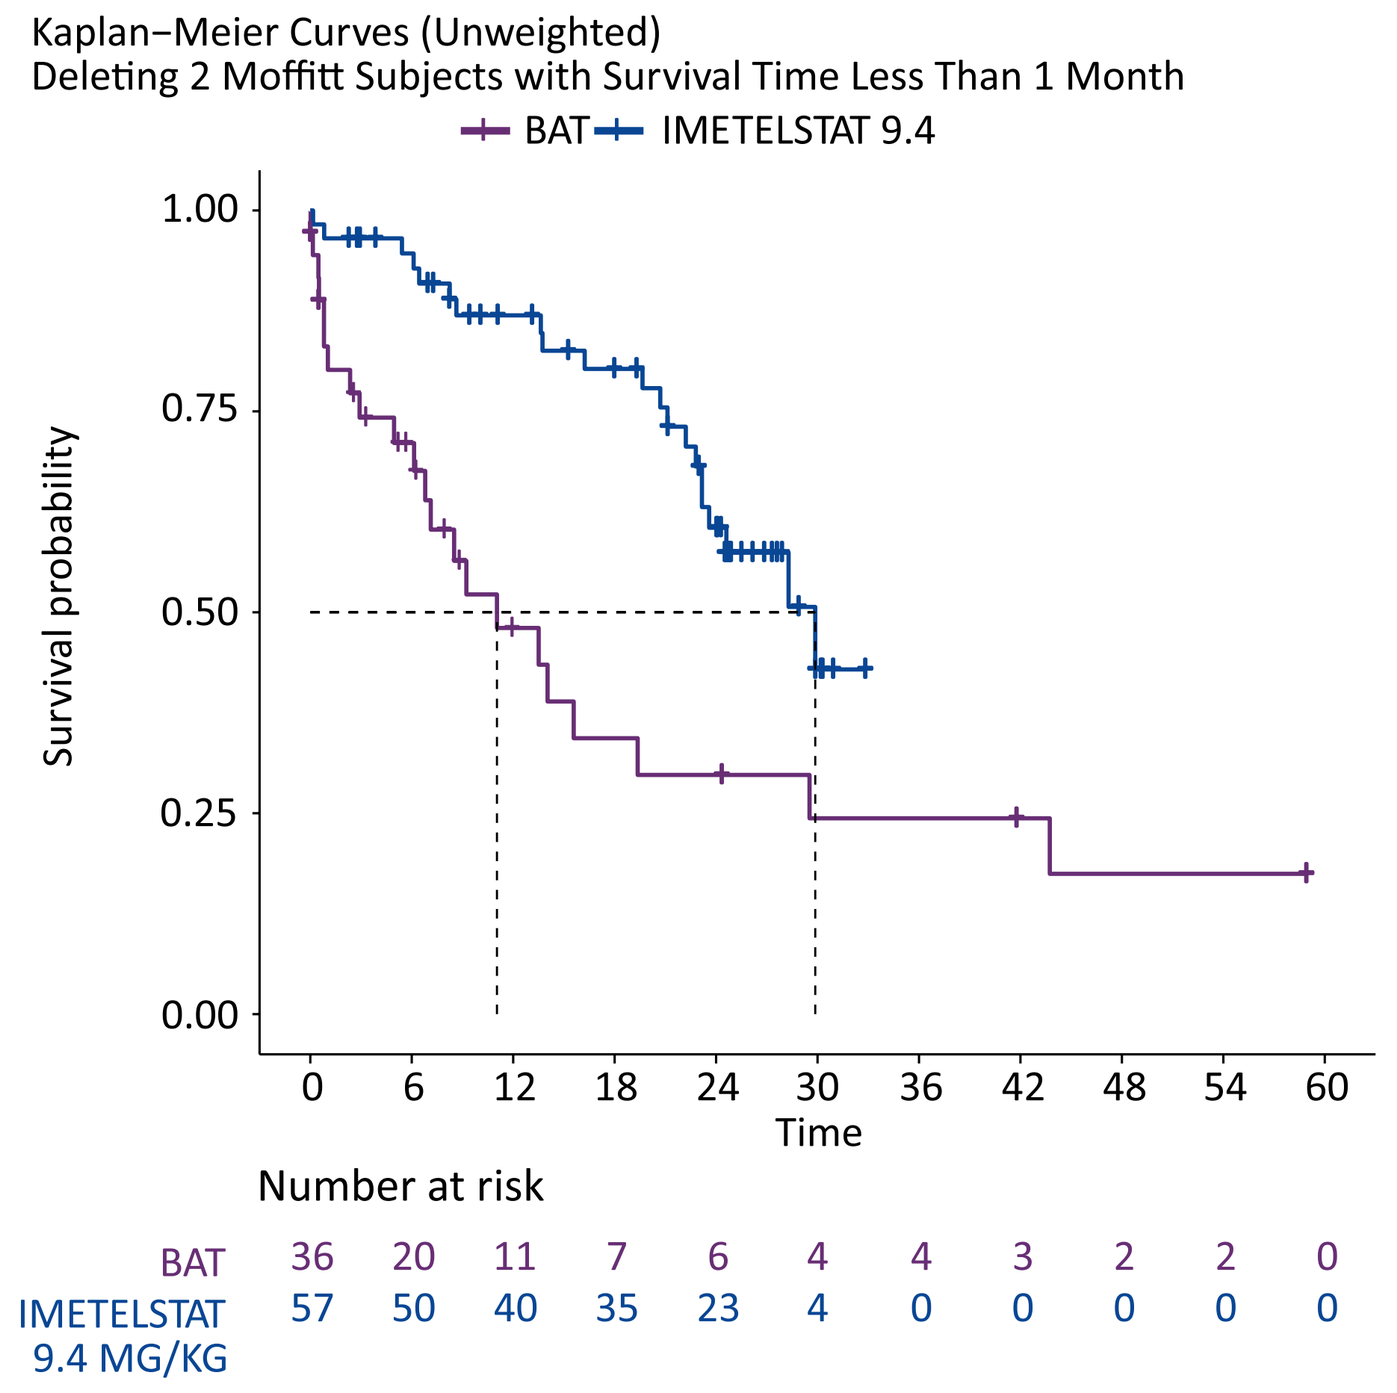
Kaplan–Meier Curves (Unweighted)
Deleting 2 Moffitt Subjects with Survival Time Less Than 1 Month
BAT IMETELSTAT 9.4
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 36 20 11 7 6 4 4 3 2 2 0
IMETELSTAT 9.4 MG/KG 57 50 40 35 23 4 0 0 0 0 0

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	29.86 (23.59, NE)	11.04 (6.80, 43.74)
Q1, Q3	21.13, NE	2.91, 29.53
Range	(0.16, 32.92+)	(0.02, 59.00+)
Overall survival rates (95% CI)		
12-month	87% (78%, 96%)	48% (33%, 70%)
24-month	61% (48%, 77%)	30% (16%, 55%)
Hazard ratio (95% CI)	0.35 (0.19, 0.65)	
P-value	0.0008	

B. Propensity score analysis with ATO (overlap weighting)*†

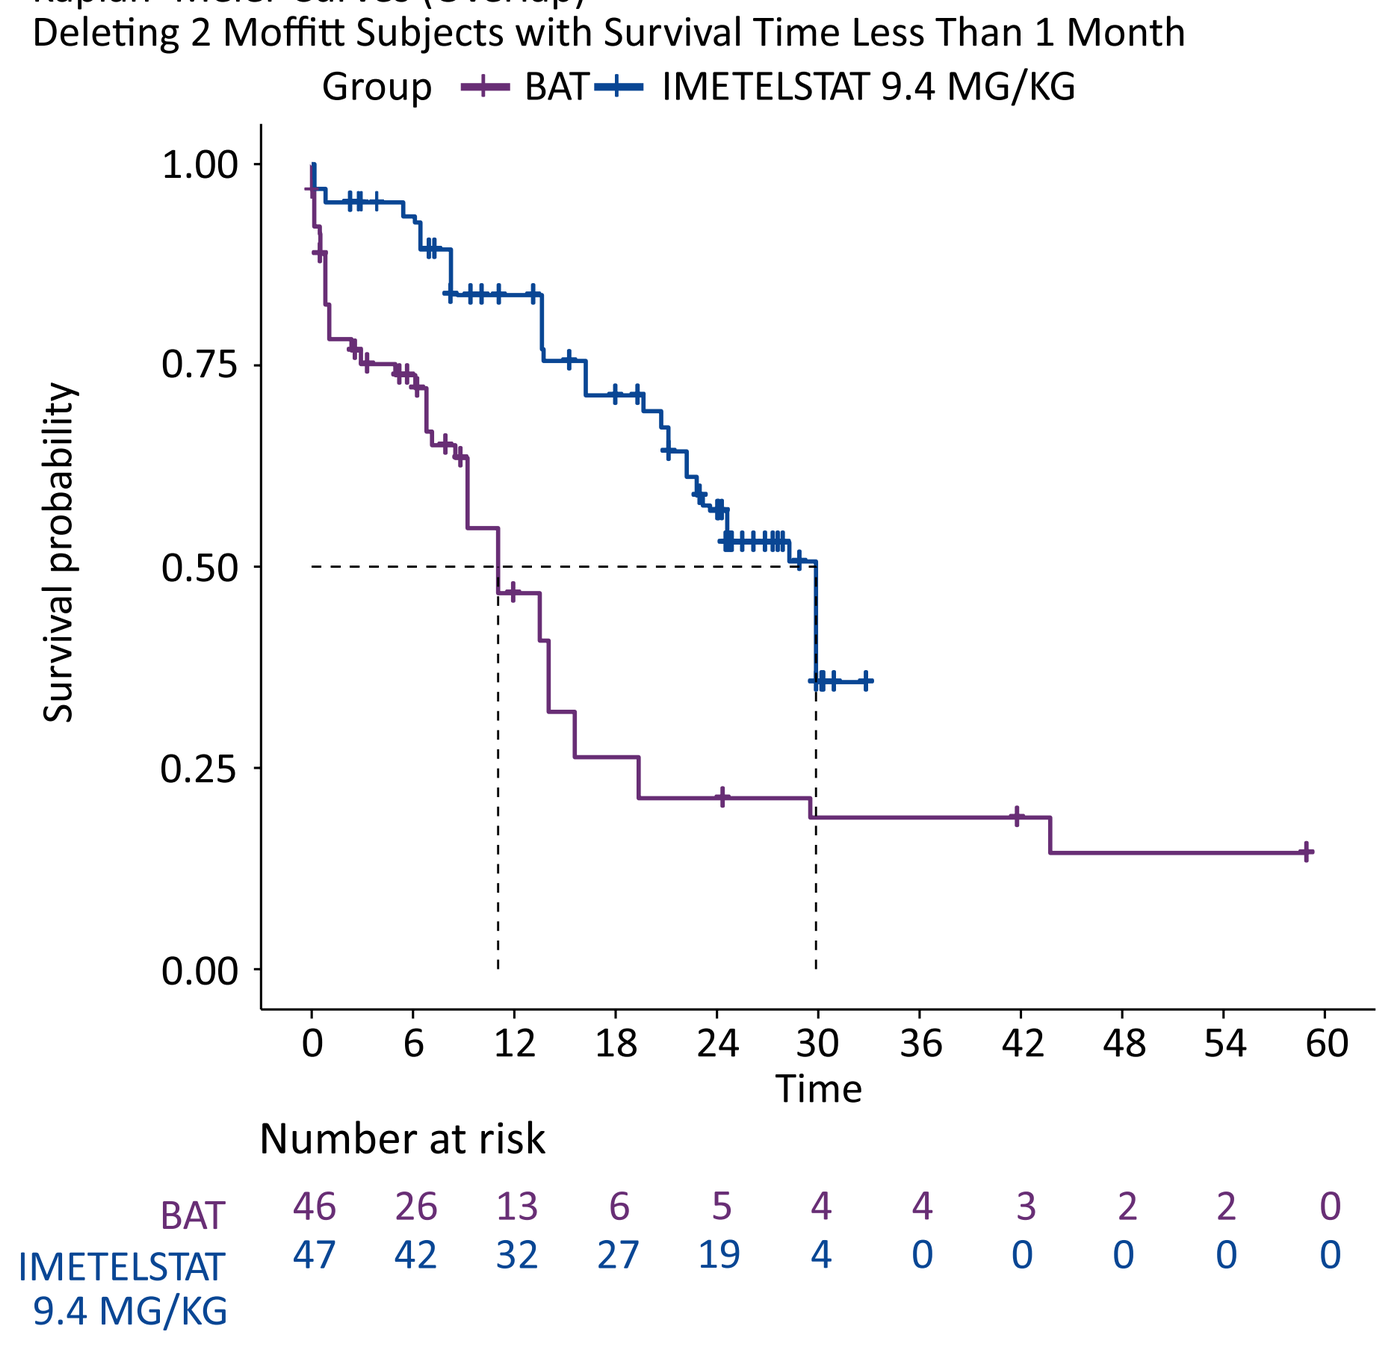
Kaplan–Meier Curves (Overlap)
Deleting 2 Moffitt Subjects with Survival Time Less Than 1 Month
Group BAT IMETELSTAT 9.4 MG/KG
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 46 26 13 6 5 4 4 3 2 2 0
IMETELSTAT 9.4 MG/KG 47 42 32 27 19 4 0 0 0 0 0

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	29.86 (21.13, NE)	11.04 (7.13, 19.36)
Q1, Q3	16.23, NE	4.95, 19.36
Range	(0.16, 32.92+)	(0.02, 59.00+)
Overall survival rates (95% CI)		
12-month	84% (71%, 98%)	47% (30%, 72%)
24-month	57% (41%, 78%)	21% (10%, 44%)
Hazard ratio (95% CI)	0.36 (0.18, 0.73)	
P-value	0.0044	

C. Stabilized inverse probability treatment weighting (sIPTW)*†

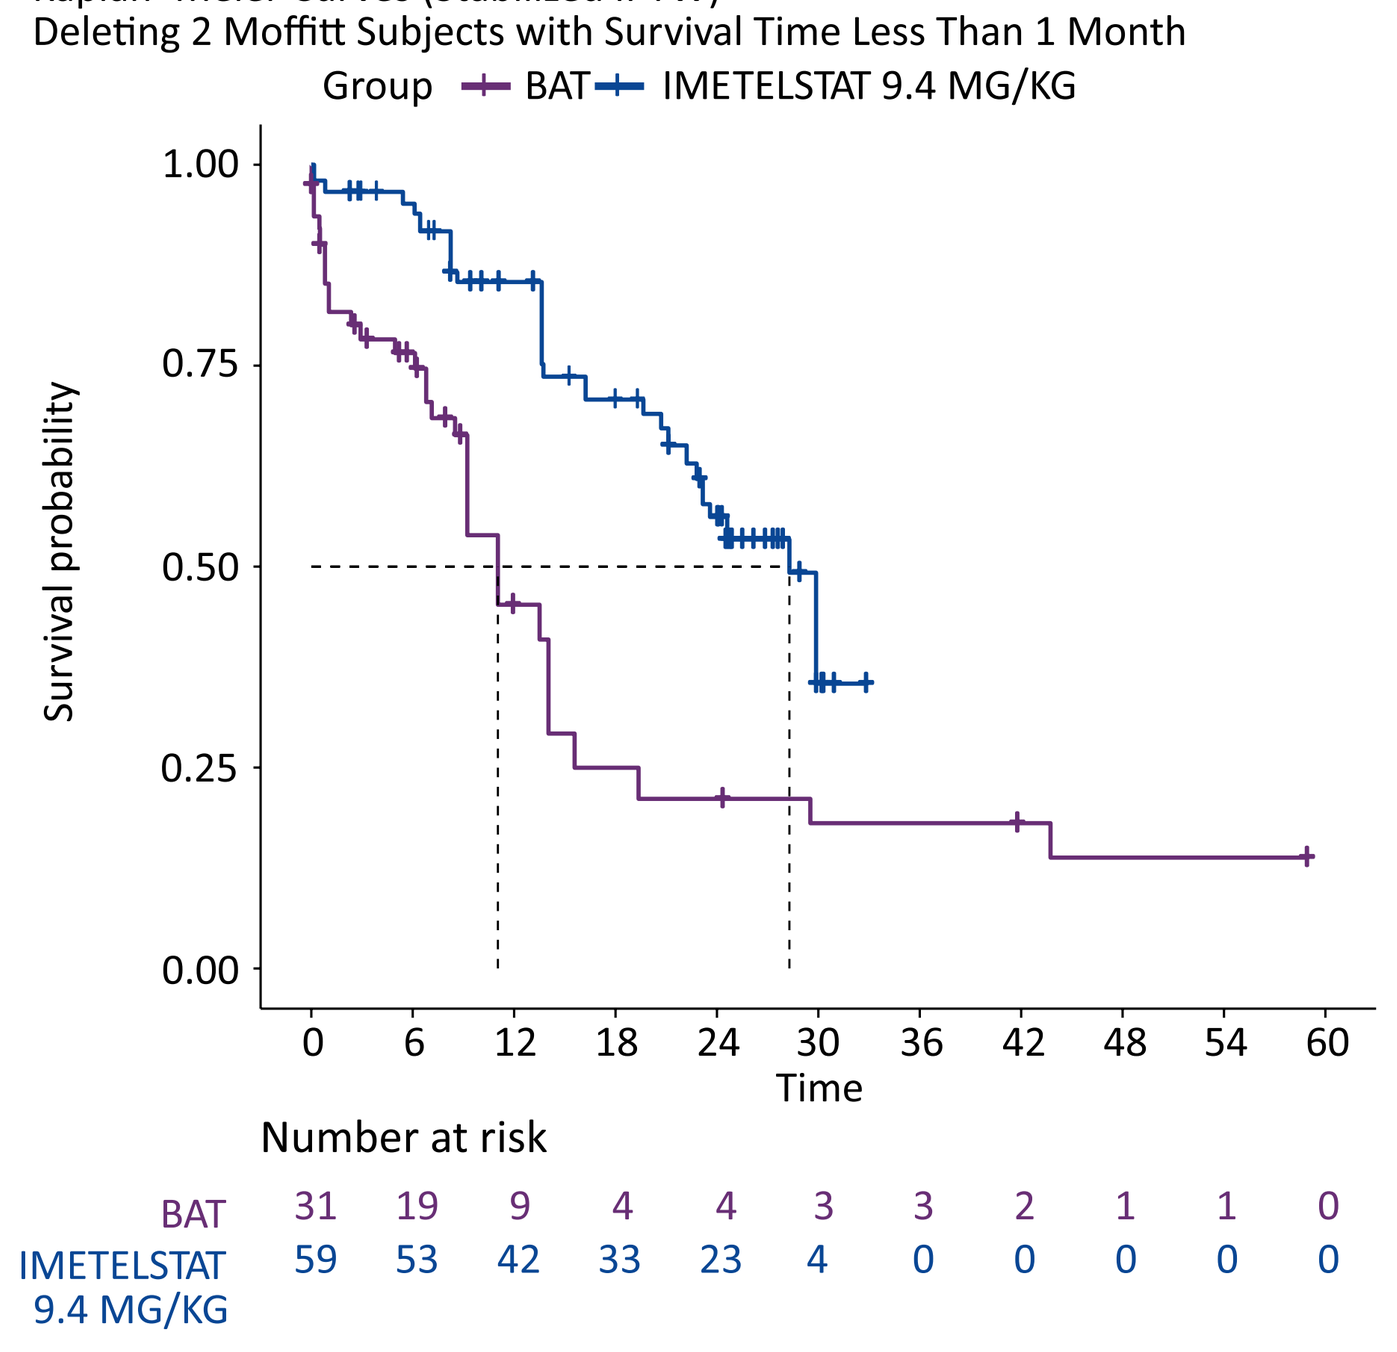
Kaplan–Meier Curves (Stabilized IPTW)
Deleting 2 Moffitt Subjects with Survival Time Less Than 1 Month
Group BAT IMETELSTAT 9.4 MG/KG
Survival probability
1.00
0.75
0.50
0.25
0.00
0 6 12 18 24 30 36 42 48 54 60
Time
Number at risk
BAT 31 19 9 4 4 3 3 2 1 1 0
IMETELSTAT 9.4 MG/KG 59 53 42 33 23 4 0 0 0 0 0

	Imetelstat (MYF2001)	BAT (Moffitt)
Overall survival (months)		
Median (95% CI)	28.29 (21.13, NE)	11.04 (9.23, 14.03)
Q1, Q3	13.73, NE	6.14, 15.58
Range	(0.16, 32.92+)	(0.02, 59.00+)
Overall survival rates (95% CI)		
12-month	85% (75%, 98%)	45% (28%, 72%)
24-month	56% (41%, 77%)	21% (11%, 41%)
Hazard ratio (95% CI)	0.34 (0.18, 0.66)	
P-value	0.0012	

BAT, best available therapy.
*Measured from the date of randomization to the date of death by any causes for MYF2001 and from 1 month post JAKi discontinuation to the date of death by any causes for the Moffitt dataset; overall survival was censored at transplant. †Adjusted for a continuous variables age, platelet count, time from diagnosis to JAKi discontinuation, duration of JAKi, and spleen size, and categorical variables sex, DIPSS score, ECOG, type of MF, transfusion status, and JAK2 mutation.

SUMMARY/CONCLUSION

- These analyses showed that treatment with imetelstat was associated with a lower risk of death compared to BAT in closely matched patients from RWD with Int-2 or High-Risk MF after JAKi failure.
 - o Median OS was 33.77 months with imetelstat in the unweighted primary analysis (30.69 months with propensity score matching) vs 12.04 months with BAT, with a 65–67% lower risk of death across the primary analyses.
 - o All sensitivity analyses were consistent with the primary results.
- Acknowledging the limitations of such comparative analyses between RWD and clinical trial data, favorable OS of imetelstat treatment in this very poor-prognosis patient population warrants further evaluation.

References: 1. Kuykendall AT, et al. Ann Hematol 2018;97:435-441; 2. Newberry KJ, et al. Blood 2017;130:1125-1131; 3. Mascarenhas J, et al. ASH 2018. Abstract & oral presentation #685

Acknowledgements: Dr. Laurie Orloski (independent medical writer) provided writing assistance and Dr. Harry Ma (Janssen Research & Development, LLC.) provided additional editorial support for this poster.

Poster Presented at the 24th European Hematology Association (EHA) Congress, June 13-16, 2019, Amsterdam, The Netherlands